Pharmacokinetic/Pharmacodynamic Modeling and Simulation for Concentration-Dependent Bactericidal Activity of a Bicyclolide, Modithromycin

SHIONOGI

Takayuki Katsube[1], Toshihiro Wajima[1], Yoshinori Yamano[2], Yoshitaka Yano[3]

[1]Clinical Research Department, Shionogi & Co., Ltd., Japan, [2]Medicinal Research Laboratories, Shionogi & Co., Ltd., Japan,
[3]Education and Research Center for Clinical Pharmacy, Kyoto Pharmaceutical University, Japan

SONG for you!

Objective

- Modithromycin (also known as S-013420) is a bicyclolide (bridged bicyclic macrolide) that exhibits potent against various pathogens of respiratory tract infections [1]. The mechanism of the bactericidal activity of modithromycin involves inhibition of amplification by binding to the 50S subunit of the bacterial ribosome as observed with telithromycin and clarithromycin [1].
- The aim of this study was to develop PK/PD model of modithromycin to explain its concentration-dependent activity against *S. aureus*, *H. influenzae* and *S. pneumoniae* based on our drug-bacterium interaction model [2-5] and to simulate efficacy of modithromycin in patients using the PK/PD model.

Method

Modeling

- The PK/PD model reported for β-lactams [2-5] was used as a base model (Fig. 1).
- An effect compartment model and a turnover model were incorporated into the base model to explain PK/PD time lag and reduction of efficacy, respectively (Fig. 1).
- *In vitro* time–kill data in 2 types of experiments [6, 7] were used for the modeling.
 - Static experiments, experiments using static concentration in medium
 - Dynamic experiments, experiments using mechanically simulated concentration in medium
 - Test drug: modithromycin, telithromycin and clarithromycin
 - Test strain: *S. aureus* SR20405, *H. influenzae* SR24159, SR24165 and SR24169, and *S. pneumoniae* SR26132
- The bacterial count data were fitted to the PK/PD model for the drug-bacteria combinations by the M3 method [8] using NONMEM (Version 7.1).

Simulation

- The followings were considered to simulate time courses of bacterial counts for modithromycin in patients with pneumonia infected with *S. aureus*, *H. influenzae* and *S. pneumoniae*.
 - MIC distribution of clinical isolates collected at Japanese medical facilities in 2004 [6] (Fig. 2).
 - PD parameters (K_{max}/KC_{50} or Slope) based on the simulated MIC using the relationships shown in Fig. 4.
 - PK parameters estimated from the data in Ph1 studies [9, 10] as typical values in patients.
 - 20 of fraction in epithelial lining fluid (ELF) to plasma to simulate lung distribution [11].
 - Unbound fraction of 0.8 in ELF
 - 30% of inter-individual variability for parameters assuming a log-normal distribution.
 - Initial inoculum of 10^4 and 10^7 CFU/lung to simulate different severities.
- Three QD regimens with 2-fold dosage as an initial loading dose were simulated.
- 1000 simulations were performed for each scenario.
- Probability of bacterial eradication [log(CFU/lung) < 0] in simulated patients was calculated as efficacy index.

