6/12/2012

A translational threshold model to assess exposure-driven QTc changes

N. Frances (1), C. Meille (1), A. Caruso (1), J. Fretland (2), A. C. Harrison (2), A. Greiter-Wilke (3), M. Sanders (3), T. Lavé (1)

F. Hoffmann-La Roche Ltd., Pharma Research and Early Development, Non Clinical Safety, (1) Drug metabolism and Pharmacokinetics (DMPK), Modeling and Simulation; (2) DMPK (3) Safety Pharmacology

ABSTRACT

Objective: QT prolongation in the electrocardiogram is a surrogate marker of 'torsade de pointes' (fatal arrhythmias). This QT parameter is investigated during pre-clinical cardiovascular telemetry studies and it is critical, in case of QT prolongation, to define the safety margin for initiation of human trials. The objective of this study was to evaluate a threshold model to link QT with exposure and explore its translational capability.
Methods: The threshold model described below was applied to 3 compounds in pre-clinical development and for one compound for which clinical data was available. QTc (heart rate corrected QT) change was linked to exposure by a threshold model using a population approach (Monolix® 3.2). This model combines a baseline parameter (QTc baseline value), a threshold parameter which corresponds to the concentration below which there is no drug induced changes in QTc expected, and a slope parameter which corresponds to the QTc increase rate according to exposure above the threshold parameter value. Inter-individual variability was estimated on baseline and slope parameter. The pre-clinically assessed threshold parameter is then used for human prediction in combination with the predicted PK profile in man by PBPK modeling (GastroPlus®).
Results: The threshold model effectively described the relationship between exposure and heart rate corrected QT when data show a range of exposure with no effect on QTc and no saturation in effect. Under these circumstances, the threshold model can be used as an alternative to other existing models. The approach successfully described QTc change in human and showed translational properties of model parameters.
Conclusion: QTc and exposure can be linked by a threshold model and demonstrate translational properties from pre-clinical to clinical studies. In case of a QTc prolongation, this model predicts the exposure where effects are expected to occur (threshold parameter, which will likely be higher than the otherwise used No Effect Level). By using the threshold model to describe the exposure response relationship and translate the model to humans, a better prediction of where QTc prolongation is likely to occur can be estimated which provides more confidence in the dose escalation strategy.

4) CONCLUSION

A threshold model was developed to describe the relationship between exposure and QTc as an alternative to linear and Emax models. This model predicts the exposure where QTc is expected to be prolonged. This exposure can then be converted back into a dose level in the species were it has been generated, to support estimation of the NOEL.
The threshold model proved to be very appropriate to describe the exposure response relationship in animals and to translate the finding to humans using PBPK [3]. Such knowledge in the QTc prolongation vs. exposure relationship provides more confidence for designing first in human studies.
The threshold model allows also formal estimation of significant QTc prolongation relative to QTc baseline variability. This is important for the interpretation of the guidance for industry in QTc evaluation where it is recommended to assess drug effect leading to 5 to 10 ms QTc prolongation.
Additional factors may have to be accounted for when translating the findings from animals to man. This includes for example species differences in receptor affinity, and some kinetics aspects to reach the receptor site.
Additional investigations are ongoing to describe relationship between in vitro HERG [8] value and the threshold parameter.

1) INTRODUCTION

Regulatory agencies require in vivo assessments of drug associated QT interval prolongation [6].
QT is considered as an in vivo surrogate marker of 'torsade de pointes' (fatal arrhythmias).
Because of its inverse relationship to heart rate, the measured QT interval is routinely corrected by means of various formulae to a less heart rate dependent value known as the QTc interval.
Solid evaluation of the QT/QTc prolongation vs. exposure in pre clinical studies is necessary to support first dose recommendation in human.
The objective of this study was to evaluate a threshold model to link QTc with exposure and explore its translational capability. Results presented here are from 3 different compounds (A, B and C).

2) MATERIAL AND METHODS

- **Experimental conditions**

For all compounds and preclinical experiments, QT was recorded via telemetry using jacketed or implanted animals.

Compound	A	B	C	
Species	Dog	Monkey	Monkey	Human
Study status	GLP tox	GLP telemetry	GLP telemetry	Phase 1
n	32	8	6	35
Administration route	oral	oral	oral	oral
Dosing	4 groups: 3 doses levels + vehicle	All individuals: 3 dose levels + vehicle	All individuals: 3 doses levels + vehicle	Several groups: 8 dose levels + vehicle
Schedule of administration	MAD 3 weeks twice daily	SAD Latin square crossover design	MAD Latin square crossover design	SAD 2 to 3 dose levels per individuals
PK data	Sparse data	Sparse data	Sparse data	Rich data
QT data	Sparse data: predose and at max effect on day 9 and 15	Rich data (every hour during 24h)	Rich data (every hour during 24h)	Rich data (5 samples + predose value)

- **Data analysis**

The modeling analysis was done on dQTc which is :
- The heart rate corrected QT (QTc) calculated by Holzgrefe formula [1]
- And the difference between QTc value at one dose level and :
 - Either QTc value for time matched control (when available)
 - Or the pre dose value.

