PK/PD modeling of brivaracetam in epilepsy using daily seizure counts

Rik Schoemaker[1], Armel Stockis[2]

[1]SGS Exprimo NV, Mechelen, Belgium
[2]UCB Pharma, Braine l'Alleud, Belgium

Objectives

To determine the population PK/PD relationship between brivaracetam (BRV) concentration and daily seizure counts in three Phase III studies in the adjunctive treatment of partial onset seizures, and to assess the influence of covariates on BRV effect.

Methodology

A population exposure-response model describing the effect of BRV on daily seizure counts was developed with the following components:

- daily seizure rates were described using a negative binomial statistical distribution with inter-individual variability in the over-dispersion factor
- the seizure rate on a given day was influenced by the number of seizures on the preceding day [1]
- a Box-Cox transformation was used to transform the baseline daily seizure rates improving the description of the baseline seizure rate distribution
- daily BRV average concentration (Cav) was used to drive the BRV effect, where an Emax model was used to describe the relationship between Cav and daily seizure rate
- a mixture model with two populations was applied separating the subjects into a mixture-model responder population and a mixture-model placebo-like population

Daily seizure count sequences with dependence on preceding-day seizures were simulated using NONMEM, allowing examination of the simulation properties of the model. A covariate analysis was performed to investigate factors influencing the effect of BRV on daily seizure counts.

Results

The population exposure-response model provided an excellent description of the data, where visual predictive checks (VPCs) for median % change in daily seizure frequency from baseline and fraction of subjects with more than 50% decrease in seizure frequency from baseline indicated that the model was perfectly capable of simulating the observed outcomes (see Figure 1).

Covariate analysis indicated that levetiracetam (LEV) co-administration effectively reduced the fraction of subjects in the mixture-model responder population to close to zero.

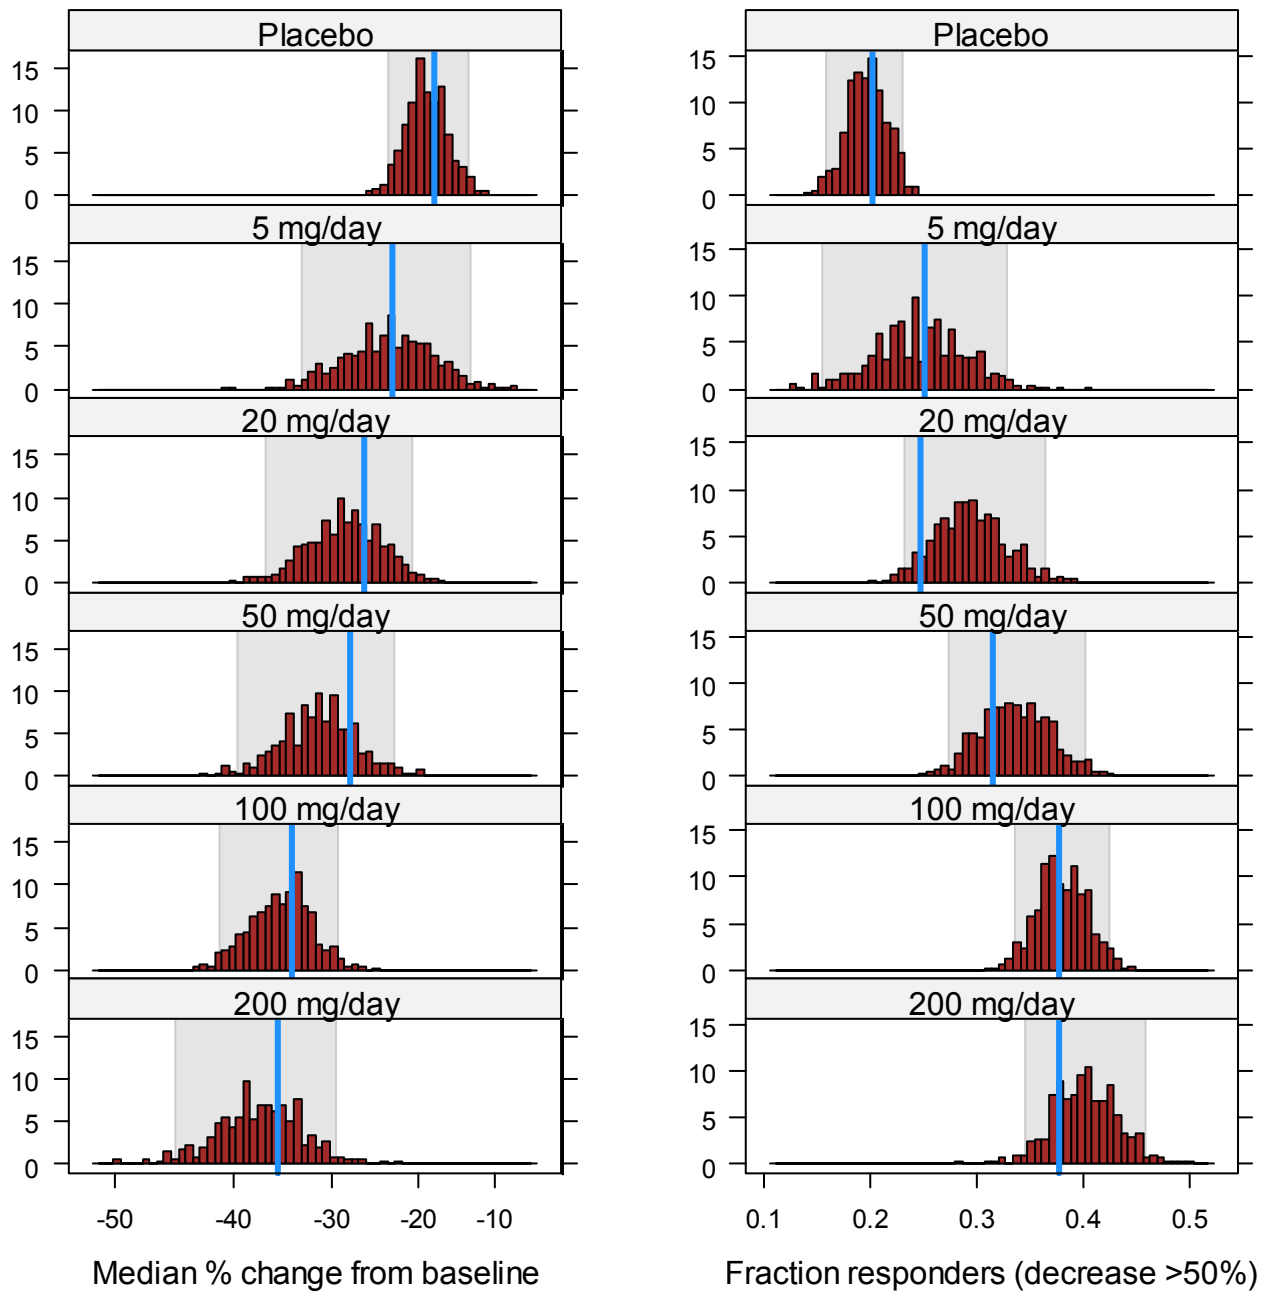
Placebo
5 mg/day
20 mg/day
50 mg/day
100 mg/day
200 mg/day
Median % change from baseline
Fraction responders (decrease >50%)