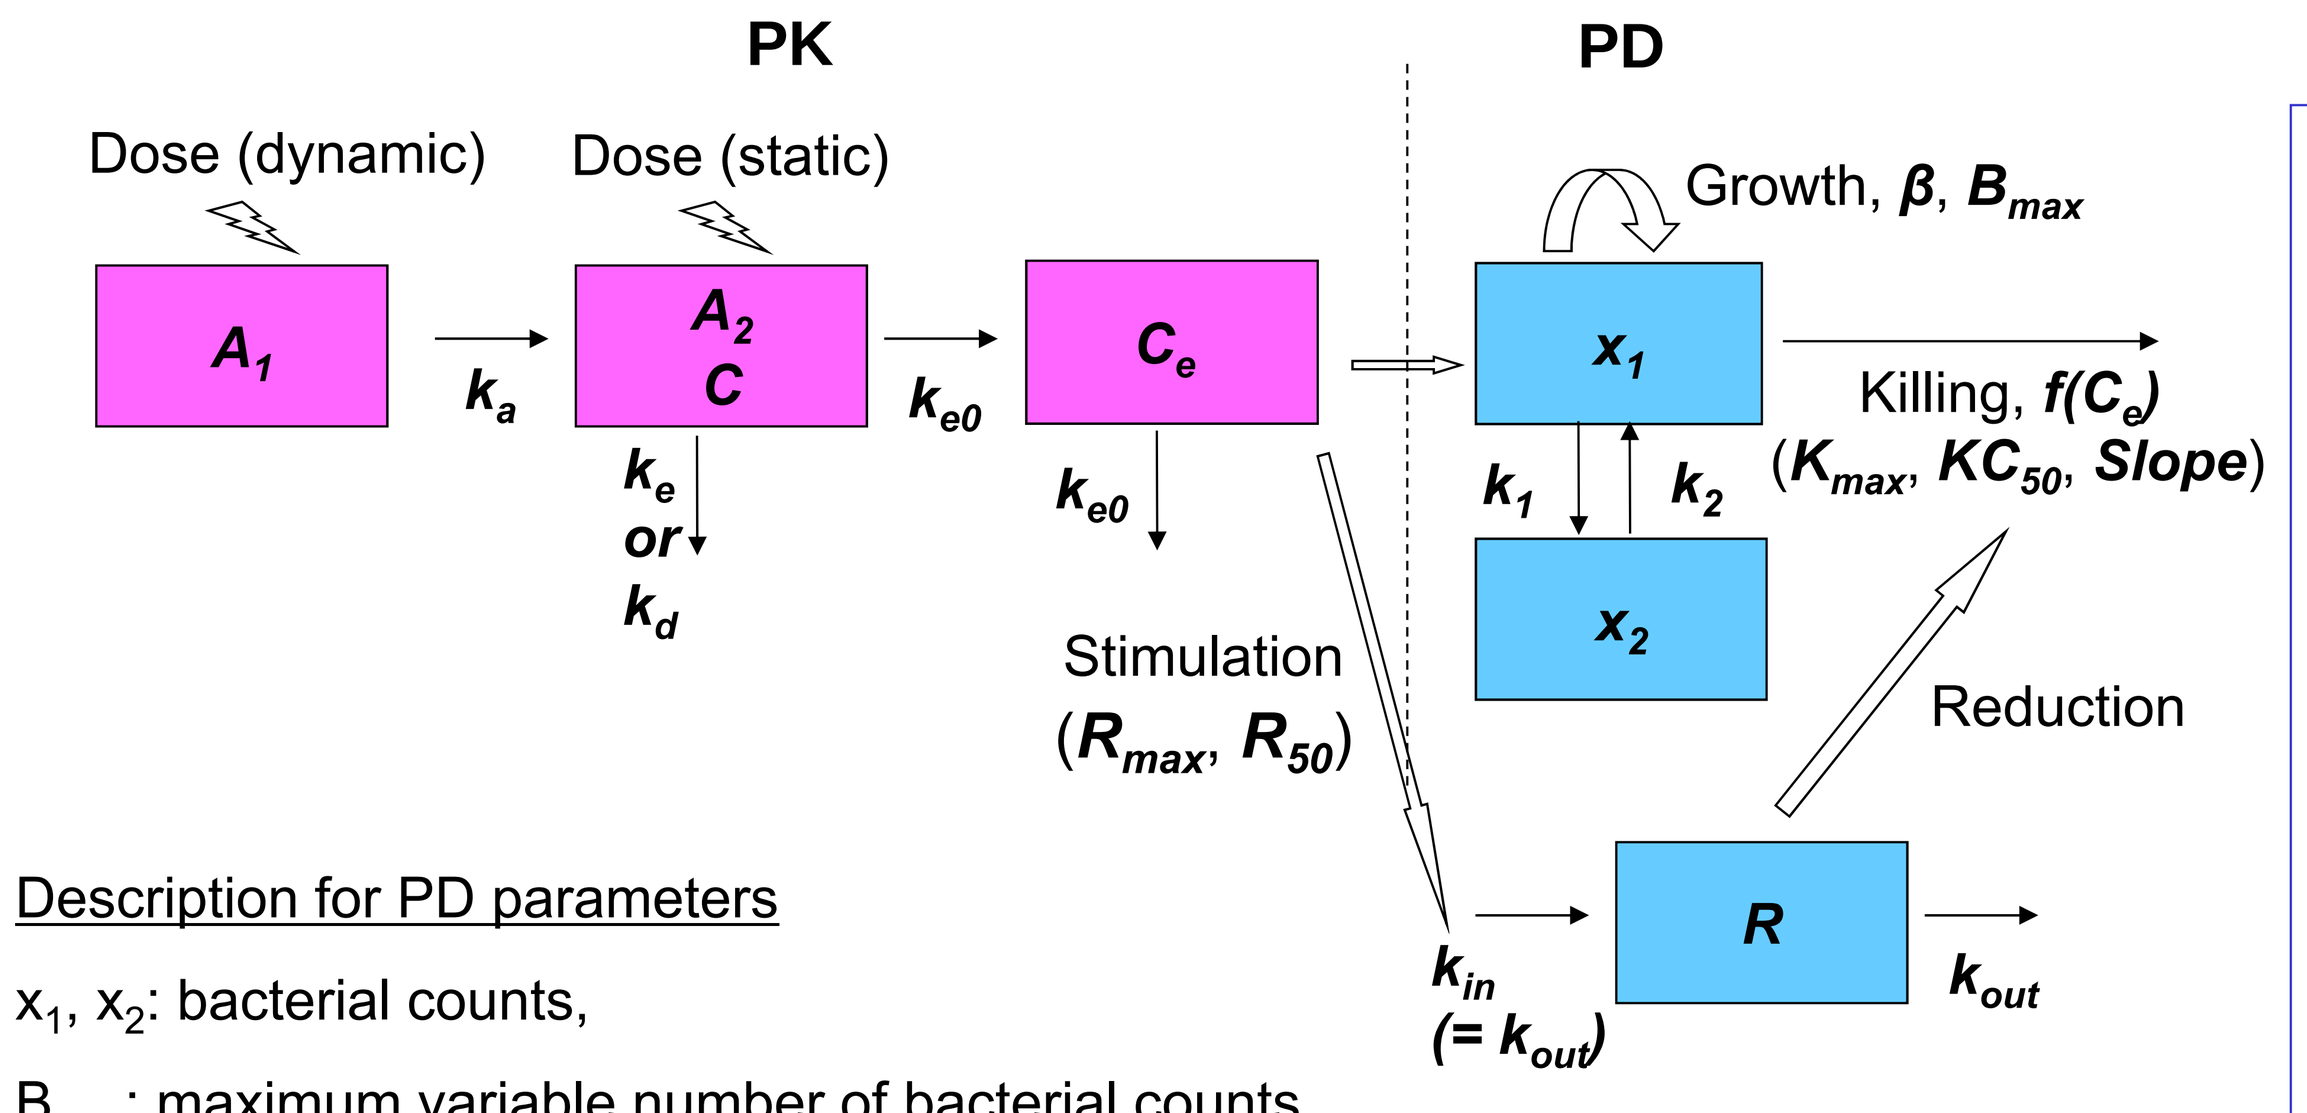

PK
PD
Dose (dynamic)
Dose (static)
A1
A2 C
Ce
x1
x2
R
ka
ke0
ke or kd
k1
k2
Growth, β, Bmax
Killing, f(Ce) (Kmax, KC50, Slope)
Stimulation (Rmax, R50)
Reduction
kin (= kout)
kout

$$\frac{dx_1}{dt} = \beta\cdot(B_{max}-x_1)\cdot x_1 - f(C_e)\cdot x_1 - k_1\cdot x_1 + k_2\cdot x_2$$

$$\frac{dx_2}{dt} = k_1\cdot x_1 - k_2\cdot x_2$$

$$f(C_e)=\frac{K_{max}\cdot C_e}{R\cdot KC_{50}+C_e} \quad \text{or} \quad f(C_e)=Slope\cdot C_e/R$$

$$\frac{dR}{dt} = k_{in}\cdot\left(1+\frac{R_{max}\cdot C_e}{R_{50}+C_e}\right) - k_{out}\cdot R$$

Dynamic

$$\frac{dA_1}{dt} = -k_a\cdot A_1$$

$$\frac{dA_2}{dt} = k_a\cdot A_1 - k_e\cdot A_2$$

$$\frac{dC_e}{dt} = k_{e0}\cdot\frac{A_2}{V_d/F} - k_{e0}\cdot C_e$$

Static

$$\frac{dC}{dt} = -k_d\cdot C$$

$$\frac{dC_e}{dt} = k_{e0}\cdot C - k_{e0}\cdot C_e$$

Description for PD parameters

x_1, x_2: bacterial counts,

B_{max}: maximum variable number of bacterial counts,

β: growth rate constant, K_{max}: maximum killing rate constant,

KC_{50}: Michaelis-Menten constant, k_1, k_2: 1st-order hypothetical transfer rate constant,

C_e: drug concentration in the effect compartment,

R: hypothetical variable in the turnover model related to increase in KC_{50},

R_{max}: maximum rate constant, R_{50}: Michaelis-Menten constant,

k_{out}: 1st-order rate constant for the loss of R, k_{in}: 0-order input rate,

Fig. 1 Scheme of the PK/PD model for describing the bactericidal activity.

