Use of mixture models in pharmacometric model-based analysis of confirmatory trials: part II – control of the type I error with real placebo data

UPPSALA UNIVERSITET

Estelle Chasseloup[1], Adrien Tessier[1], Mats O. Karlsson[1]
[1]Department of Pharmaceutical Biosciences, Uppsala University, Uppsala, Sweden - [2]Division of Clinical Pharmacokinetics and Pharmacometrics, Institut de Recherches Internationales Servier, Suresnes, France

Objectives

The control of the type I error during trials designed to establish drug efficacy is critical to avoid unnecessary and unethical studies. To analyse such studies, **the standard (STD) non-linear mixed-effect models (NLMEM) have good power properties[1,2], but unsatisfactory type I error control because of model misspecifications and bias in drug effect estimates**. We used real placebo data to explore a **new NLMEM approach using mixture models, called individual model averaging (IMA)[3], comparing its type I error rate sensitivity to model misspecification and its bias in drug effect estimates to the STD NLMEM approach.**

Methods

Real placebo data

- **Likert pain score** (categorical data) from 230 patients with neuropathic pain receiving placebo[4,5]
- **ADAS-Cog** (continuous data): severity test from the natural history of 800 patients with Alzheimer's disease[6]

Patients were randomized (1:1) to artificial placebo or drug treatment (TRT). The **type I error rate** is the frequency with which the full model (TRT as covariate, H_1) is significant (α=0.05) relative to the base model (no TRT information, H_0) according to the likelihood ratio test (LRT).

