Effect of Fluticasone Furoate Treatment on Cortisol Circadian Rhythm in Healthy Chinese Subjects

Yan Ren and Peiming Ma

Department of Clinical Pharmacology Modelling and Simulation, R&D Shanghai, GlaxoSmithKline, China

Creation Date: 04/20/2014

Objectives

Fluticasone furoate (FF) is a novel inhaled corticosteroid (ICS) with potent glucocorticoid activity. It is being developed as a monotherapy, and also as a potential steroid component in a once daily ICS/long-lasting beta agonist (LABA) combination with vilanterol (VI) for asthma and COPD. Corticosteroids, but not VI, have the potential to induce dose-dependent systemic effects on the hypothalamic-pituitary-adrenal (HPA) axis. Cortisol suppression has been observed in asthma patients with normal HPA axis function at baseline on receiving high doses of ICSs, and is associated with adverse effects on a number of physiological processes. The relationship between FF systemic exposure and 24-hr weighted mean serum cortisol has been well described previously [Allen A., Clin. Pharmacokinet. 2013]. In this study cortisol concentrations in healthy Chinese male subjects before and after repeat FF dosing were measured and characterized by a cosine model. The direct influence of FF on cortisol circadian rhythm was explored.

Clinical Trial

A single-centre, double-blind, placebo-controlled, crossover, randomized, single- and repeat-dose study was conducted in healthy male Chinese subjects. The study conformed to the requirements of Declaration of Helsinki and its amendments to Good Clinical Practice guidelines. The study objective was to evaluate the pharmacokinetic-pharmacodynamic profile, safety and tolerability of 50, 100 and 200 mcg of FF in combination with 25 mcg VI. An incomplete Latin-square design of 4 sequences by 4 treatment periods was used. A total of 16 subjects aged 18-45 years were randomized with the aim of achieving at least 10 evaluable subjects in each treatment period. Subjects were fasted on each dosing day from previous midnight to 4 hours post dose. Water was allowed freely except for 1 hour before and after dosing. Standard meals were provided. Baseline serum cortisol concentrations were measured 5 min before dosing on Day 1. Steady-state FF concentrations were achieved after repeat dosing for 4 days. On Day 7 blood samples were collected at 1, 2, 4, 6, 8, 10, 12, 16, 24 hr post dose with the purpose of evaluating the effect of FF treatment on cortisol concentrations.

Plasma FF and serum cortisol concentrations were analyzed using validated analytical methods based on solid phase extraction and protein precipitation, respectively, followed by HPLC-MS/MS. The lower limits of quantification (LLQ) of FF and serum cortisol concentrations were 10 pg/ml and 5 ng/ml respectively.

Methods

Nonlinear mixed-effect modelling was used (NONMEM® 7.2) with the first-order conditional estimation with interaction as the method for minimization. R 3.0.2 was used for data handling, exploratory diagnostics, and plotting.

1. Cortisol circadian rhythm

Time profile of cortisol circadian rhythm Circ(t) were described by 3 cosine functions.

❖ $\mathrm{Circ}(t) = A\cdot\cos(2\pi(t-\alpha)/24) + B\cdot\cos(2\pi(t-\beta)/12) + C\cdot\cos(2\pi(t-\gamma)/8)$,

where A, B and C are the amplitudes of cosine terms; α, β and γ are acrophases (times to reach peak) of the cosine terms.

Baseline cortisol concentration per subject per treatment period were directly used as a covariate, called BASE, for the cortisol response Cort(t).

❖ $\mathrm{Cort}(t) = \mathrm{Circ}(t) + \mathrm{INT} + \mathrm{BI}\cdot\mathrm{BASE}$

where INT and BI are, respectively, intercept and slope terms.

2. Effect of FF on cortisol

Observed FF concentration was used directly to affect cortisol profile R(t). Missing FF concentrations due to LLQ were imputed as half of the LLQ (i.e., 5 pg/ml) at the first time point and 0 afterwards in each period.The following models were tested to describe the relationship of FF concentration and R(t):

a) $R(t) = \mathrm{Cort}(t) - E\cdot FF/(EC50 + FF)$
b) $R(t) = \mathrm{Cort}(t) - S\cdot FF$
c) $R(t) = \mathrm{Cort}(t)\cdot(1 - E\cdot FF/(EC50 + FF))$

where in a) and c) E is the maximum effect of FF on cortisol lowering, and EC50 is the FF concentration reaching 50% of the maximum cortisol lowering effect, and in b) S is the slope quantifying the FF effect.