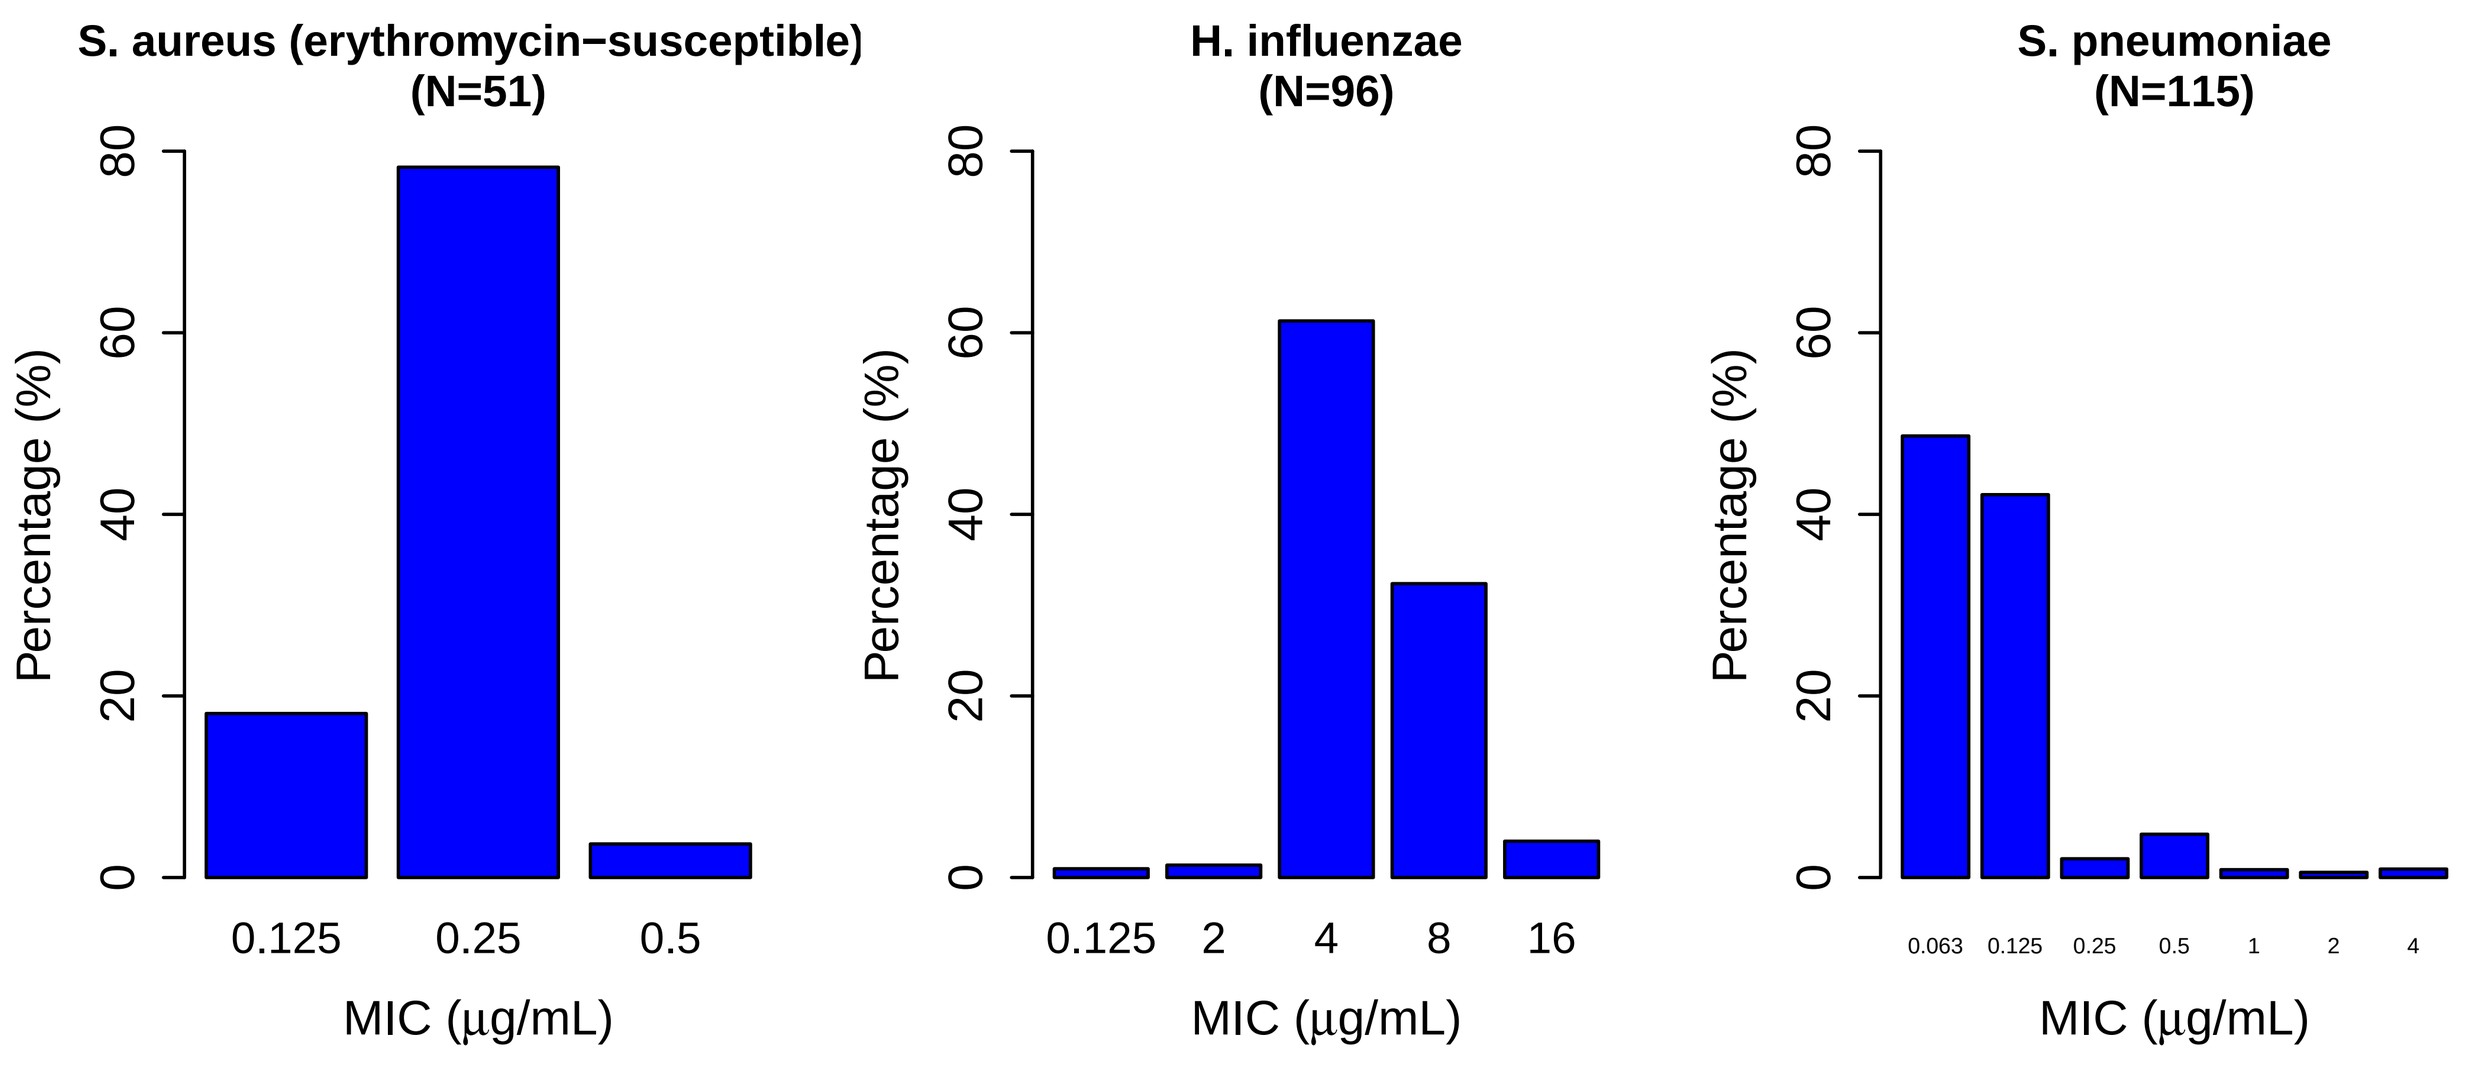

S. aureus (erythromycin−susceptible) (N=51)
H. influenzae (N=96)
S. pneumoniae (N=115)
Percentage (%)
MIC (μg/mL)

Fig. 2 MIC distribution of each bacterium used for the simulations.

Result - Modeling

- The developed PK/PD model well described *in vitro* time-kill data for all combinations of drugs and bacterium (Fig. 3 as typical cases).
- The PD parameters (K_{max}/KC_{50} and Slope) strongly correlated with MIC (Fig. 4).
- The simulated bactericidal activity of modithromycin strongly correlated to AUC/MIC (Fig. 5).