The threshold model can be an alternative to the linear [4] and Emax models [5] in the presence of a range of exposure without significant effect, or when a maximum effect is not reached.
3 parameters are needed for the threshold model:
- Baseline parameter: dQTc value when there is no effect.
- Threshold parameter: Concentration below which no QTc increase is expected.
- Slope parameter: indicates the rate of QTc increase when concentration is above the threshold value.

A population approach using the software package Monolix® [2] (version 3.2) was used to estimate the parameters. All graphical representations were performed in Matlab® [7] (version 2009b).

Compound A

No full PK and PD profiles were available due to sparse sampling. The relationship between observed Cmax and observed QTc max is depicted on Figure A1. The threshold model showed a better fit (AIC) compared to linear and Emax model.
The baseline parameter was different from 0 indicating a significant effect in the vehicle treated group.
The model turned out to be very appropriate to describe the data obtained from 2 other individuals (Figure A2).

Compound B

The threshold model was applied as described for compound A. The observed maximum effect on QTc was related to the model predicted Cmax.
One individual with QTc profile unrelated to dose increase and one with missing data were excluded from the analysis.

Compound C

The threshold model was applied as described for compound A on full PK and PD profiles (direct link).
Human data was also available allowing the translational capability of the threshold model to be evaluated. To account for the species differences and for the non linearity in protein binding, the QTc prolongation was represented as a function of free concentration in monkey and human.

3) RESULTS

A

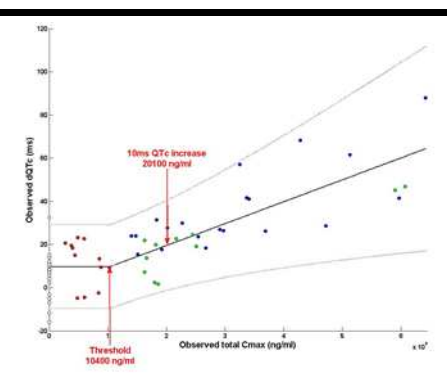
10ms QTc increase
20100 ng/ml
Threshold
10400 ng/ml
Observed dQTc (ms)
Observed total Cmax (ng/ml)

Figure A1: Observed dQTc at maximum effect versus observed total Cmax in dog with compound A.
Observed data (white, red, green and blue circle for control, low, mid and high dose respectively) and model predicted data (straight line is the mean prediction and dashed line the 95% confidence interval of the prediction).

Intra specie prediction performance

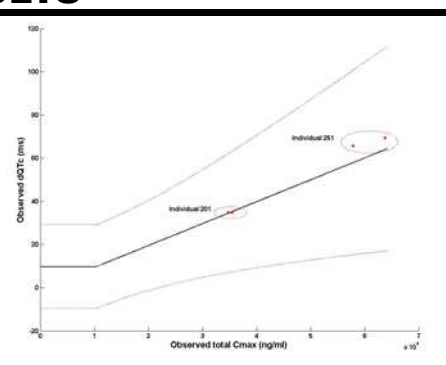
Observed dQTc (ms)
Observed total Cmax (ng/ml)

Figure A2: Observed dQTc at maximum effect versus observed total Cmax in dog with compound A in an additional study.
Observed data (red star) and model prediction (from figure A1).

- **Despite sparse data, the approach provides a good indication on the NOEL (Figure A1).**
- **Model prediction was a posteriori checked with other data from an additional study (Figure A2).**

B

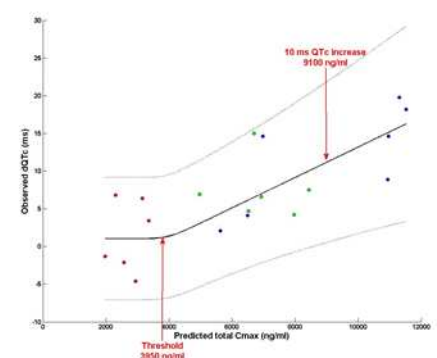
10 ms QTc increase
9100 ng/ml
Threshold
3850 ng/ml
Observed dQTc (ms)
Predicted total Cmax (ng/ml)

Figure B: Observed dQTc at maximum effect versus predicted total Cmax in monkey with compound B.
Observed data (red, green and blue circle for low, mid and high dose respectively) and model predicted data (straight line is the mean prediction and dashed line the 95% confidence interval of the prediction).