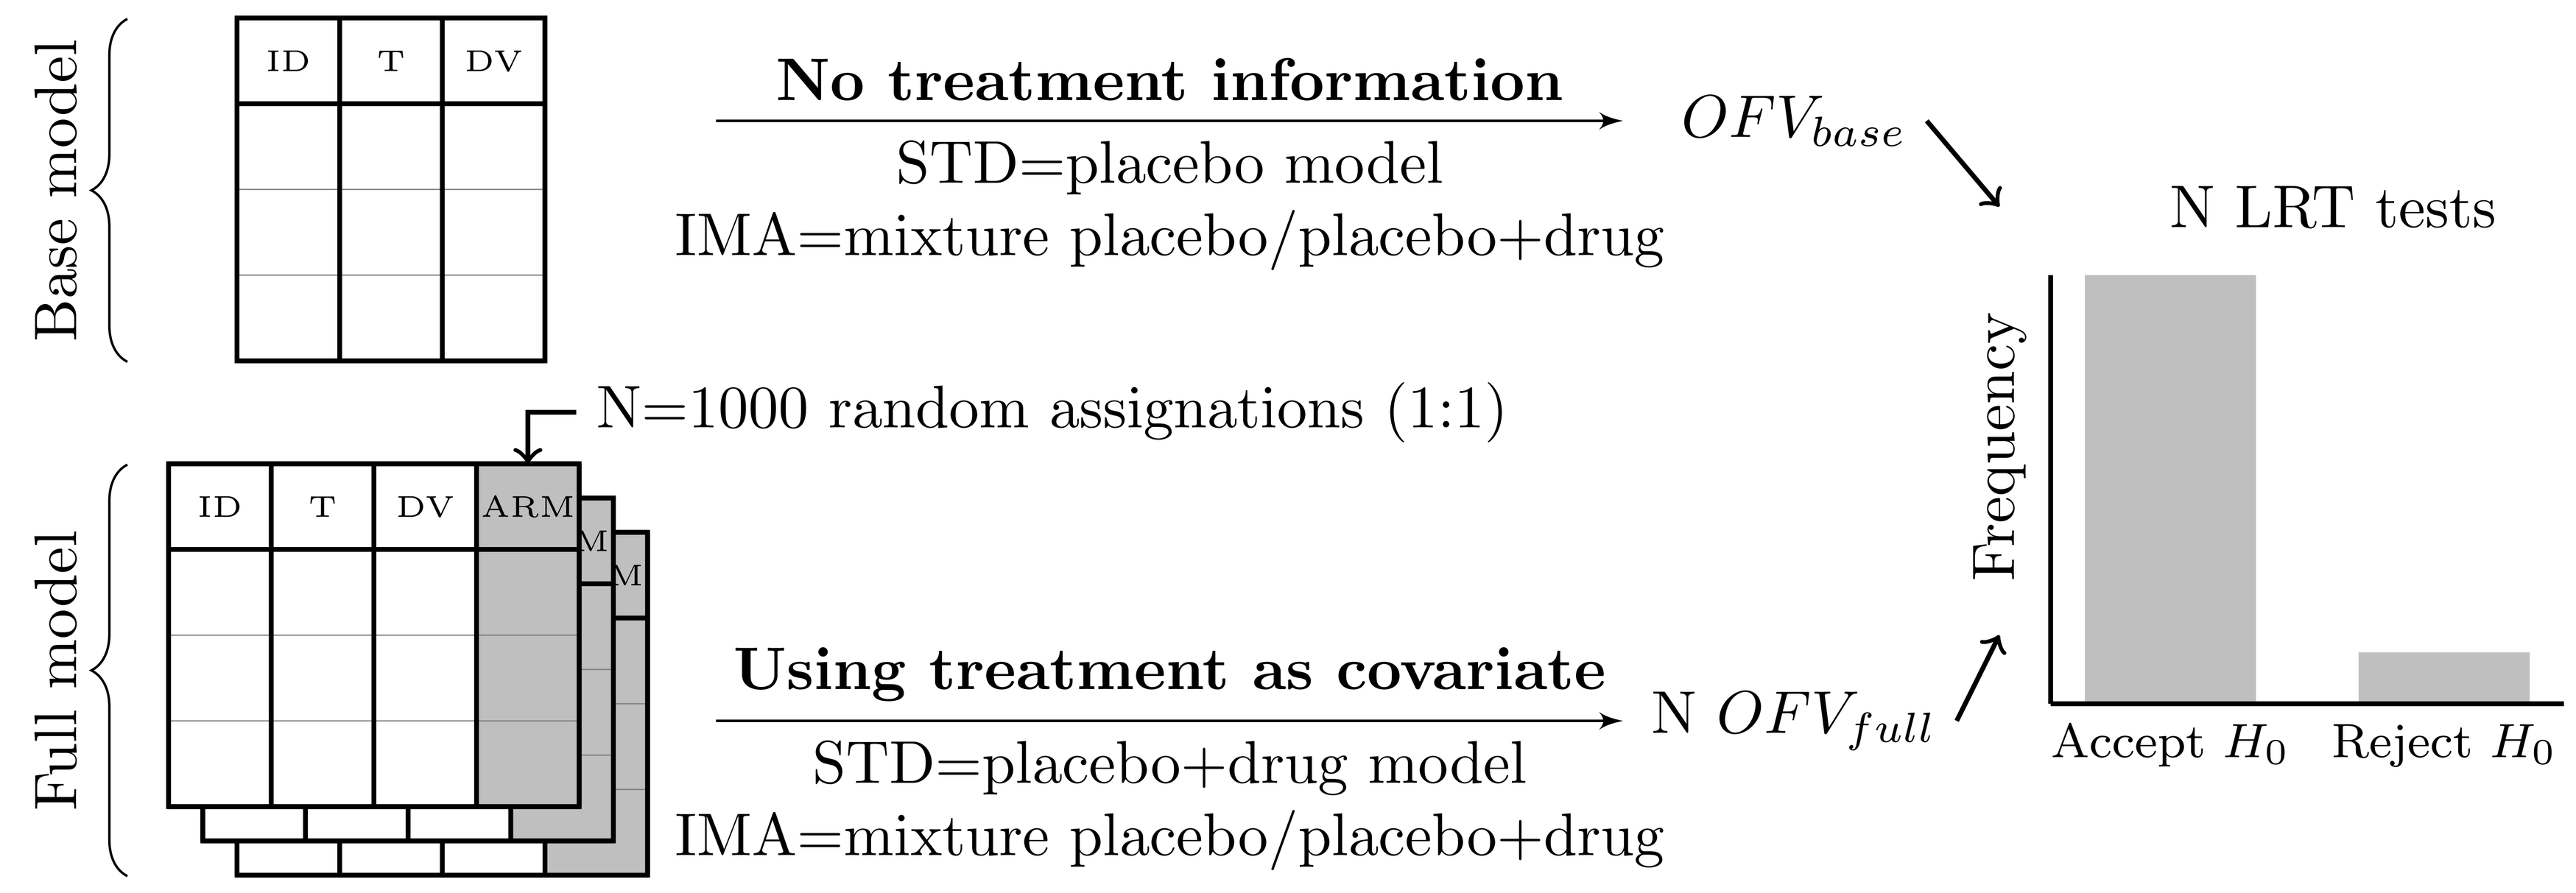
Base model
ID T DV
No treatment information
STD=placebo model
IMA=mixture placebo/placebo+drug
OFV_{base}
N=1000 random assignations (1:1)
Full model
ID T DV ARM
Using treatment as covariate
STD=placebo+drug model
IMA=mixture placebo/placebo+drug
N OFV_{full}
N LRT tests
Frequency
Accept H_0
Reject H_0

Figure 1: Workflow used to compute the type I error

Table 1: Description of the base and full models for each approach

	Base model	Full model
STD	$BASE + PLB$	$BASE + PLB + DRUG * TRT$
IMA	Mixture 1: $BASE + PLB$ and mixture 2: $BASE + PLB + DRUG$	
	Mixture probability set to randomization: P(1) = 0.5 P(2) = 1 - P(1)	Mixture probability estimated: P(1) = TRT*θ_{MIX}+(1-TRT)*(1-θ_{MIX}) P(2) = 1 - P(1)

$BASE$: baseline model, PLB: placebo model, $DRUG$: drug effect model, θ_{MIX}: mixture probability, TRT: treatment allocation (0 for placebo and 1 for treatment).

