Population PK-QT Analysis across Phase I Studies for a p38 Mitogen Activated Protein Kinase Inhibitor – PH-797804

Rujia Xie[1], Jakob Ribbing[2], Peter A Milligan[3], Lutz Harnisch[3], Grant Langdon[4]
[1]Clinical Pharmacology & Pharmacometrics, Pfizer (China) R&D, China, [2]Pharmacometrics, Pfizer, Sweden,
[3]Pharmacometrics, Pfizer, UK, [4]PTx Solution Ltd. UK

Objectives:

Development of a PK-QTc model to 1) characterize the time-course of QTc interval prolongation following administration of PH-797804, and 2) inform dose selection for a future thorough QT (TQT) study.

Methods:

PK-QTc model 6 Phase I studies were analyzed (129 subjects receiving single or multiple oral doses over range 0.3-60 mg or placebo, total of 3980 QT records). Typically, 3 replicate 12-lead ECG were recorded at each time point. PK-QT analysis was conducted in four steps:

1)Nonparametric PK model to predict concentrations at time of QT measurements
2)PK-RR interval analysis
3)Assessment of correction methods using baseline QT data (eq.1)
4)PK-QTc analysis (eq. 2-4)

Different drug effect models on QTc, Linear, ln-linear, step ln-linear (eq. 2-3), step function, E_{max} and sigmoidal E_{max} models were evaluated:

$$\ln\left(\hat{QT}_{ij}\right) = \ln(QTCM_i) + CIRC_i + BETA_i * \ln\left(\frac{RR_{ij}}{1000}\right) \qquad \text{eq. 1}$$

$$step = \frac{CONC^{10}}{\left(CONC^{10} + CT^{10}\right)} \qquad \text{eq. 2}$$

$$DRUG = step * slope * LN(CONC) \qquad \text{eq. 3}$$

$$\hat{QTc}_{ij} = QTCM_i * (1 + CICR_i) + DRUG_{ij} \qquad \text{eq. 4}$$

$$QTc_{ij} = \hat{QTc}_{ij} * \left(1 + \varepsilon_{ij} + \varepsilon_{ik}\right) \qquad \varepsilon \in \left(0, \sigma^2\right) \qquad \text{eq. 5}$$

Where QT_{ij} and RR_{ij} are the observed QTc interval and heart rate for subject i at time point j, respectively. *QTcm* represents the 24 hour mean corrected QT interval at baseline. $BETA_i$ is individual correction. $CIRC_i$ denotes the circadian rhythm, which is the cosine function with wavelength of 24. k indexes replicate. ε_{ij} and ε_{ik} are residual errors between and within replicate, respectively.

NONMEM was used to analyze PK, RR, and QT data. Residual correlation among the QT-Triplicate measures was accounted for by using the L2 option (eq. 5).

TQT trial simulation setting Fixed effect population parameter estimates, with uncertainty, and random effects (IIV and residuals) from the final QTcIS model were used to predict the following design: cross-over in fasted healthy volunteers; sample size N=60; dose range: 0-24mg; 14 days washout period among each dose; triplicate measurements (2 min apart); sampling time: 0, 2, 3, 4, 5, 6, 8, 12, and 24hr; 500 simulation replicates.

Results:

PH-797804 concentrations were well characterized by a linear interpolation method. In the RR interval model, a negative slope was estimated but a bootstrap 95%CI resulted in included zero, -0.436 [95%CI: -0.899, 0.208]. Six correction methods were evaluated: QTcB, QTcF, QTcP (population), QTcS (study), QTcI (individual) and QTcIS (individual correction and an extra study correction factor).

The relationship between QTc and RR interval during baseline and treatment periods is shown in Figure 1. QTcIS was selected as the DV in developing the PK-QTc model. The final PK-QTcIS model is described by eq.4 and consists of one cosine circadian term; a step ln-linear concentration effect on QTcIS; and an effect of gender on baseline. The slope was estimated as 1.44 [95%CI: 0.93, 1.91] (ms change when concentrations increase 2.72 times) and a "concentration step" estimated as 1.27 [95%CI: 0.22, 4.36] ng/ml. Baseline QTcIS in females was 7.36 ms longer than in males (404ms). A large replicate variability (ε correlation 0.32) was determined.