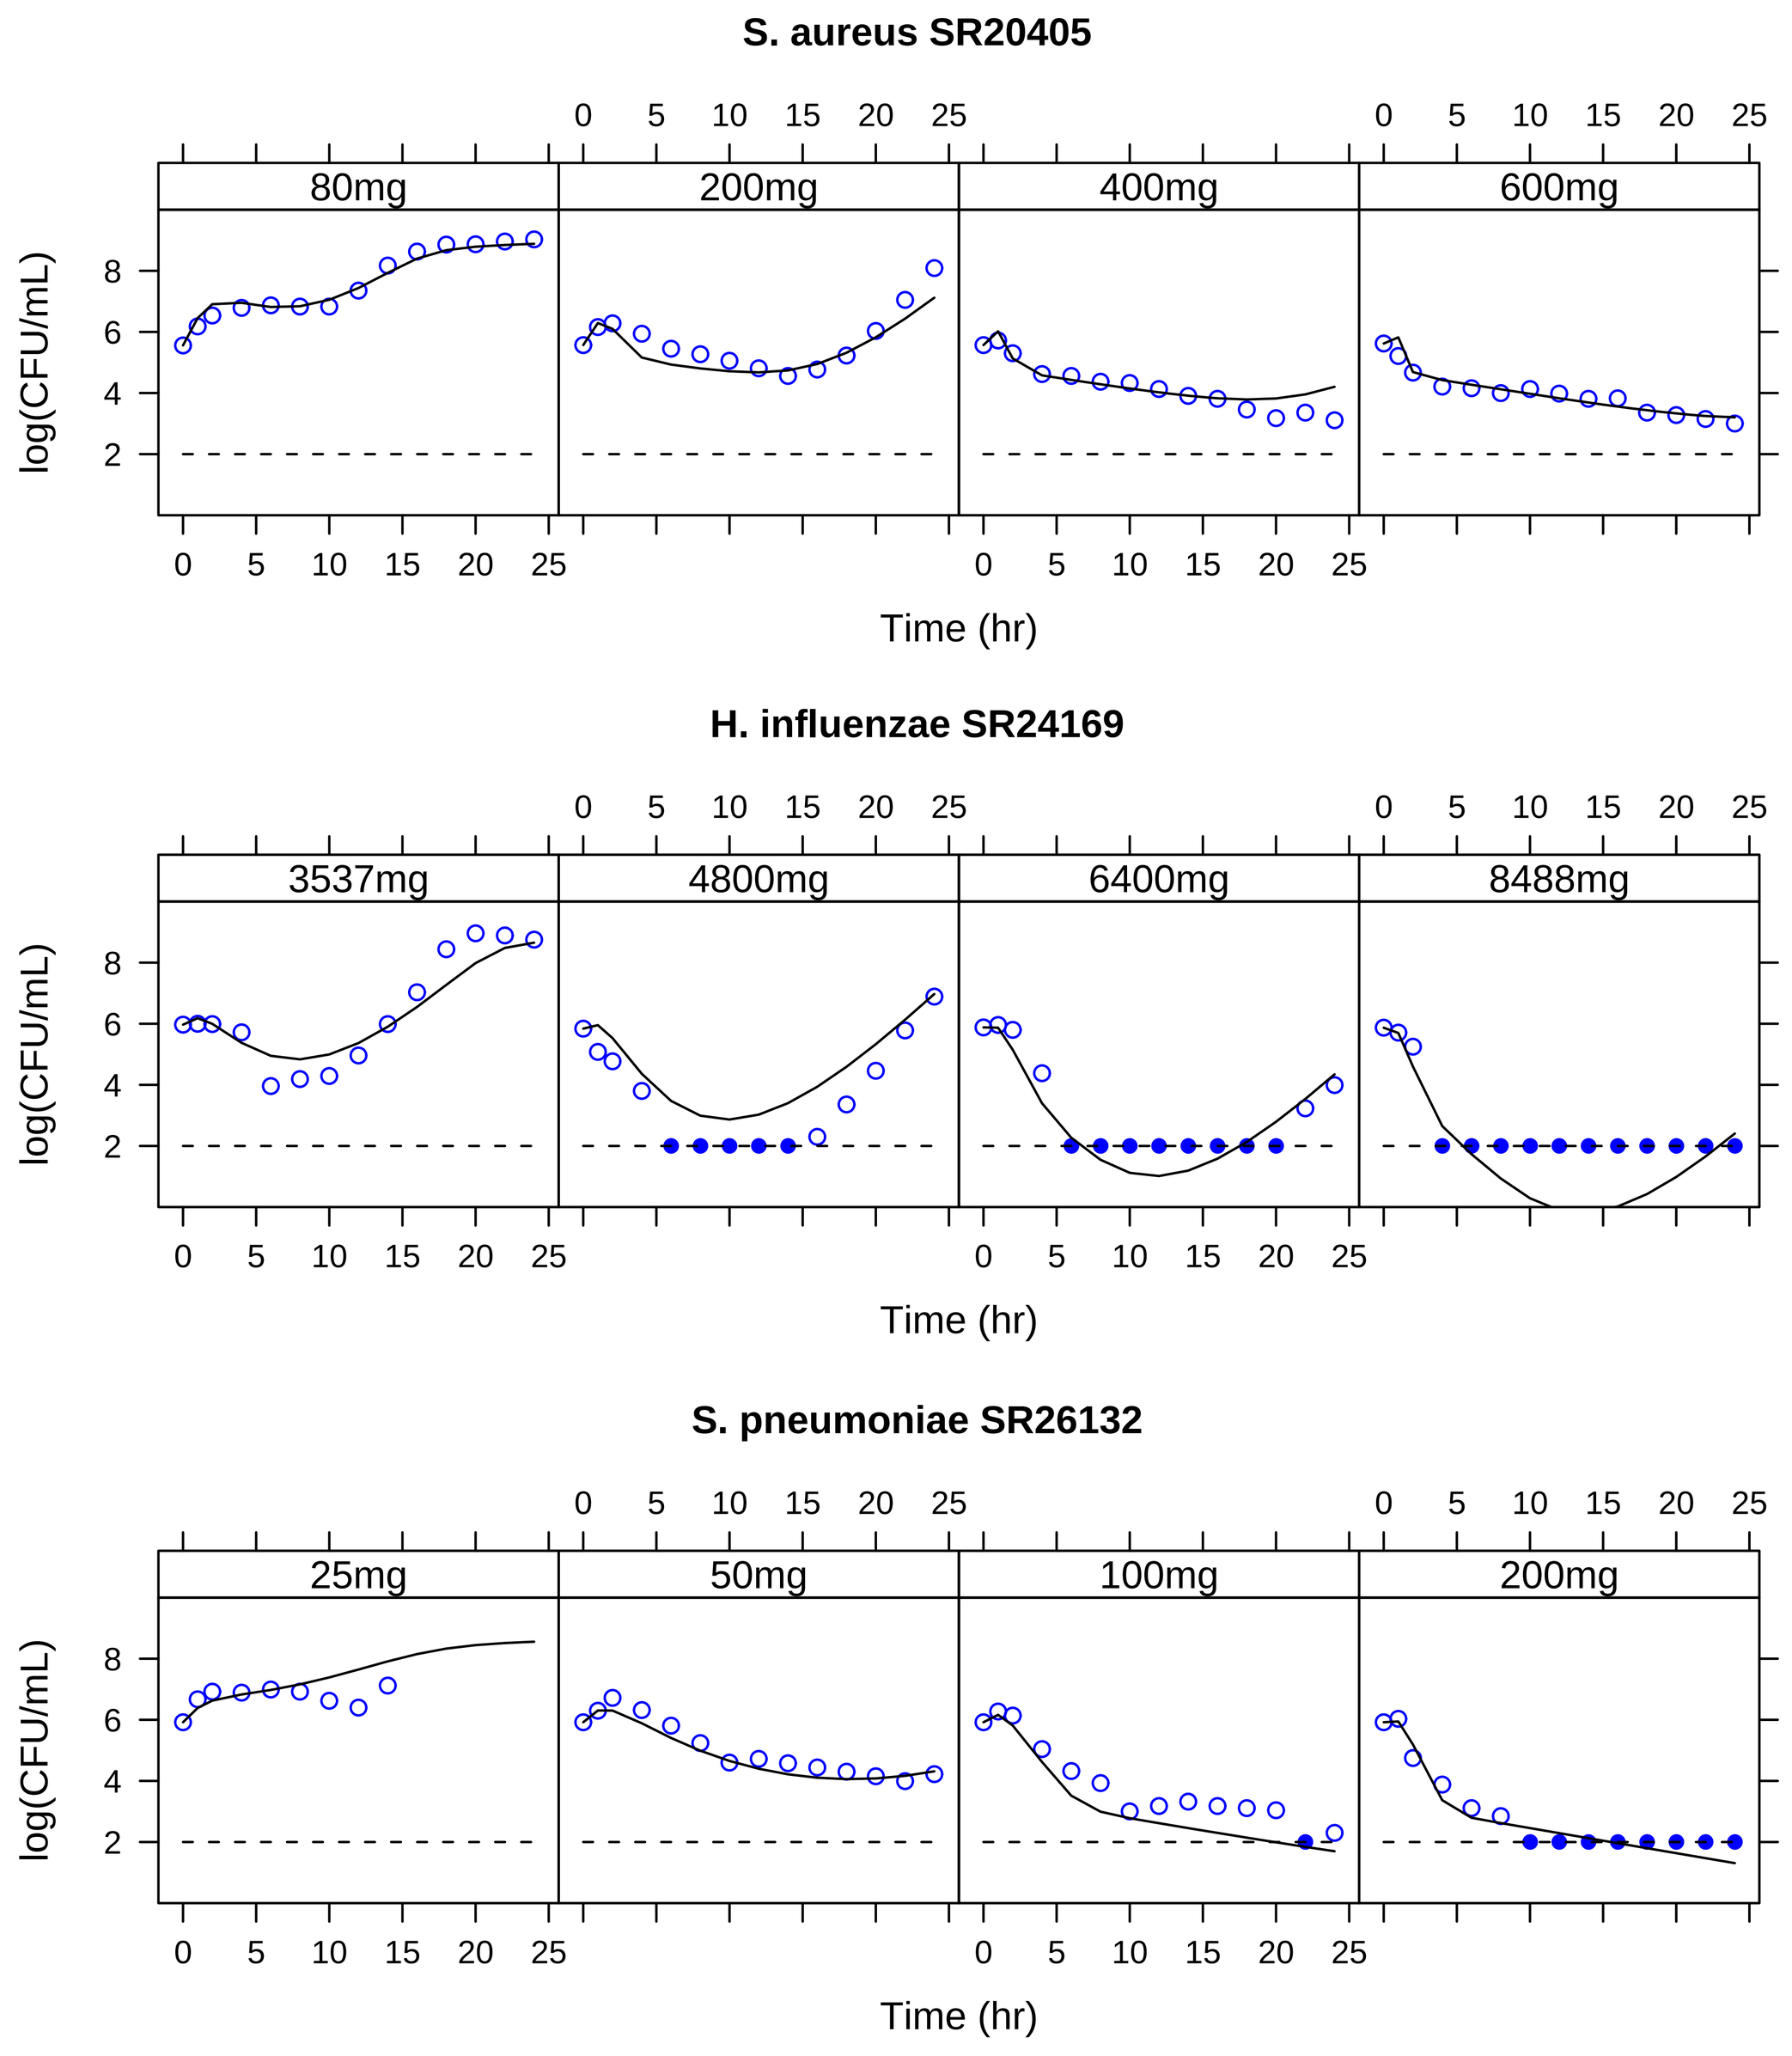

S. aureus SR20405
80mg
200mg
400mg
600mg
H. influenzae SR24169
3537mg
4800mg
6400mg
8488mg
S. pneumoniae SR26132
25mg
50mg
100mg
200mg
log(CFU/mL)
Time (hr)

Fig. 3 Typical results of model fitting with *in vitro* bacterial count data of modithromycin against each strain.
Dotted line: the lower limit of quantification.
The data below the limit of quantification were plotted as 2 of log(CFU/mL) using closed circles.