Table 2: Computation of the median drug effect

Drug model	Drug effect parameterization	Median drug effect
Offset	$BASE + PLB + \theta_{drug}$	$(\theta_{MIX} * 2 - 1) * \theta_{drug}$
Linear with time *Likert pain score data*	$BASE + PLB + \theta_{drug} * t$	$(\theta_{MIX} * 2 - 1) * \theta_{drug} * t_{last}$
Linear with placebo *ADAS-cog data*	$BASE + PLB + \theta_{drug} * PLB$	$(\theta_{MIX} * 2 - 1) * \theta_{drug} * PLB$

$BASE$: baseline model, PLB: placebo model, θ_{drug}: drug effect parameter, θ_{MIX}: mixture probability (set to 1 for the standard approach), t_{last}: time at the end of the study.

Type I error results

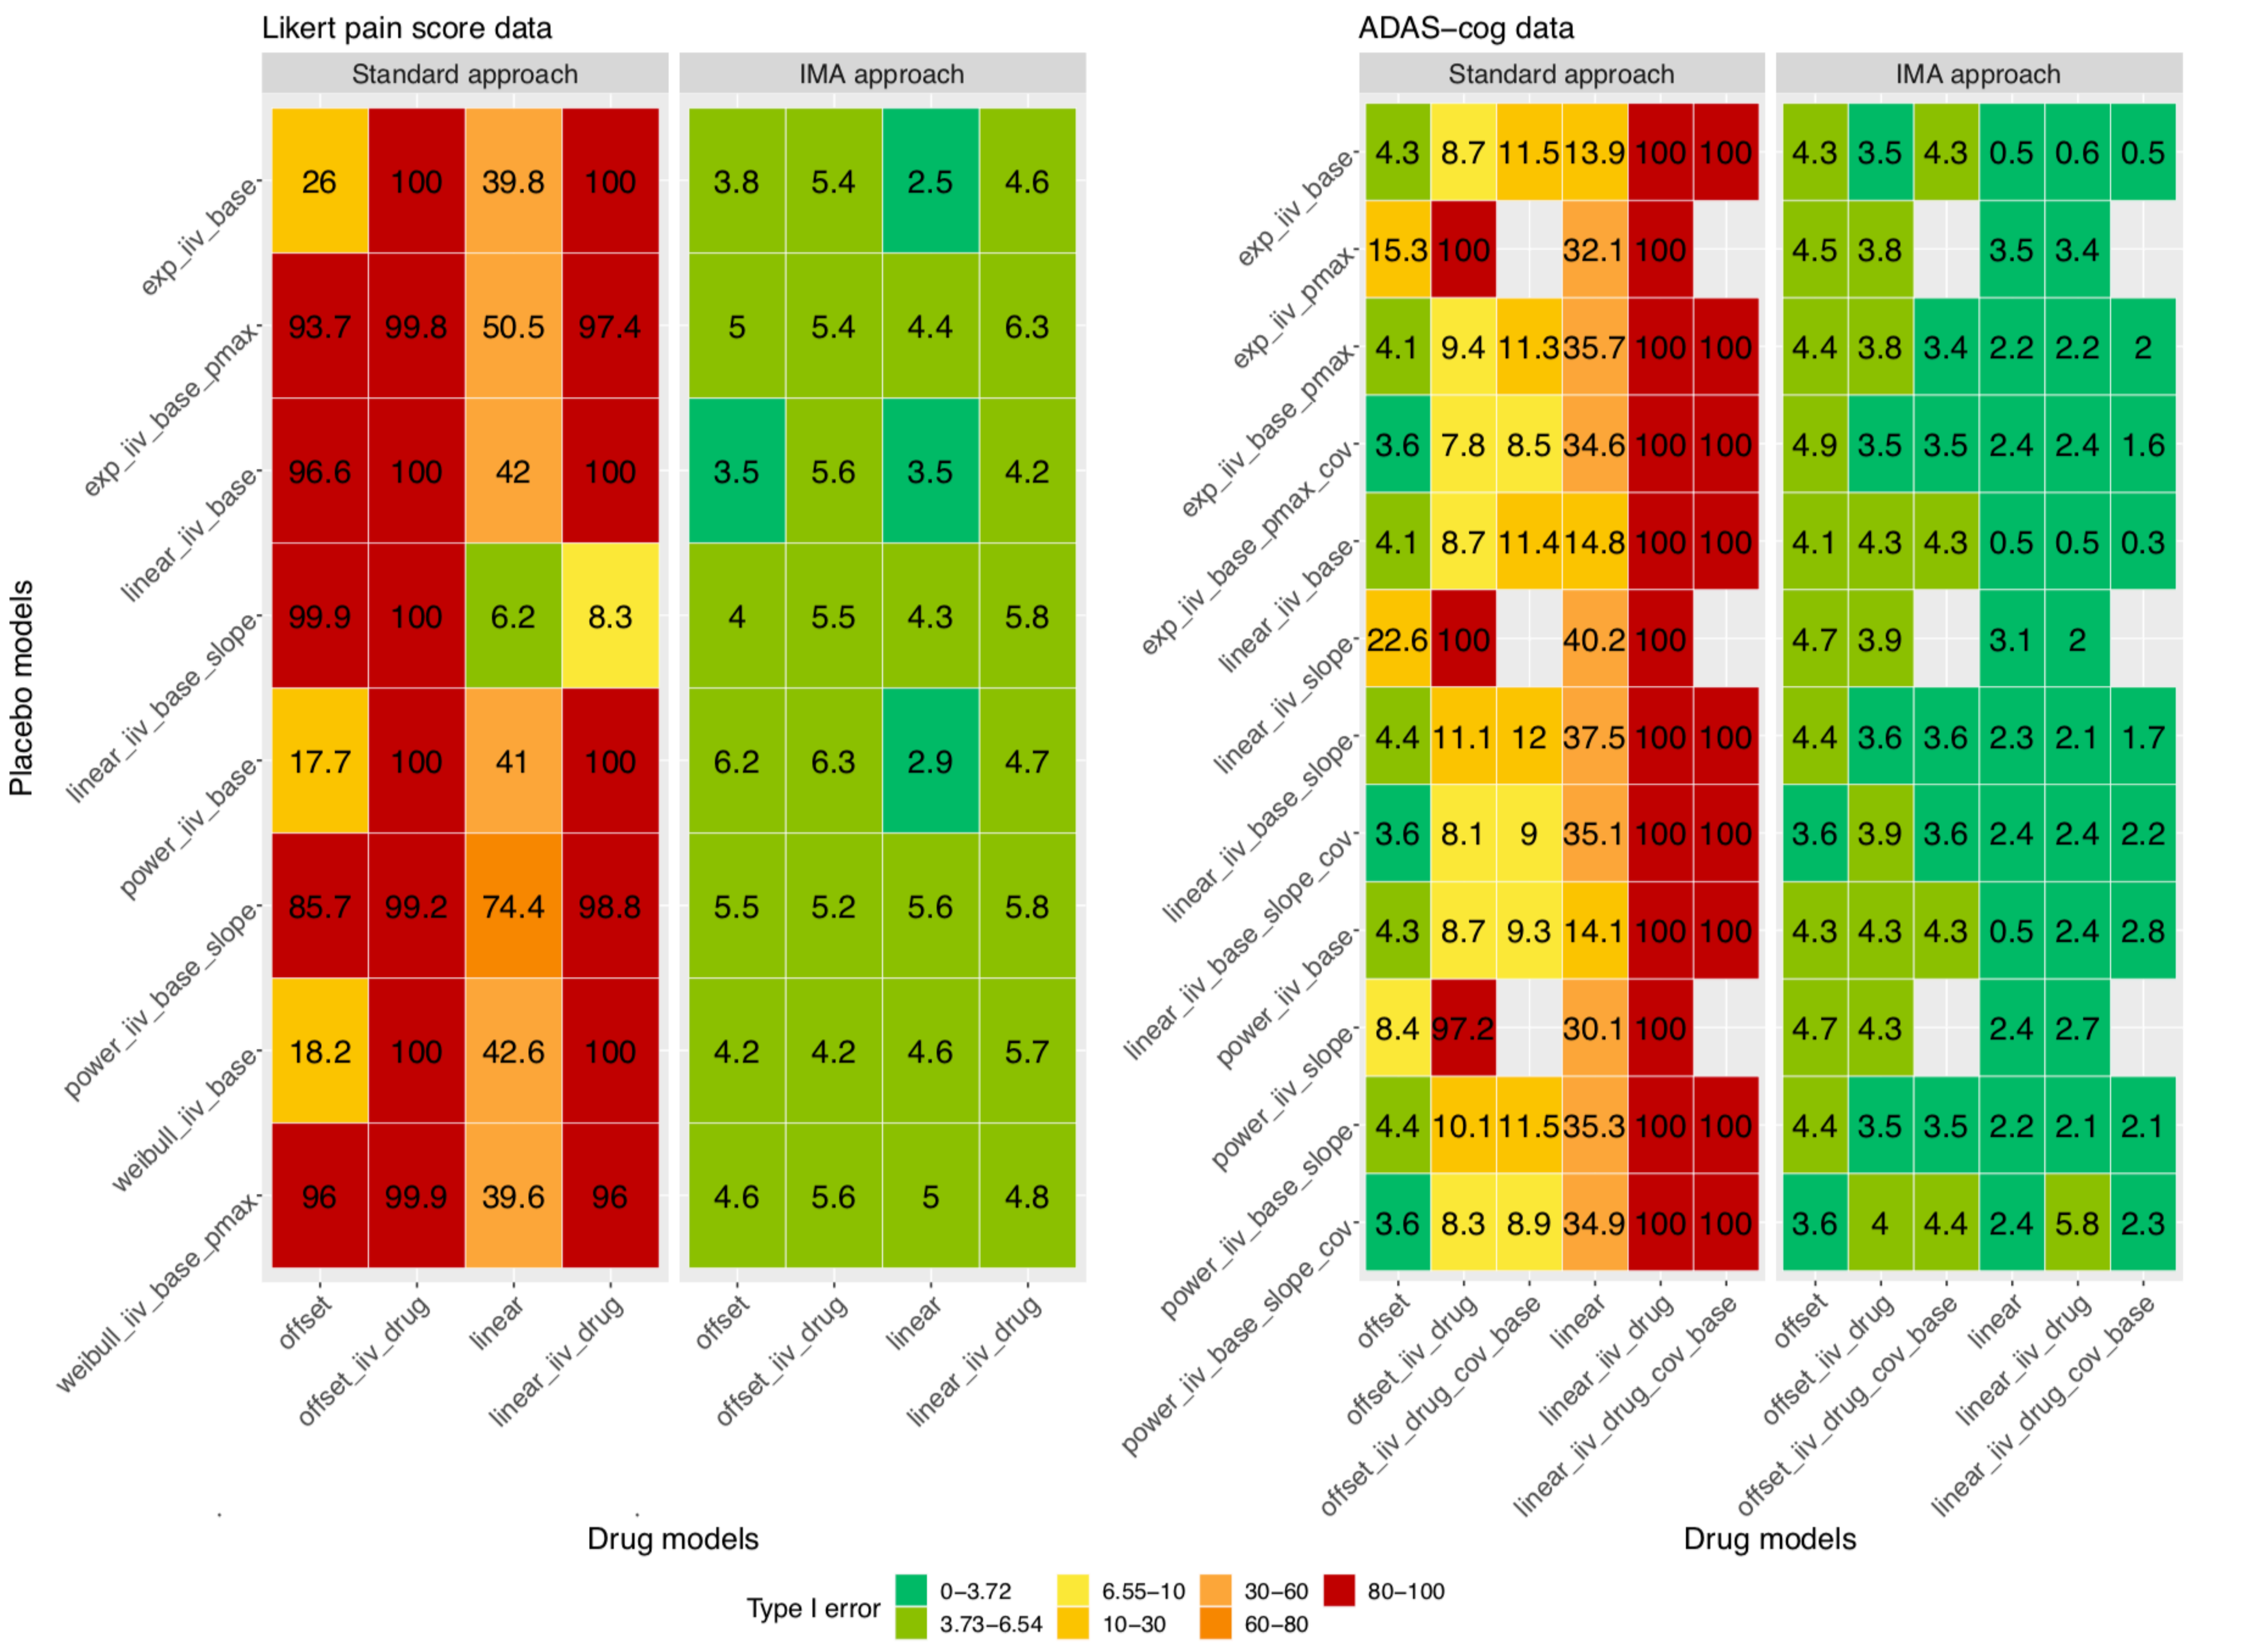
Likert pain score data
Standard approach
IMA approach