3. Covariate effects

Age and body weight were tested as continuous covariates on INT, dosing regimen sequence, treatment period and dosage amount were tested as categorical covariates of R(t). These covariates were not kept in the final model since they did not decrease the objective function value significantly.

4. Variability

Inter-subject variability was implemented on parameters. Various residual error models were tried. Inter-occasion variability was tested on INT but not kept in the final model.

Model a) was selected as final.

Results

Thirteen subjects reported ≥ 1 adverse events (AEs). All were of mild intensity. Incidence of AEs in FF/VI 100/25 & 200/25 groups was similar to or lower than the placebo group. No clinically relevant findings in vital signs, ECG or physical examination were observed.

The final model a) appeared to have adequately described the endogenous cortisol circadian rhythm. Although data were limited (e.g., cortisol was not measured from10 pm to 8 am), results showed cortisol profiles consistent with previous publication [Zhao ZY et al., Steroids, 2003]. FF concentration, instead of FF dose, was sufficient when used as a covariate affecting cortisol change using an Emax model. Parameter estimates are listed below. Some goodness-of-fit plots are shown in Figure 1. The effect of FF concentration after repeat dosing on cortisol circadian time profile is described in Figure 2.

Table. Parameter estimates from the final model

Parameter	Estimate (%RSE)	Variability (%RSE)	Residual Error (%RSE)
A (ng/ml)	31.0 (9.97)	--	Proportional Error
B (ng/ml)	10.9 (30.5)	0.075 (67.5), Exp.	0.354 (3.36)
C (ng/ml)	6.35 (28.8)	--	Additive Error (ng/ml)
α (hr)	8.11 (5.14)	--	--
β (hr)	4.80 (6.21	0.717 (85.9), Add.	
γ (hr)	4.91 (6.17)	--	
Base INT (ng/ml)	34.7 (7.09)	0.333 (36.3), Exp.	
Base Slope	0.16 (16.7)	--	
E (ng/ml)	50.6 (46.4)	--	
EC50 (pg/ml)	99.4 (65.3)	--	