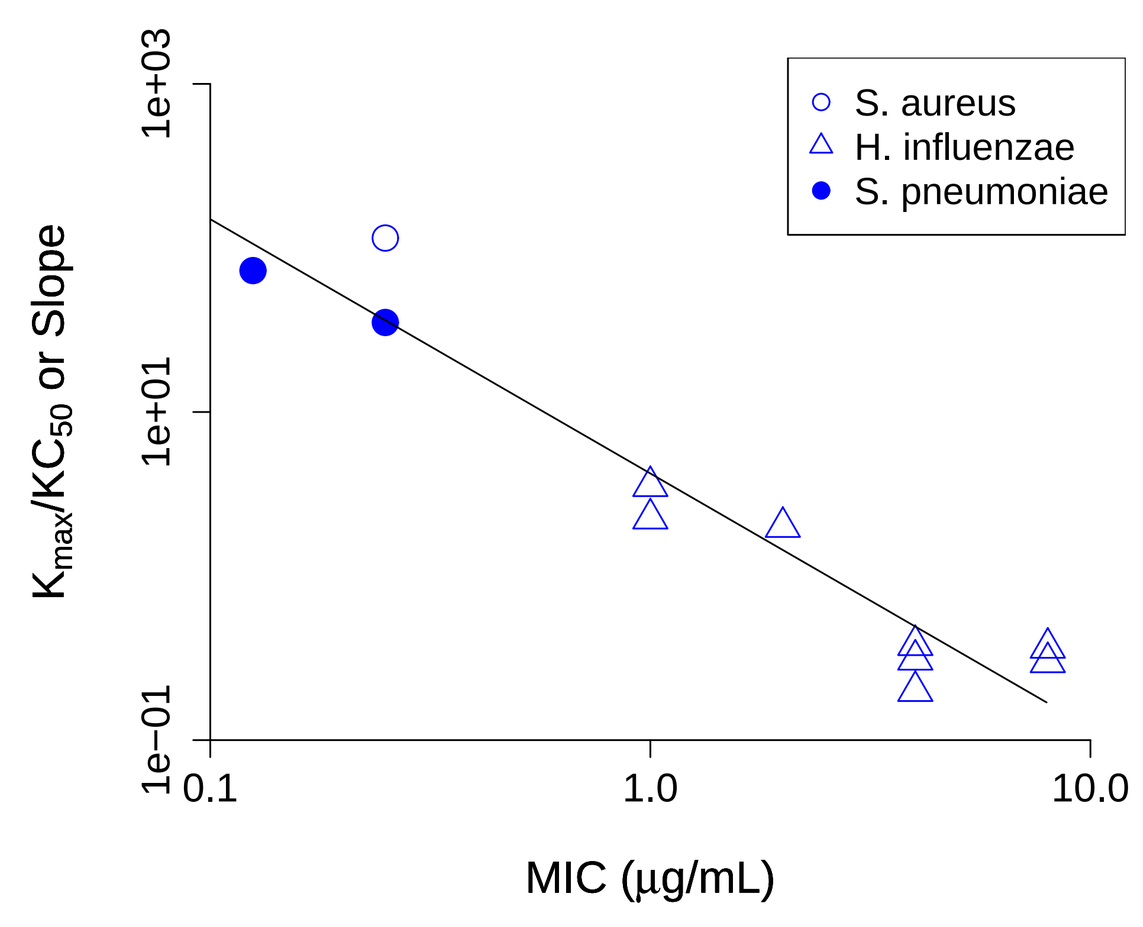

S. aureus
H. influenzae
S. pneumoniae
Kmax/KC50 or Slope
MIC (μg/mL)

Fig. 4 Relationships between PD parameters and MIC.