exp_iiv_base 26 100 39.8 100 3.8 5.4 2.5 4.6
exp_iiv_base_pmax 93.7 99.8 50.5 97.4 5 5.4 4.4 6.3
linear_iiv_base 96.6 100 42 100 3.5 5.6 3.5 4.2
linear_iiv_base_slope 99.9 100 6.2 8.3 4 5.5 4.3 5.8
power_iiv_base 17.7 100 41 100 6.2 6.3 2.9 4.7
power_iiv_base_slope 85.7 99.2 74.4 98.8 5.5 5.2 5.6 5.8
weibull_iiv_base 18.2 100 42.6 100 4.2 4.2 4.6 5.7
weibull_iiv_base_pmax 96 99.9 39.6 96 4.6 5.6 5 4.8
offset offset_iiv_drug linear linear_iiv_drug
Placebo models
Drug models
ADAS–cog data
Standard approach
IMA approach
exp_iiv_base 4.3 8.7 11.5 13.9 100 100 4.3 3.5 4.3 0.5 0.6 0.5
exp_iiv_pmax 15.3 100 32.1 100 4.5 3.8 3.5 3.4
exp_iiv_base_pmax 4.1 9.4 11.3 35.7 100 100 4.4 3.8 3.4 2.2 2.2 2
exp_iiv_base_pmax_cov 3.6 7.8 8.5 34.6 100 100 4.9 3.5 3.5 2.4 2.4 1.6
linear_iiv_base 4.1 8.7 11.4 14.8 100 100 4.1 4.3 4.3 0.5 0.5 0.3
linear_iiv_slope 22.6 100 40.2 100 4.7 3.9 3.1 2
linear_iiv_base_slope 4.4 11.1 12 37.5 100 100 4.4 3.6 3.6 2.3 2.1 1.7
linear_iiv_base_slope_cov 3.6 8.1 9 35.1 100 100 3.6 3.9 3.6 2.4 2.4 2.2
power_iiv_base 4.3 8.7 9.3 14.1 100 100 4.3 4.3 4.3 0.5 2.4 2.8
power_iiv_slope 8.4 97.2 30.1 100 4.7 4.3 2.4 2.7
power_iiv_base_slope 4.4 10.1 11.5 35.3 100 100 4.4 3.5 3.5 2.2 2.1 2.1
power_iiv_base_slope_cov 3.6 8.3 8.9 34.9 100 100 3.6 4 4.4 2.4 5.8 2.3
offset offset_iiv_drug offset_iiv_drug_cov_base linear linear_iiv_drug linear_iiv_drug_cov_base
Drug models
Type I error
0–3.72
3.73–6.54
6.55–10
10–30
30–60
60–80
80–100