Figure 1 VPC for the final model by dose for median percent change from baseline (left) and fraction of 50% responders (right). *Histograms provide the distribution of outcomes for 500 simulated trials, the blue vertical line displays the result for the observed data.*

Covariate analysis also indicated that subjects with high baseline seizure rates had a much lower probability of ending up in the mixture-model responder population. In the absence of LEV co medication, the probabilities for ending up in the mixture-model responder population were estimated to be 54.8% at 0.14 seizures/day (1 seizures/week), 29.3% at 0.32 seizures/day (the median baseline seizure frequency), and 0.8% at 6 seizures/day. No further significant covariates influencing BRV effectiveness were detected.

The concentration at half the maximum response (EC_{50}) in the mixture-model responder population was estimated to be 0.57 mg/L, corresponding to 50 mg/day (see Figure 2). Further seizure reductions may be obtained for doses up to 150-200 mg/day.

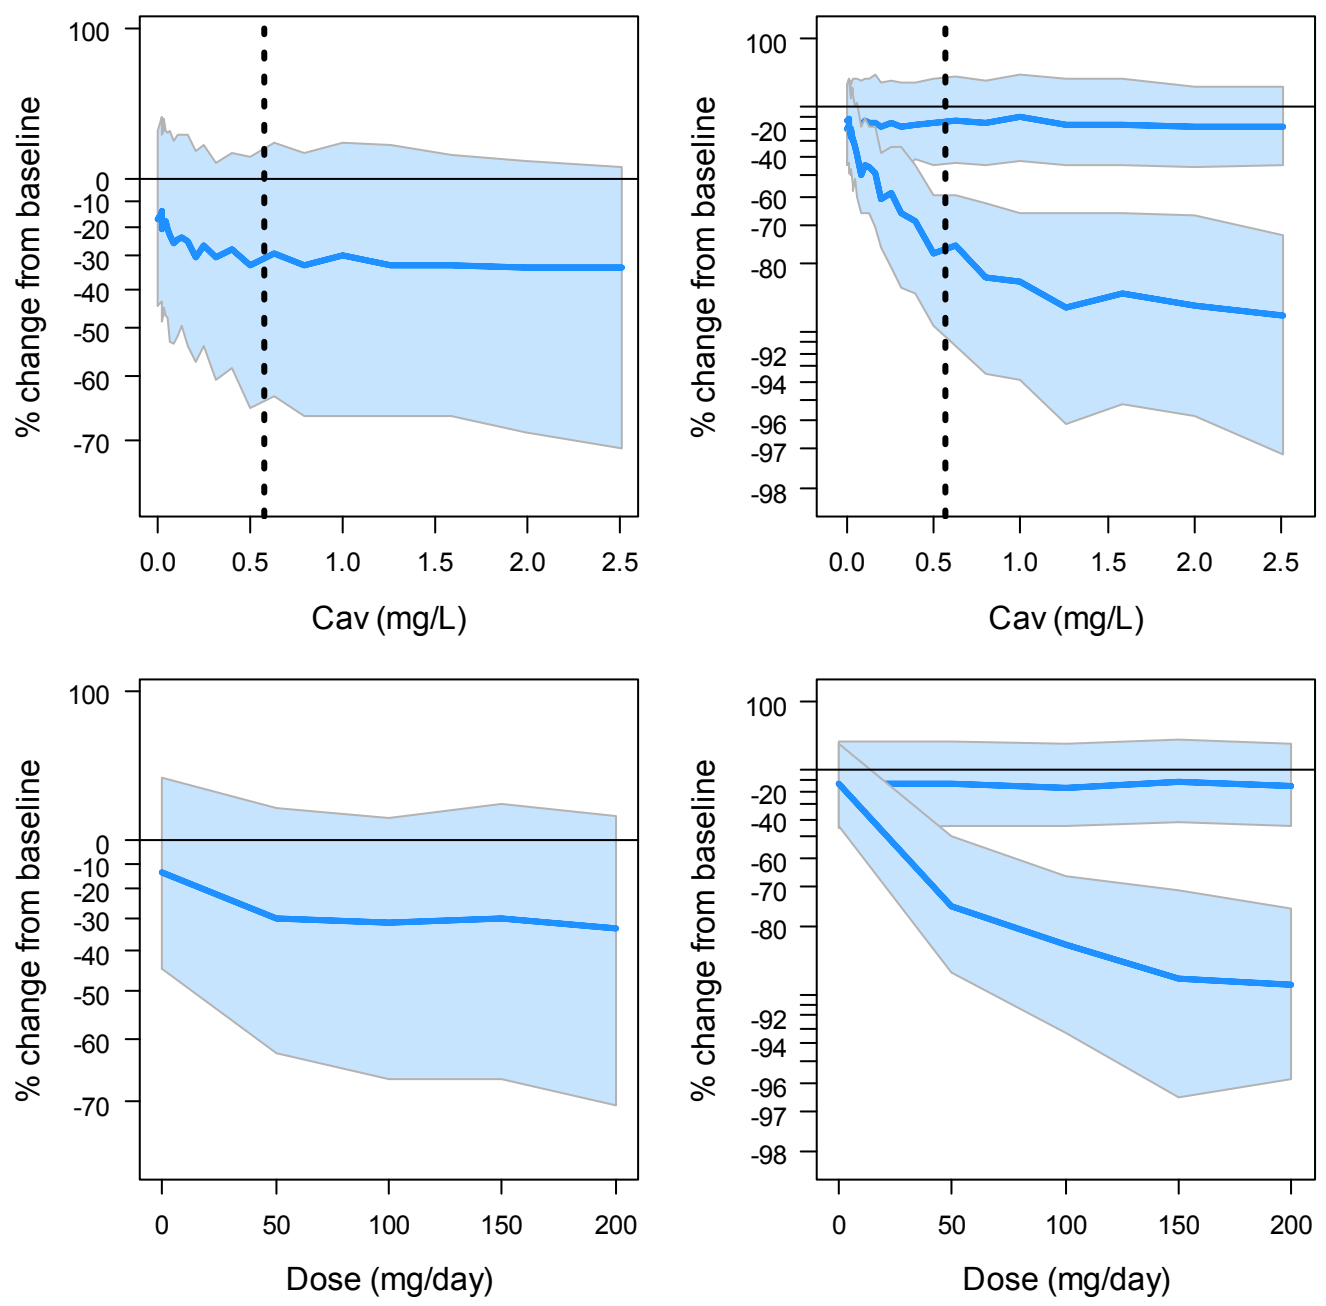
% change from baseline
Cav (mg/L)
Dose (mg/day)

Figure 2 Simulated overall BRV effect (left) and split by mixture model population (right), by BRV Cav (top) and daily dose (bottom). *Median and interquartile range of simulated individuals*

Conclusions

- **A population PK/PD model was developed allowing the mathematical description of the relationship between BRV exposure and its effect on daily seizure counts**
- **Only LEV co-administration and baseline seizure frequency were shown to significantly influence the response to BRV treatment**
- **The mixture model indicates that in responders the seizure frequency reduction reaches a plateau at 100 to 200mg/day.**

References

[1]. Ahn JE, Plan EL, Karlsson MO and Miller R. Modeling Longitudinal Daily Seizure Frequency Data From Pregabalin Add-On Treatment. J Clin Pharmacol 2012 52: 880-892.

This work was sponsored by UCB Pharma.