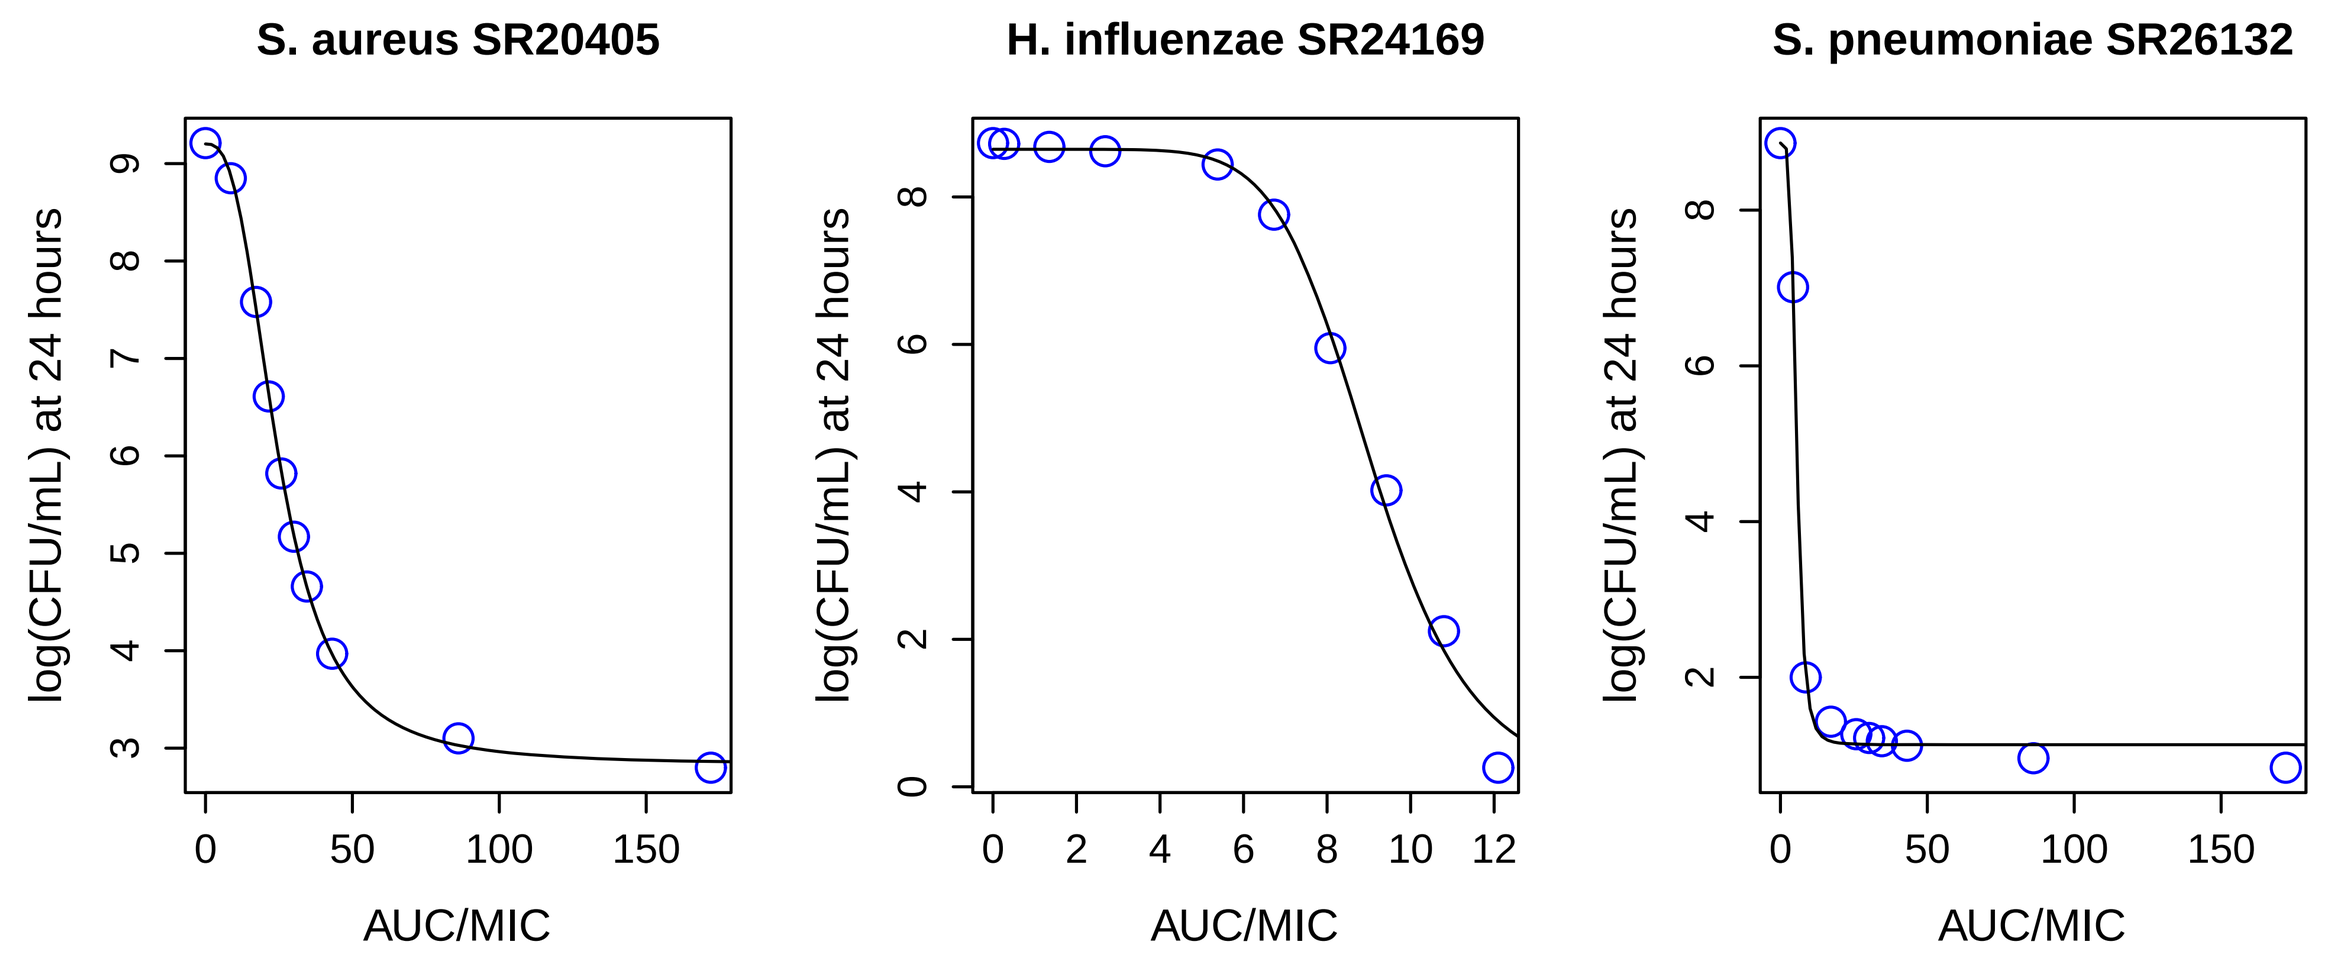

S. aureus SR20405
H. influenzae SR24169
S. pneumoniae SR26132
log(CFU/mL) at 24 hours
AUC/MIC

Fig. 5 Relationships between simulated bacterial counts at 24 hours and AUC/MIC of modithromycin for each strain.

Result - Simulation

- The parameters used in the simulations are shown in Table 1.
- Typically any dosage regimens exhibited bactericidal effect regardless the initial inoculums, except for the lowest regimen against *H. influenzae* (Fig. 6).
- The probability as efficacy index against *S. aureus*, *H. influenzae* and *S. pneumoniae* exceeded 80% at 200 mg QD, 400 mg QD and 100 mg QD, respectively, in both initial inoculums (Fig. 7).
- The probability in smaller and larger initial inoculums reached 80% within 3 days and 5 days from the start of effective dosage regimen, respectively (Fig. 7).