Figure 2: Type I error rate comparison between the two approaches with categorical data (left panel) and continuous data (right panel)

Standard approach

The type I error is very sensitive to model misspecifications: low only when the drug model offer little opportunity to improve the fit in the presence of placebo model misspecification.

IMA approach

The type I error is controlled for both categorical and continuous data (α =0.05), and often below α, especially with the ADAS-cog data.

Drug effect bias results

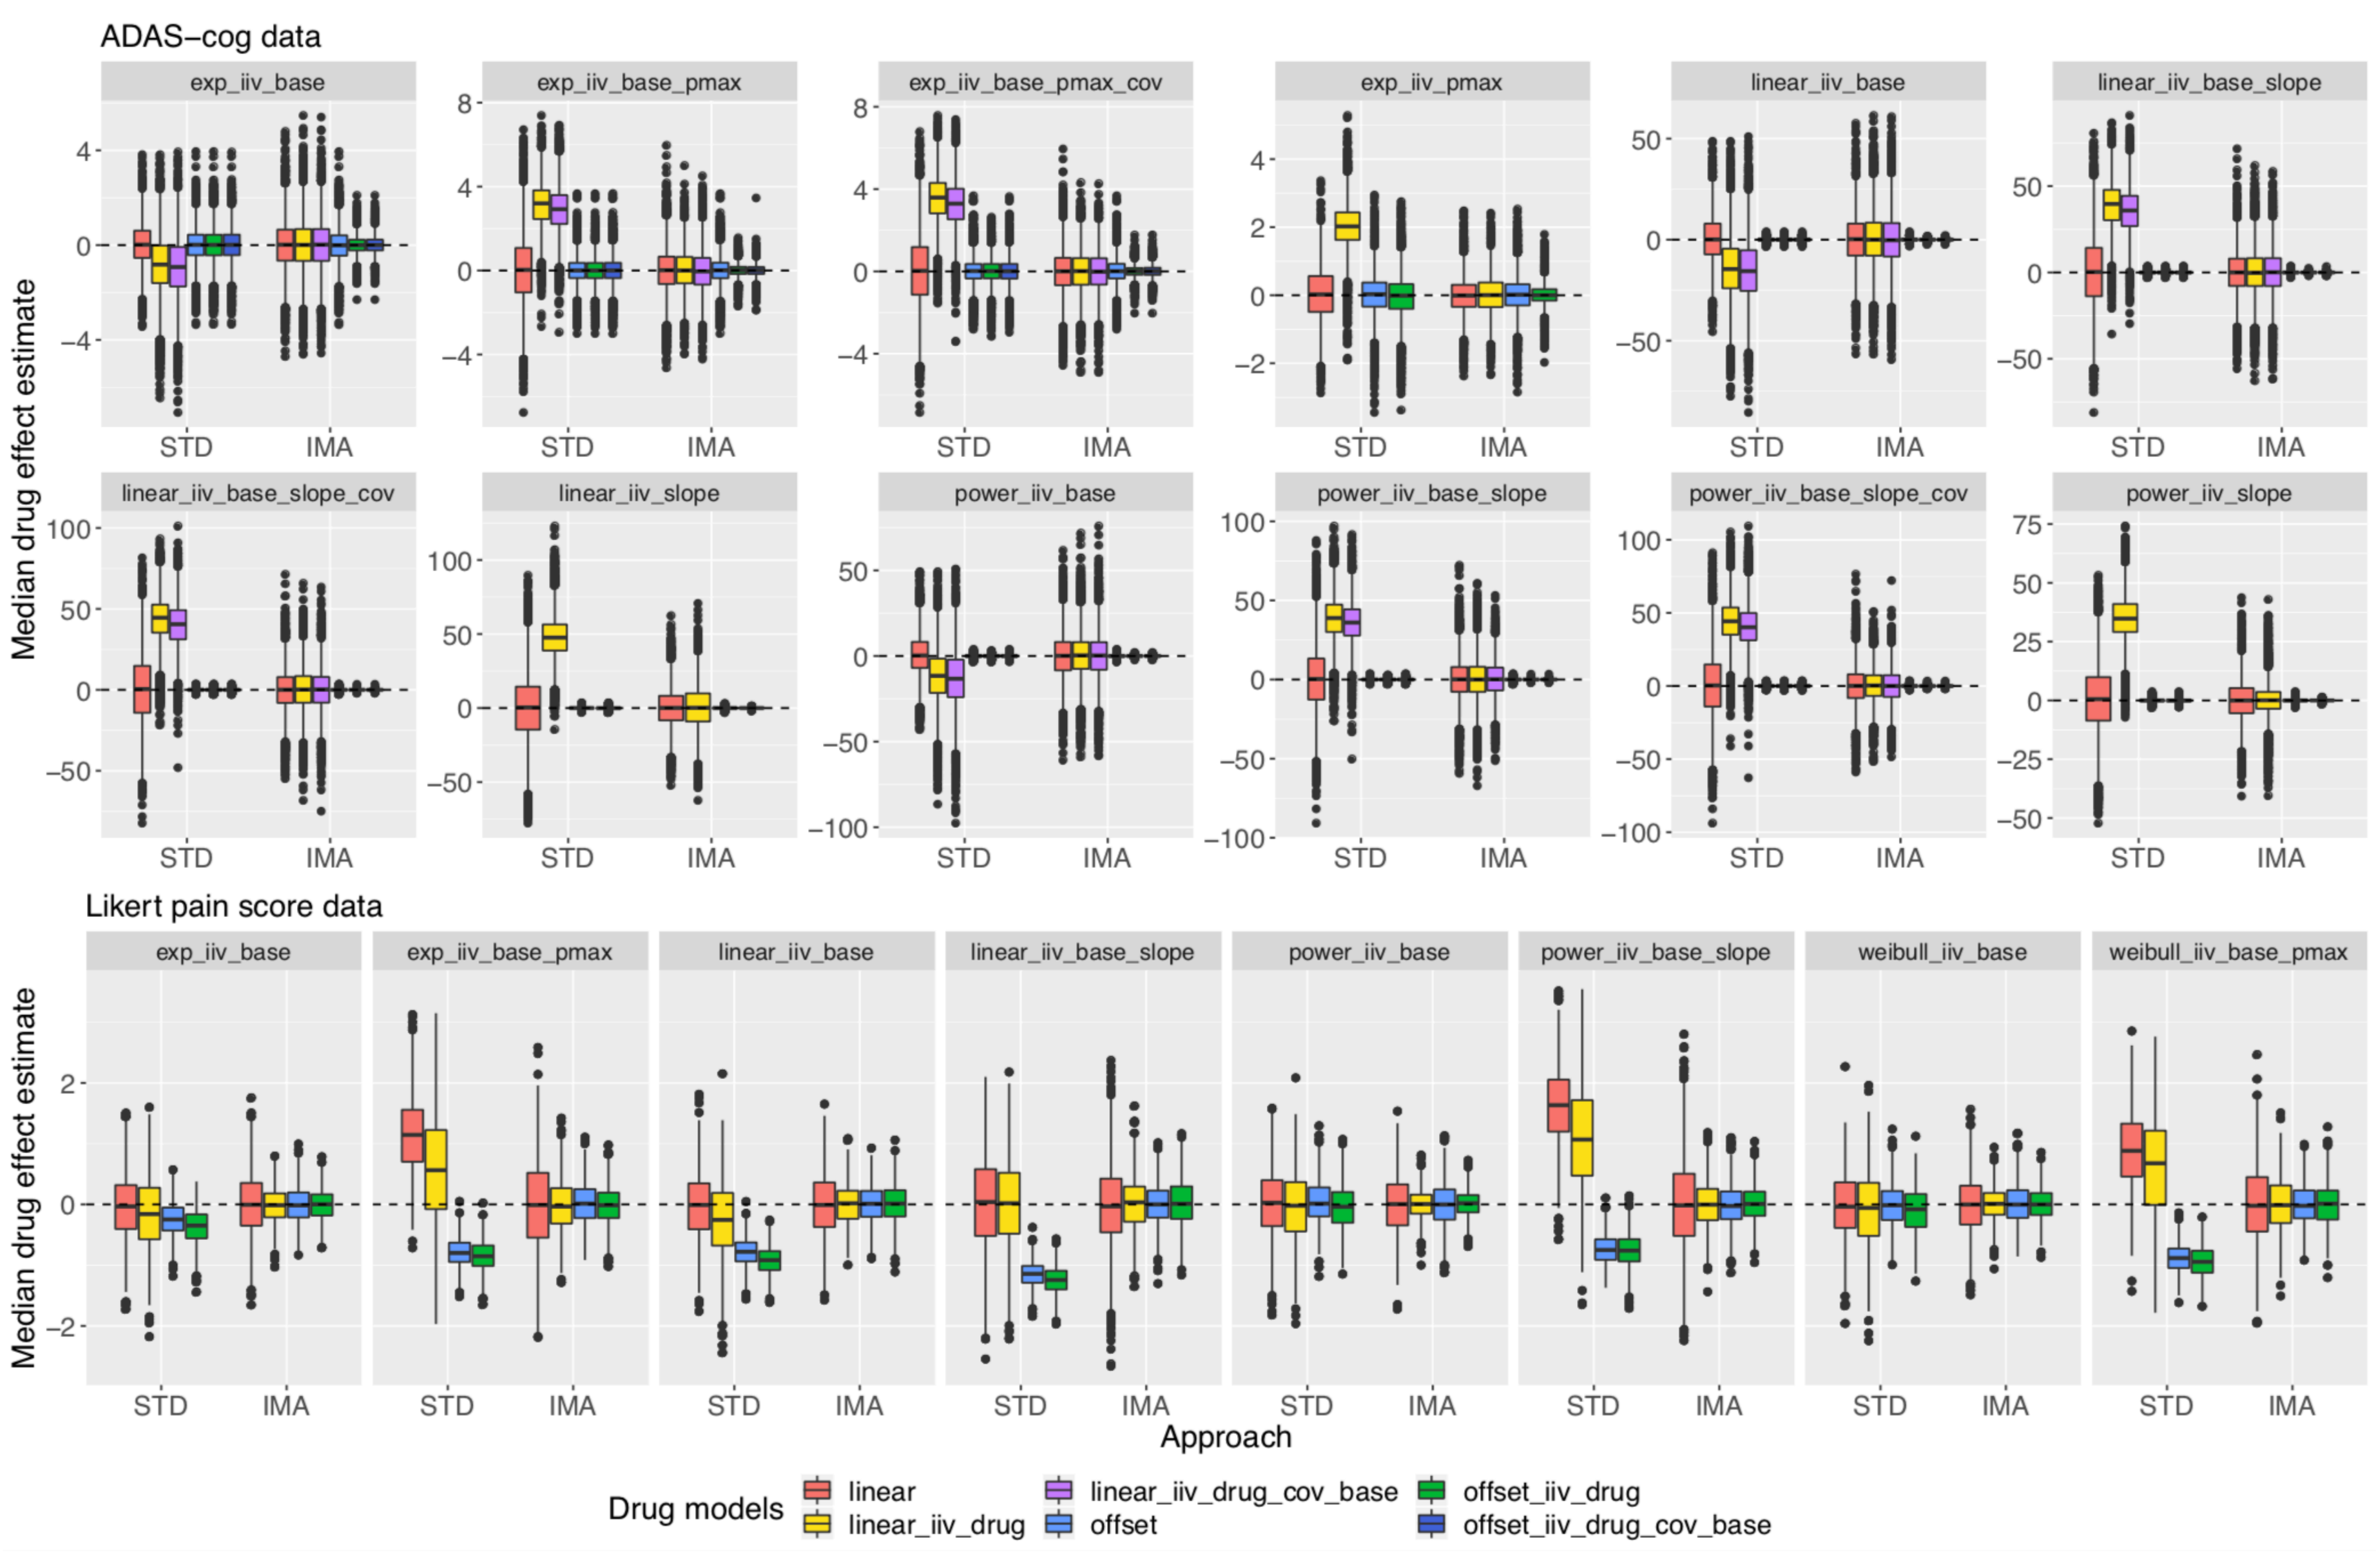
ADAS–cog data
exp_iiv_base
exp_iiv_base_pmax
exp_iiv_base_pmax_cov
exp_iiv_pmax
linear_iiv_base
linear_iiv_base_slope
linear_iiv_base_slope_cov
linear_iiv_slope
power_iiv_base
power_iiv_base_slope
power_iiv_base_slope_cov
power_iiv_slope
Median drug effect estimate
STD
IMA
Likert pain score data
exp_iiv_base
exp_iiv_base_pmax
linear_iiv_base
linear_iiv_base_slope
power_iiv_base
power_iiv_base_slope
weibull_iiv_base
weibull_iiv_base_pmax
Approach
Drug models
linear
linear_iiv_drug
linear_iiv_drug_cov_base
offset
offset_iiv_drug
offset_iiv_drug_cov_base