- **Limited PK and PD data: some limitation in precisely estimating the parameters.**
- **Nevertheless, it provides an indication on the NOEL.**

C

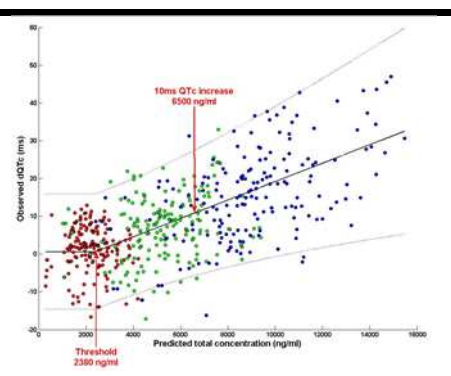
10ms QTc increase
6500 ng/ml
Threshold
2380 ng/ml
Observed dQTc (ms)
Predicted total concentration (ng/ml)

Figure C1: Observed dQTc versus predicted total concentration in monkey with compound C.
Observed data (red, green and blue circle for low, mid and high dose respectively) and model prediction (straight line is the mean prediction and dashed line the 95% confidence interval of the prediction).

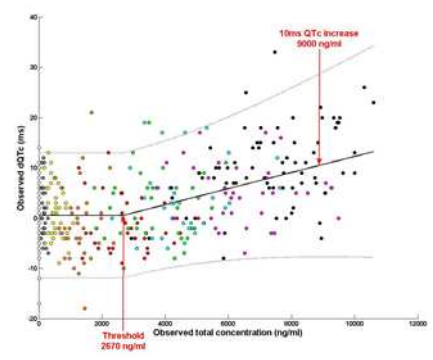
10ms QTc increase
9000 ng/ml
Threshold
2670 ng/ml
Observed dQTc (ms)
Observed total concentration (ng/ml)

Figure C2: Observed dQTc versus observed total concentration in human with compound C.
Observed data (circles with different colors corresponding to 8 dose levels + control) and model prediction (straight line is the mean prediction and dashed line the 95% confidence interval of the prediction).

Translational performance (Monkey to Human)

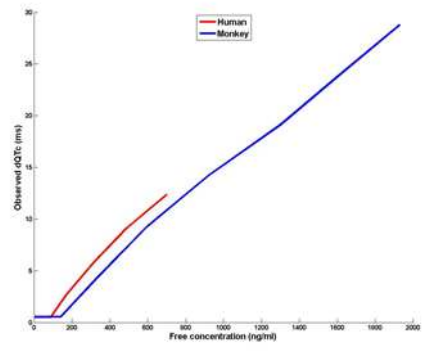
Human
Monkey
Observed dQTc (ms)
Free concentration (ng/ml)

Figure C3: Observed dQTc versus free concentration in monkey (blue line) and man (red line)

- **Threshold model successfully described the data in monkey and in human (Figure C1 and C2).**
- **'dQTc prediction versus total concentration' was transformed into 'dQTc prediction versus free concentration' (figure C3) accounting for non linear protein binding in each species:**
 - **Effect on QTc in Human similar to effect in monkey when it is a function of the free concentration.**
- **The threshold value was within a 2-fold range between species.**

[1] Henry H. Holzgrefe, Icilio Cavero, Carol R. Gleason, William A. Warner, Lewis V. Buchanan, Michael W. Gill, Dennis E. Burkett, Stephen K. Durham; Novel probabilistic method for precisely correcting the QT interval for heart rate in telemetered dogs and cynomolgus monkeys, Journal of Pharmacological and Toxiological Methods
[2] Kuhn E., Lavielle M. "Maximum likelihood estimation in nonlinear mixed effects models" Computational Statistics and Data Analysis, vol. 49, No. 4, pp 1020-1038, 2005
[3] GastroPlus v6.1, www.simulations-plus.com
[4] Kenny J. Watson, William P. Gorczyca, John Umland, Ying Zhang, Xian Chen, Sunny Z. Sun, Bernard Fermini, Mark Holbrook, Piet H. Van Der Graaf; Pharmacokinetic–pharmacodynamic modelling of the effect of Moxifloxacin on QTc prolongation in telemetered cynomolgus monkeys, Journal of Pharmacological and Toxicological methods 63 (2011) 304-313 and Poster presentation in PAGE 2011, Athens.
[5] Brandquist C., Stypinski D., Teuscher N., Girard F.; PK/PD Modeling of the Effect of Intravenous Doses of Anzemet® (dolasetron mesylate) and Its Metabolite (hydrodolasetron) on the QT Interval in Healthy Subjects; Poster presentation PAGE 2011, Athens.
[6] Guidance for Industry: E14 Clinical Evaluation of QT/QTc Interval Prolongation and Proarrhythmic Potential for Non-Antiarrhythmic Drugs, October 2005 ICH; http://www.fda.gov/downloads/Drugs/GuidanceComplianceRegulatoryInformation/Guidances/ucm073153.pdf
[7] MATLAB (2009) High-performance Numeric Computation and Visualization Software, R2009b, The Math Works, Natick MA.
[8] Sanguinetti MC, Jiang C, Curran ME, Keating MT. A mechanistic link between an inherited and an acquired cardiac arrhythmia: HERG encodes the IKr potassium channel. Cell 1995;81(2):299-307.

1