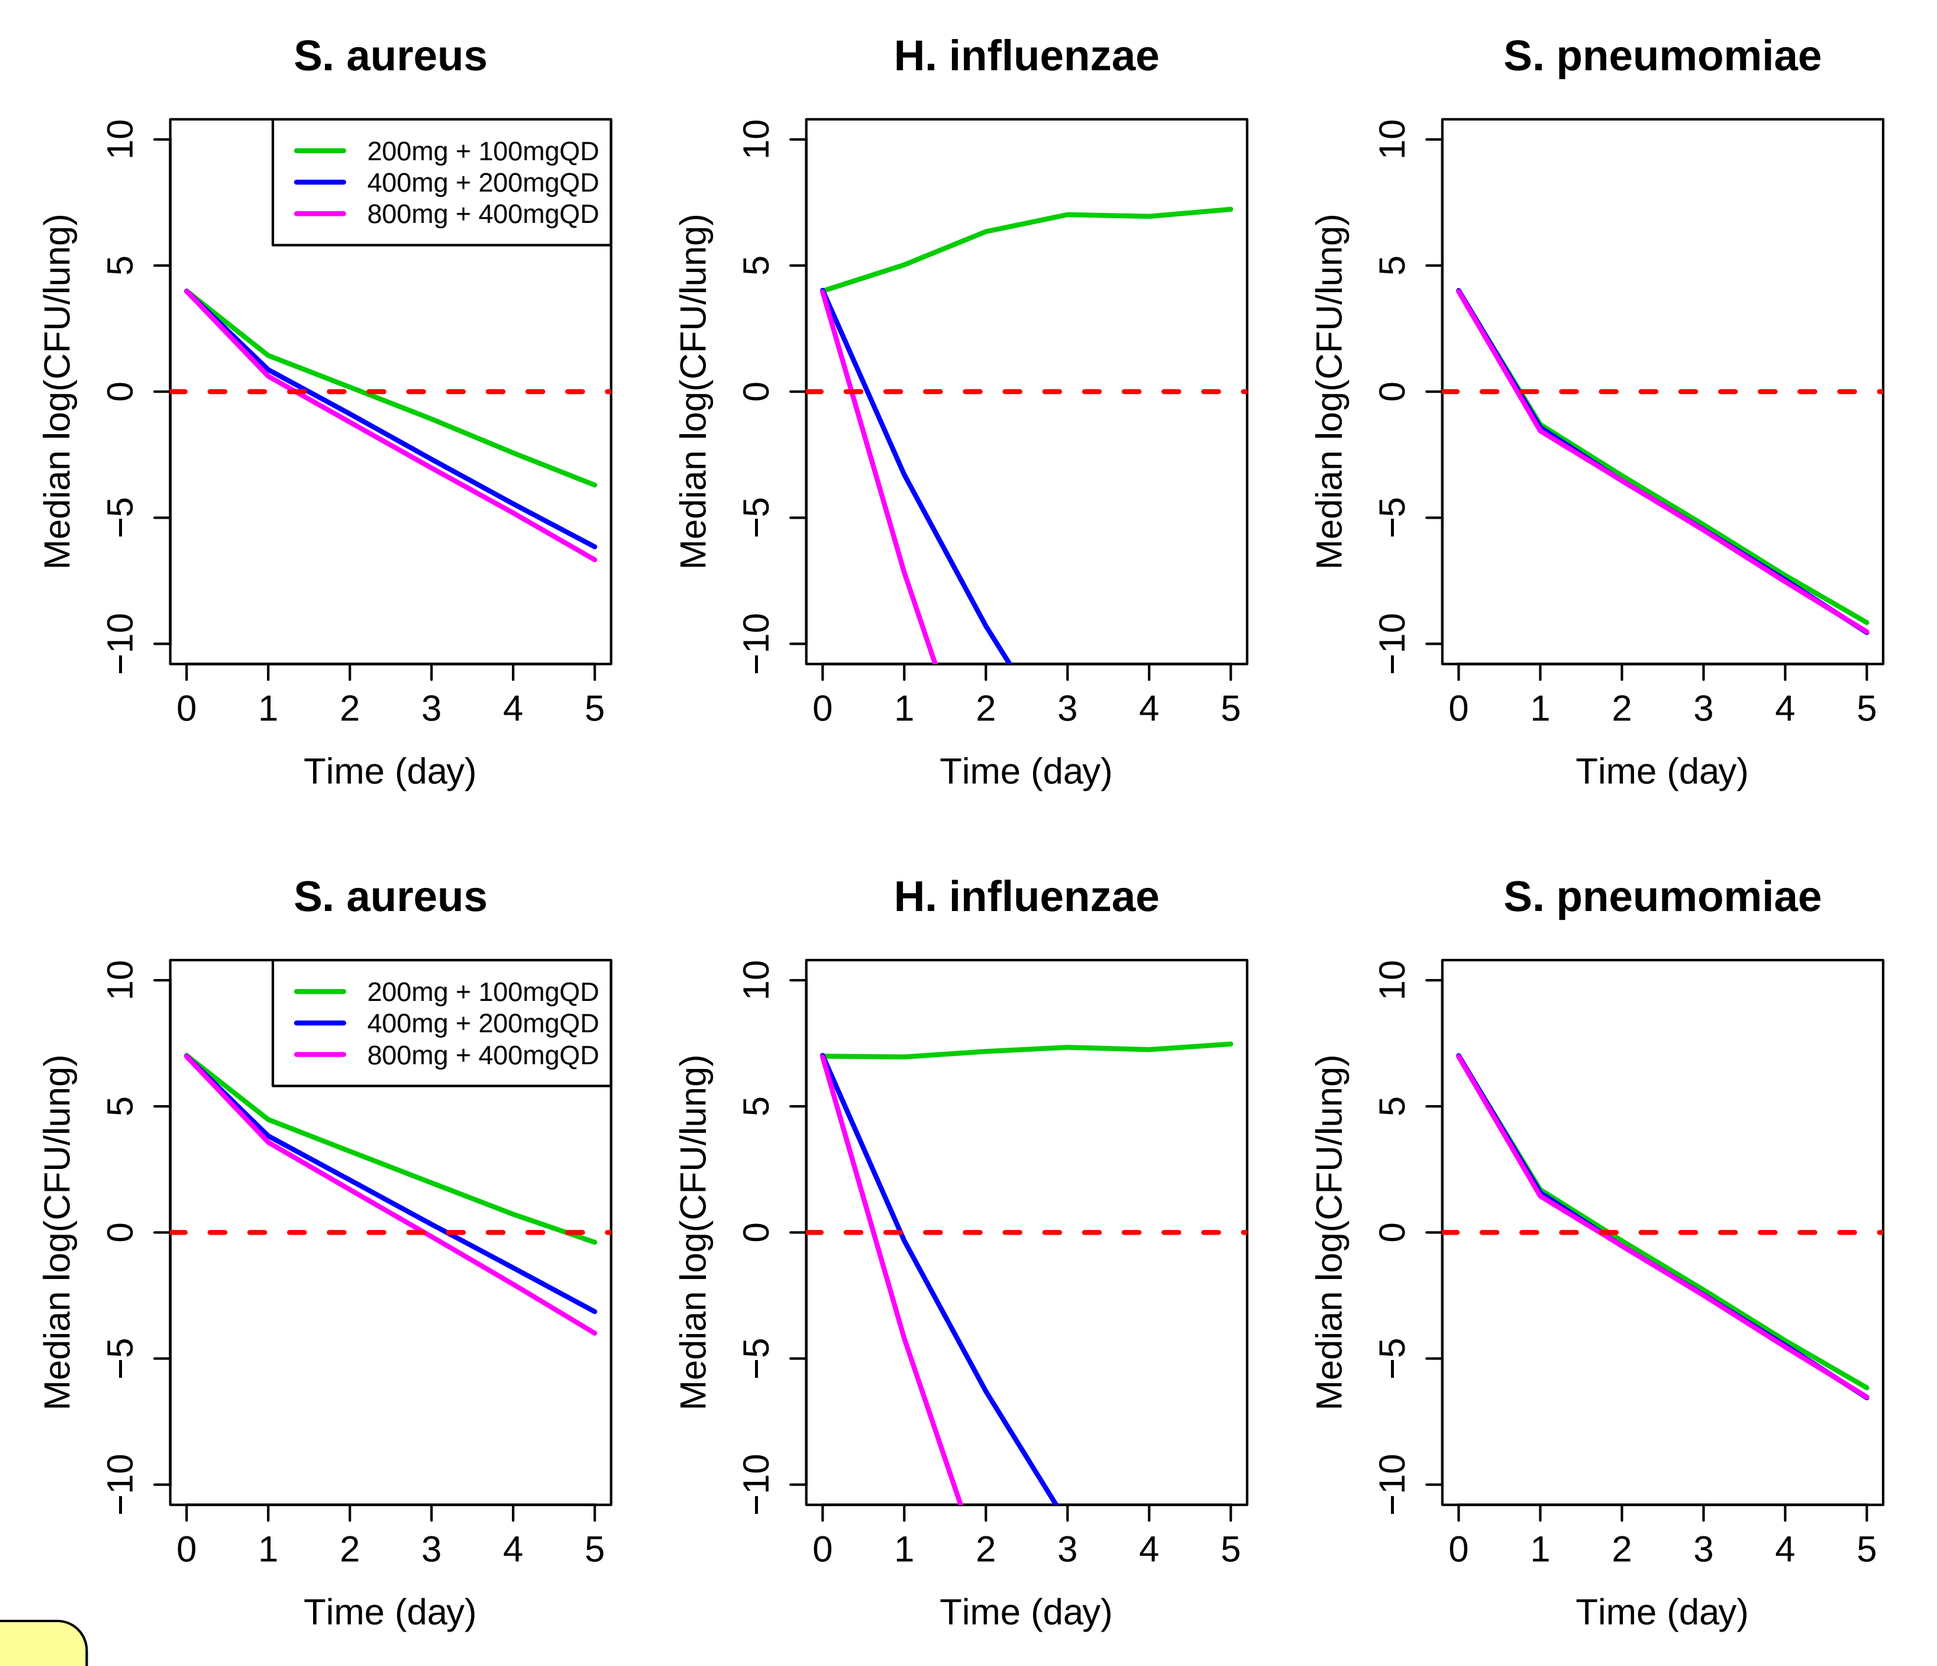

S. aureus
H. influenzae
S. pneumomiae
200mg + 100mgQD
400mg + 200mgQD
800mg + 400mgQD
Median log(CFU/lung)
Time (day)

Fig. 6 Time courses of median simulated bacterial counts in 3 dosage regimens.
Initial inoculum: 10^4 (upper) and 10^7 (lower) CFU/lung.