Figure 3: Bias in drug effect estimate facetted by placebo models for categorical data (bottom panel) and continuous data (top panel)

Standard approach

A pronounced bias in the median drug effect occurs for many drug/placebo model combinations. It is typically larger when the drug effect model contains inter-individual variability (iiv).

IMA approach

None of the placebo/drug combination tested had biased median drug estimate and the precision is slightly better than the STD approach.

Conclusions

- IMA has a better type I error control and is more robust towards misspecifications in the placebo model.
- IMA consistently provided unbiased estimates of drug effect, whereas the standard approach did not.
- IMA seems a promising method to address the drawbacks from the standard NLMEM approach to analyse efficacy trials

[1]Karlsson KE, Vong C, Bergstrand M, Jonsson EN, Karlsson MO. Comparisons of Analysis Methods for Proof-of-Concept Trials. CPT Pharmacomet Syst Pharmacol. 2013 Jan;2(1):e23. [2]Jonsson EN, Sheiner LB. More efficient clinical trials through use of scientific model-based statistical tests. Clin Pharmacol Ther. 2002 Dec;72(6):603–14. [4]Plan EL, Elshoff J-P, Stockis A, Sargentini-Maier ML, Karlsson MO. Likert pain score modeling: a Markov integer model and an autoregressive continuous model. Clin Pharmacol Ther. 2012 May;91(5):820–8. [5]Schindler E, Karlsson MO. A Minimal Continuous-Time Markov Pharmacometric Model. AAPS J. 2017;19(5):1424–35. [6]Ito K, Corrigan B, Zhao Q, French J, Miller R, Soares H, Katz E, Nicholas T, Billing B, Anziano R, Fullerton T, Alzheimer's Disease Neuroimaging Initiative. Disease progression model for cognitive deterioration from Alzheimer's Disease Neuroimaging Initiative database. Alzheimers Dement J Alzheimers Assoc. 2011 Mar;7(2):151–60.

[3]More results (including power) with simulated data, in A. Tessier et al. Poster IV-75 (abstract 9233)