Figure 1. RR-QTc Interval Relationship for Baseline (grey) and Treatment Periods (black) for all studies

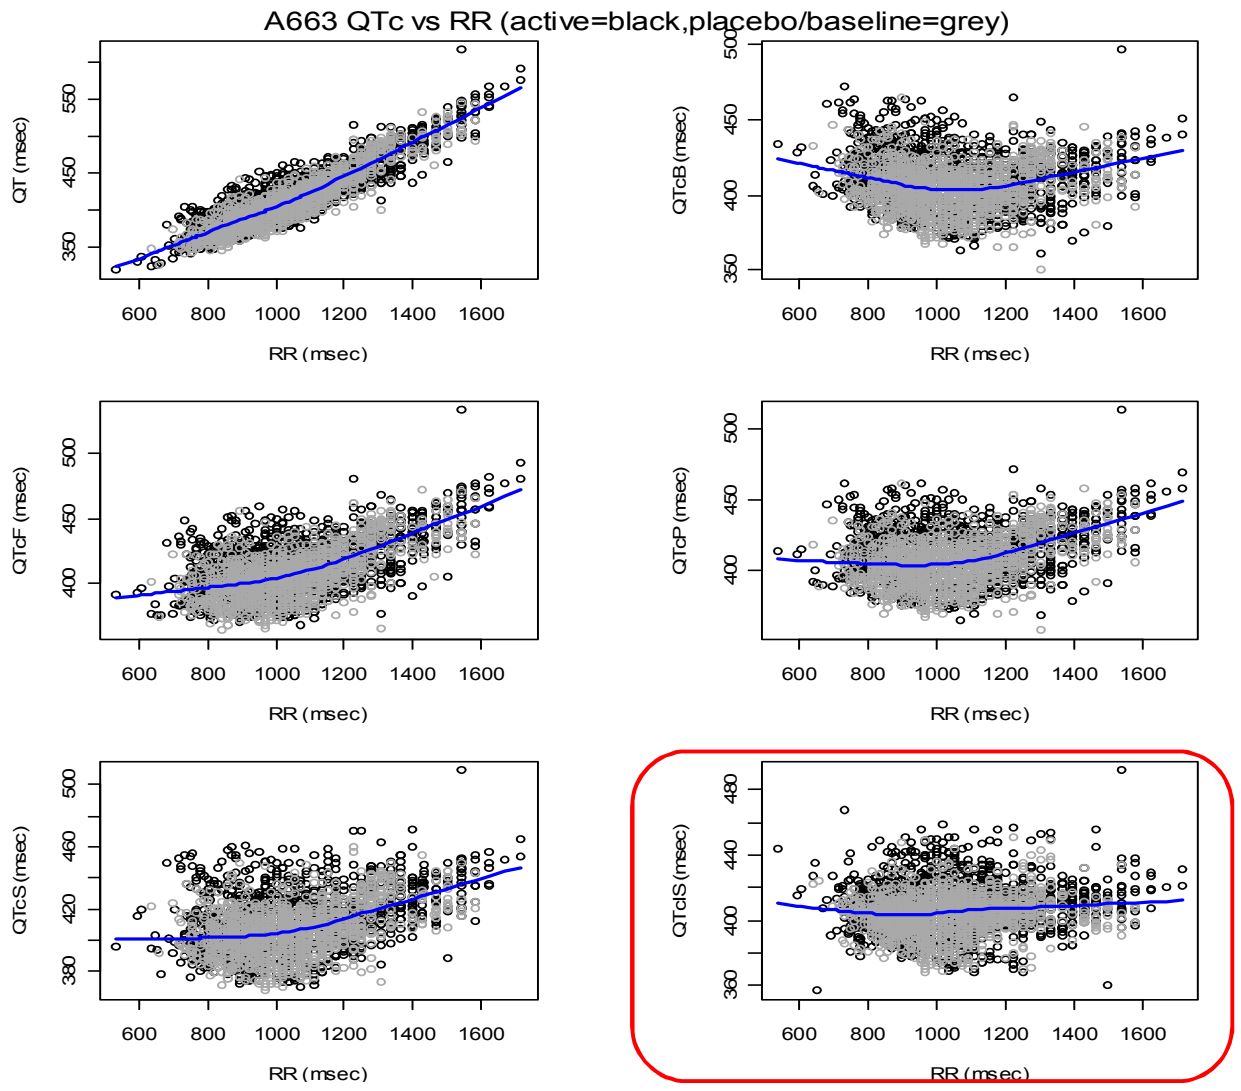
A663 QTc vs RR (active=black,placebo/baseline=grey)
QT (msec)
QTcB (msec)
QTcF (msec)
QTcP (msec)
QTcS (msec)
QTcIS (msec)
RR (msec)
600
800
1000
1200
1400
1600

A prediction corrected visual predictive check (PC-VPC) was performed (Figure 2).