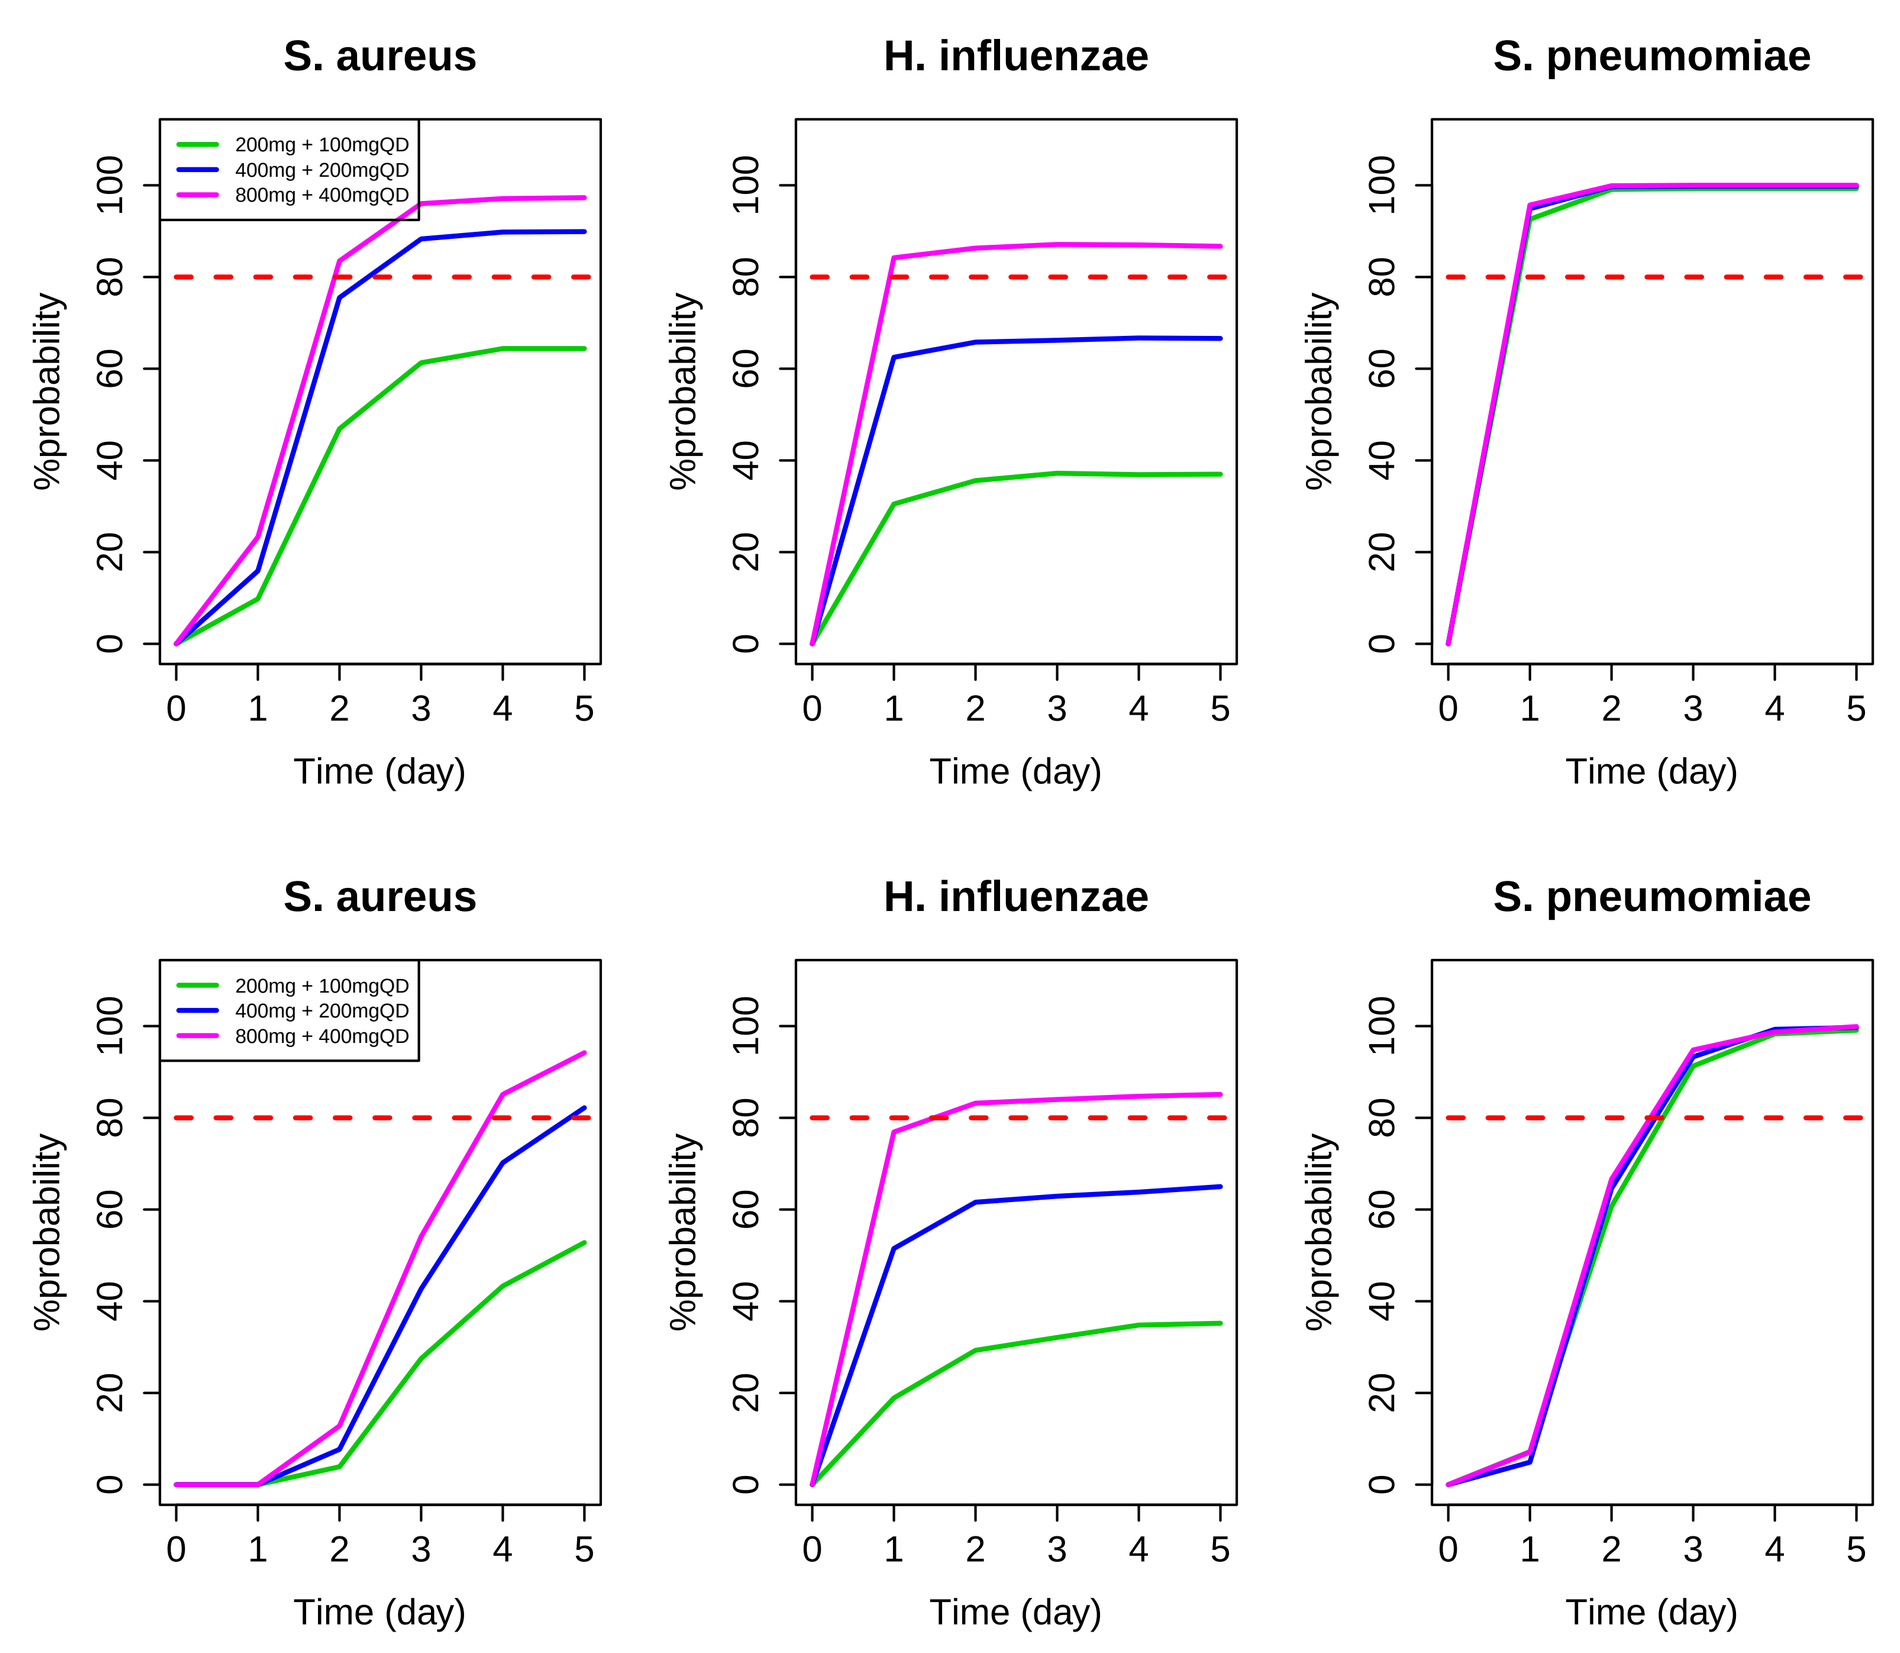

S. aureus
H. influenzae
S. pneumomiae
200mg + 100mgQD
400mg + 200mgQD
800mg + 400mgQD
%probability
Time (day)

Fig. 7 Time courses of the probability [log(CFU/lung) < 0] as efficacy index in 3 dosage regimens.
Initial inoculum: 10^4 (upper) and 10^7 (lower) CFU/lung.

Table 1 PK/PD parameters to simulate bactericidal activity of modithromycin

Bacterium	*S. aureus*	*H. influenzae*	*S. pneumoniae*
β (10^{-10}*hr^{-1}*(CFU/mL))	18.4	21.2	18.1
B_{max} (10^{10}*CFU/mL)	0.129	0.0531	0.0716
k_1 (hr^{-1})	0.0470	0.00852	0.000938
k_2 (hr^{-1})	0.184	1.08	0.192
K_{max} (hr^{-1})	199	-	5.58
KC_{50} (μg/mL)	K_{max}/[exp(1.44)*$MIC^{-1.55}$]	-	K_{max}/[exp(1.44)*$MIC^{-1.55}$]
Slope (hr^{-1}*μg/mL)	-	exp(1.44)*$MIC^{-1.55}$	-
k_{e0} (hr^{-1})	0.144	10.0	1.66
R_{max} (hr^{-1})	37.8	-	-
R_{50} (μg/mL)	0.398	-	-
K_{out} (hr^{-1})	0.129	-	-
V_d/F (L)	709	709	709
k_a (hr^{-1})	1.28	1.28	1.28
k_e (hr^{-1})	0.039	0.039	0.039

Summary and Conclusion

- The developed PK/PD model well described *in vitro* time-kill data of modithromycin against test strains.
- The initial loading dose followed by 200 mg QD, 400 mg QD and 100 mg QD of modithromycin would be potent against *S. aureus, H. influenzae* and *S. pneumoniae*, respectively.
- The 5-day dosing of modithromycin would be expected to exhibit efficacy regardless severity.

Reference

1) Van Bambeke F et al. Expert Opin Pharmacother. 2008;9:267-283.
2) Yano Y et al. J Pharm Sci. 1998;87:1177-1183.
3) Katsube T et al. J Pharm Sci. 2008. 97: 4108-4117.
4) Katsube T et al. J Pharm Sci. 2008. 97: 1606-1614.
5) Katsube T et al. J Pharm Sci. 2010. 99: 2483-2491.
6) Homma T et al. J Antimicrob Chemother. 2010;65:1433-1440.
7) Homma T et al. Antimicrob Agent Chemother. 2010;54:4300-4305.
8) Beal SL. J Pharmacokinet Pharmacodyn. 2001;28481-504.
9) Jiang LJ et al. Antimicrob Agent Chemother. 2009;53:3218-3225.
10) Shimada J et al. 47th Intersci. Conf. Antimicrob Agents Chemother, abstr. A-799; 2007.
11) Furuie H et al. Antimicrob Agent Chemother. 2010;54:866-870.