Figure 1: Diagnostic plots for the final model

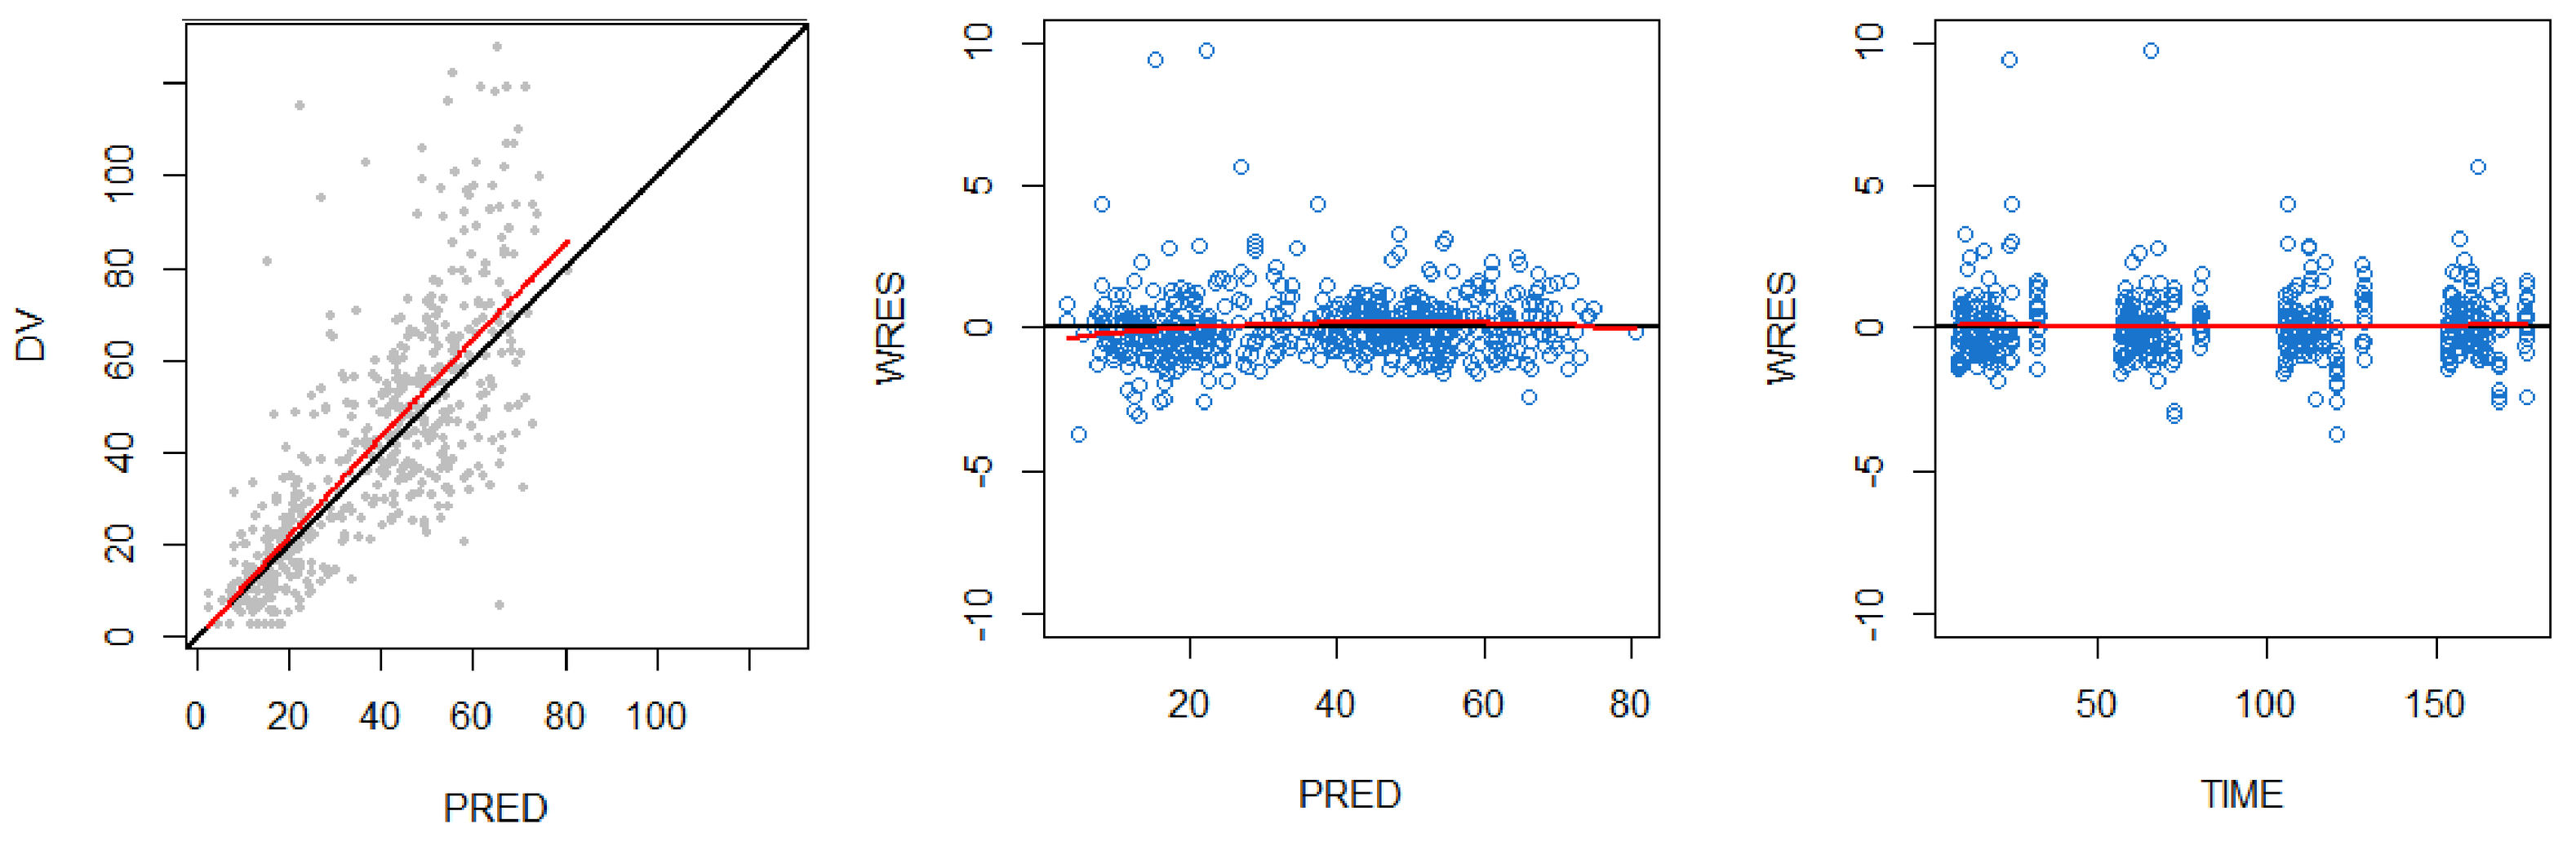
DV
PRED
WRES
PRED
WRES
TIME

Figure 2: Effect of FF on Cortisol circadian time profile at different doses.