Figure 2. PC-VPC by study

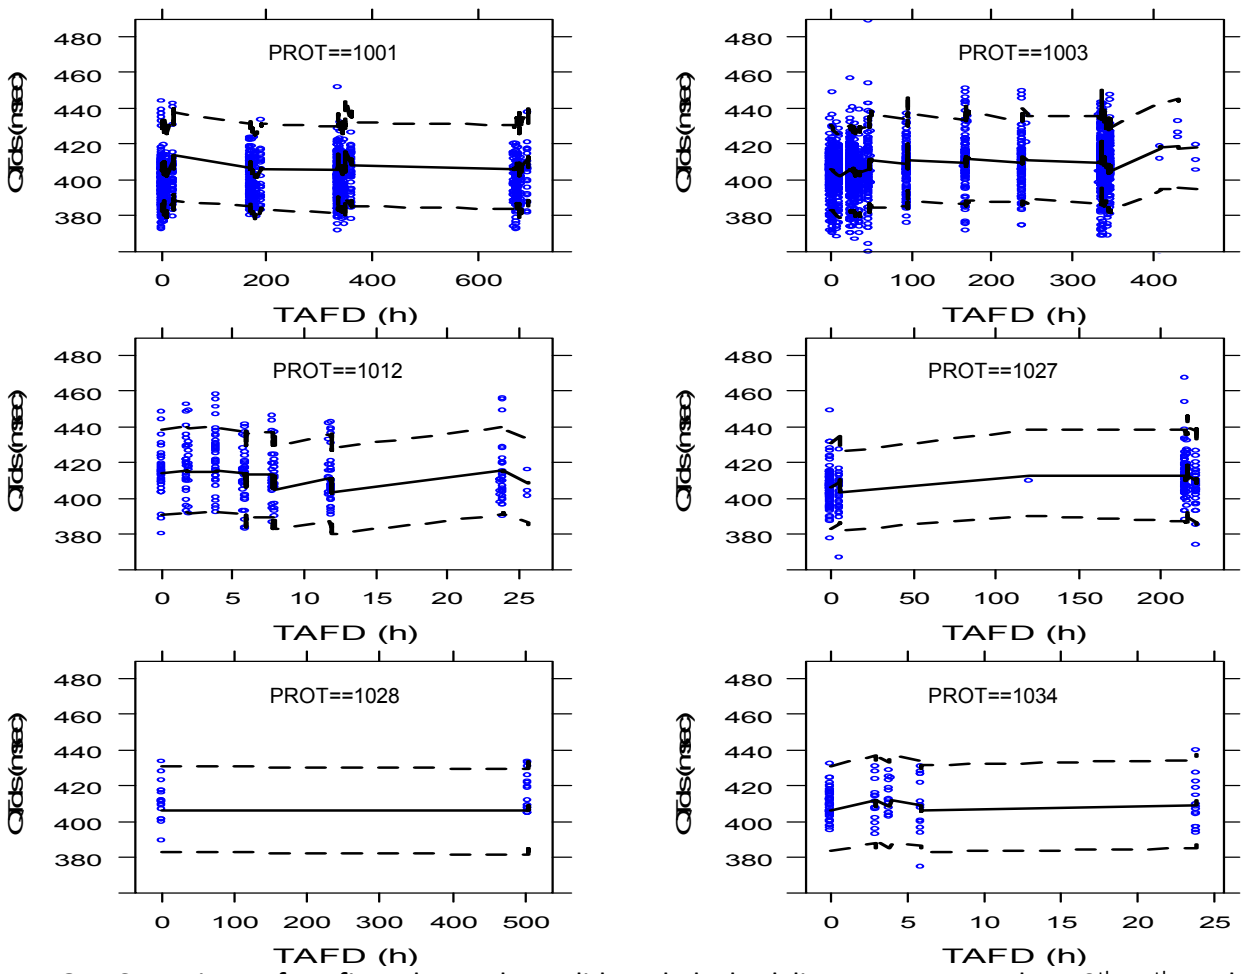
PROT==1001
PROT==1003
PROT==1012
PROT==1027
PROT==1028
PROT==1034
TAFD (h)
380
400
420
440
460
480

QTcIS vs time after first dose; the solid and dashed lines represent the 50th, 5th and 95th percentiles of 1000 simulations; circles are observed QTcIS.

TQT trial simulations demonstrated that QTcIS values >450 ms and >500 ms would occur in 6% (vs. 0.05% in placebo) and 0% subjects' respectively, following administration of a supratherapeutic dose of 24mg. QTcIS change from baseline >30ms and >60ms would occur in 33% (vs. 10% in placebo) and 0.41% subjects' respectively, and the maximum mean ΔΔQTcIS would not exceed 8.5 ms with the upper 95% CI for the mean not exceeding 10 ms for the majority of time points following administration of doses up to 24mg.

Conclusions:

A modified individual correction method, PK-QTcIS, was shown to be best approach to characterise the PK-QT relationship for PH-797804. The model prospectively informed the design of a future TQT study .

Acknowledgements:
We would like to thank the Pharmacometrics group in Sandwich Pfizer for their valuable technical contributions to the modelling work.