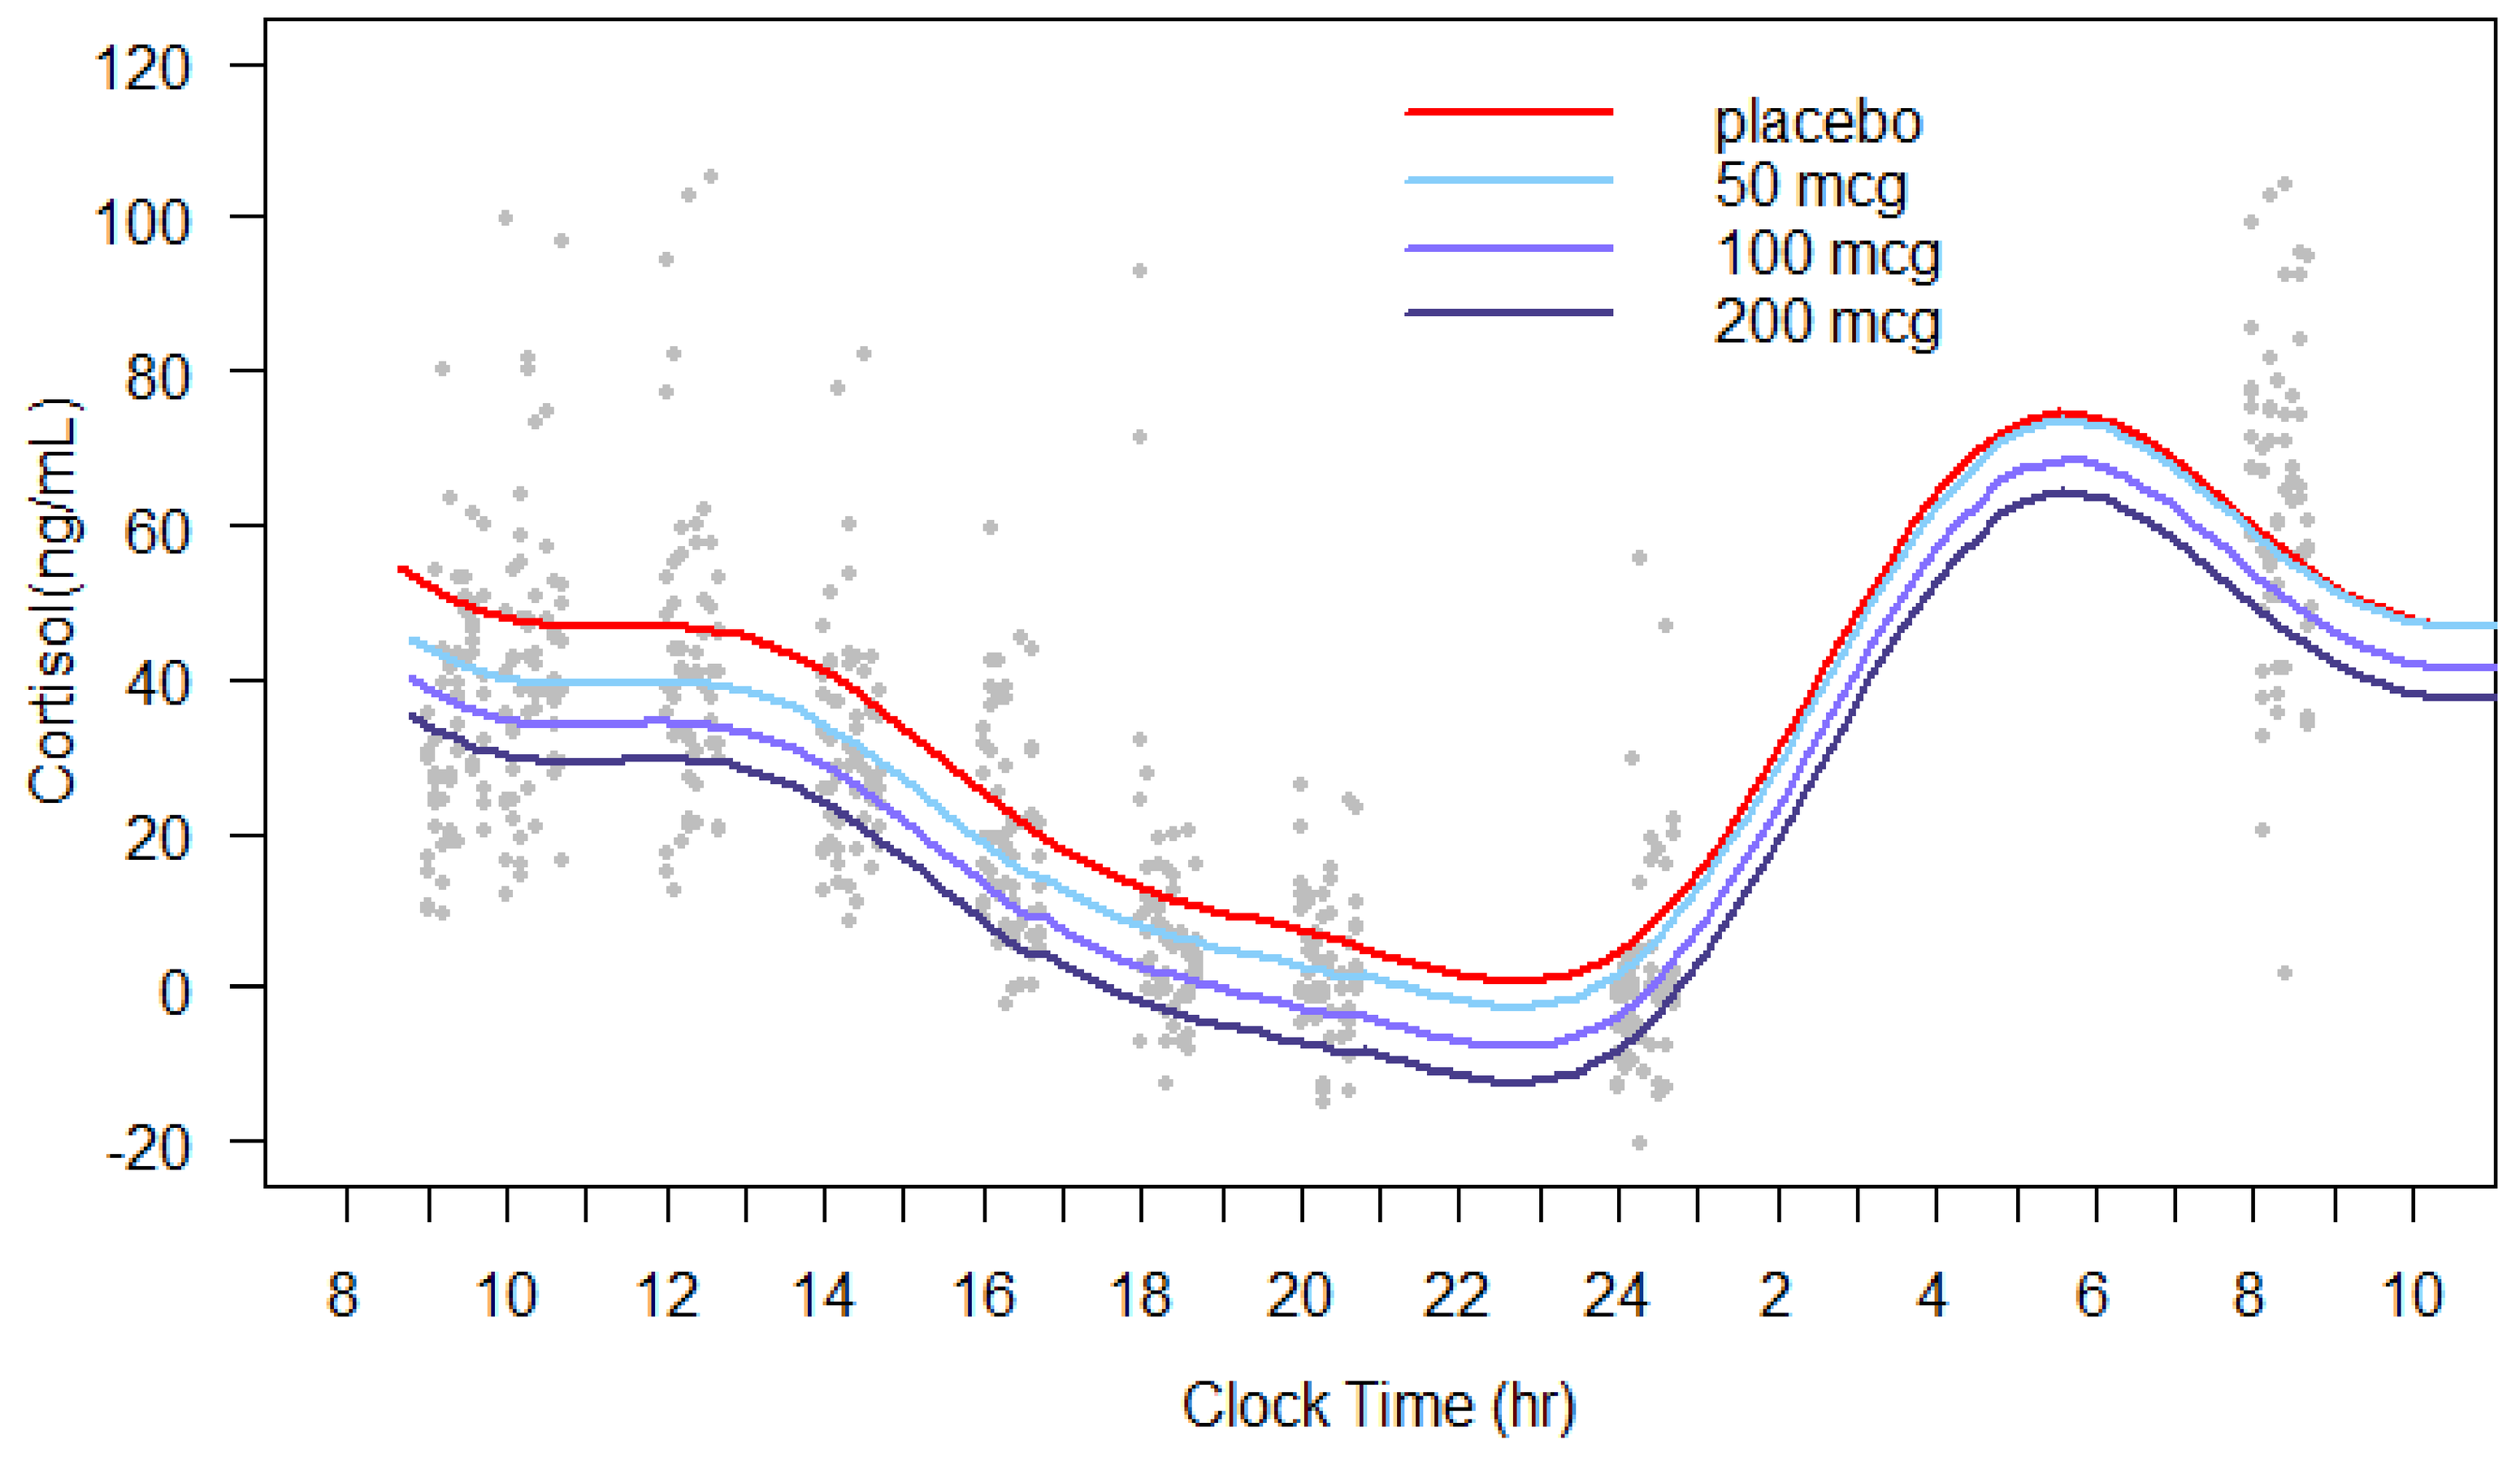
placebo
50 mcg
100 mcg
200 mcg
Cortisol(ng/mL)
Clock Time (hr)

Discussion

- Single and repeat dose administrations of FF/VI 50/25, 100/25 and 200/25 mcg were safe and well tolerated in healthy Chinese subjects.
- The cosine model with direct FF effect reasonably captured the circadian rhythm of cortisol in healthy male Chinese subjects.
- The estimated EC50 of FF on cortisol lowering is 99.4 pg/ml. The effect gradually diminishes as FF concentration in plasma decreases. The effect of FF on cortisol was also tested with a linear function (function b). The change in objective function value was not significant compared to the Emax model. Thus, the more mechanistic Emax model was selected.
- Although data supported a direct effect of FF concentration on cortisol in the final model, delay could not be entirely ruled out since assessments of FF concentrations were made only at steady state.

Created by: Digital Media Services
create | deliver | engage