A time-to-event model relating integrated craving to risk of smoking relapse across different nicotine replacement therapy formulations

UPPSALA UNIVERSITET

E Germovsek* (1), A Hansson (2), MO Karlsson (1), Å Westin (2), PA Soons (3), A Vermeulen (3), MC Kjellsson (1)

(1) Department of Pharmaceutical Biosciences, Uppsala University, Sweden; (2) McNeil AB, Helsingborg, Sweden; (3) Janssen R&D, a division of Janssen Pharmaceutica NV, Beerse, Belgium

*eva.germovsek@farmbio.uu.se

Background and objectives

Smoking causes over 7 million avoidable deaths per year [1]. Nicotine replacement therapy (NRT) facilitates smoking cessation [2] by reducing craving for nicotine. Our aim was to quantitatively investigate how a pharmacodynamic marker of long-term withdrawal symptom response (i.e. 'integrated' craving, meaning craving over a period of time) is linked to the hazard of smoking relapse.

Conclusions

- A time-to-event model was developed, where integrated craving was related to the hazard of smoking relapse in a sigmoidal E_{max} fashion.
- The model confirms the common understanding that higher craving is related to a lower probability of remaining abstinent from smoking.
- Integrated craving measured with 3 different pharmacodynamic scales was connected using a joint bounded integer model. This helped with the model development and also facilitates its use in simulations.

Results

The data included 9,323 (n active:placebo treatment arm = 6,415:2,908) adult subjects with median of 2 (range 0-99) previous attempts to quit smoking. 33% smoked their first cigarette 6-30 minutes after waking up, and 6% of subjects >60 minutes after waking up. In total, 9% of subjects (% active:placebo = 11:5) remained smoking abstinent until the end of the studies, with study lengths ranging from 3 weeks to 2 years.

A combination of a time-constant and Gompertz hazard proved best to describe the data, with hazard decreasing over time. Integrated craving was positively related with the hazard of relapsing (with a sigmoidal E_{max} function), and was included in the model as predicted craving Z-scores.

Equation 1 shows the structure of the model, and Table 1 and Figure 2 show numerical and graphical results. Plots showing how abstinence probability changes with time for different levels of craving are presented in Figure 3.

$$hazard = (constant + \lambda \cdot \exp(\alpha \cdot time)) \cdot \left(1 + \frac{E_{max} \cdot \exp(Zcraving)^{Hill}}{\exp(Zcraving)^{Hill} + \exp(Zcraving_{50})^{Hill}}\right) \cdot \exp(\theta_{active} \cdot ACTIVE), \quad \text{(Eq. 1)}$$

where λ is a scale parameter and α is a shape parameter in the Gompertz model, E_{max} is the maximal effect integrated craving can have on the hazard, *Z-craving*$_{50}$ is the integrated craving giving 50% of maximal hazard due to craving, *Hill* is the sigmoidicity parameter, *ACTIVE* is 1 for the NRT treatment arm and 0 for the placebo arm.

Table 1: Parameter estimates with uncertainty.

	Constant hazard (/day)	α	λ	E_{max}	Z-craving$_{50}$	Hill	θ_{active}
Mean (RSE)	0.0005 (7%)	-0.0127 (4%)	0.0085 (2%)	10.5 (2%)	-0.405 (6%)	3.37 (4%)	-0.361 (7%)

RSE is relative standard error obtained with NONMEM 7.4

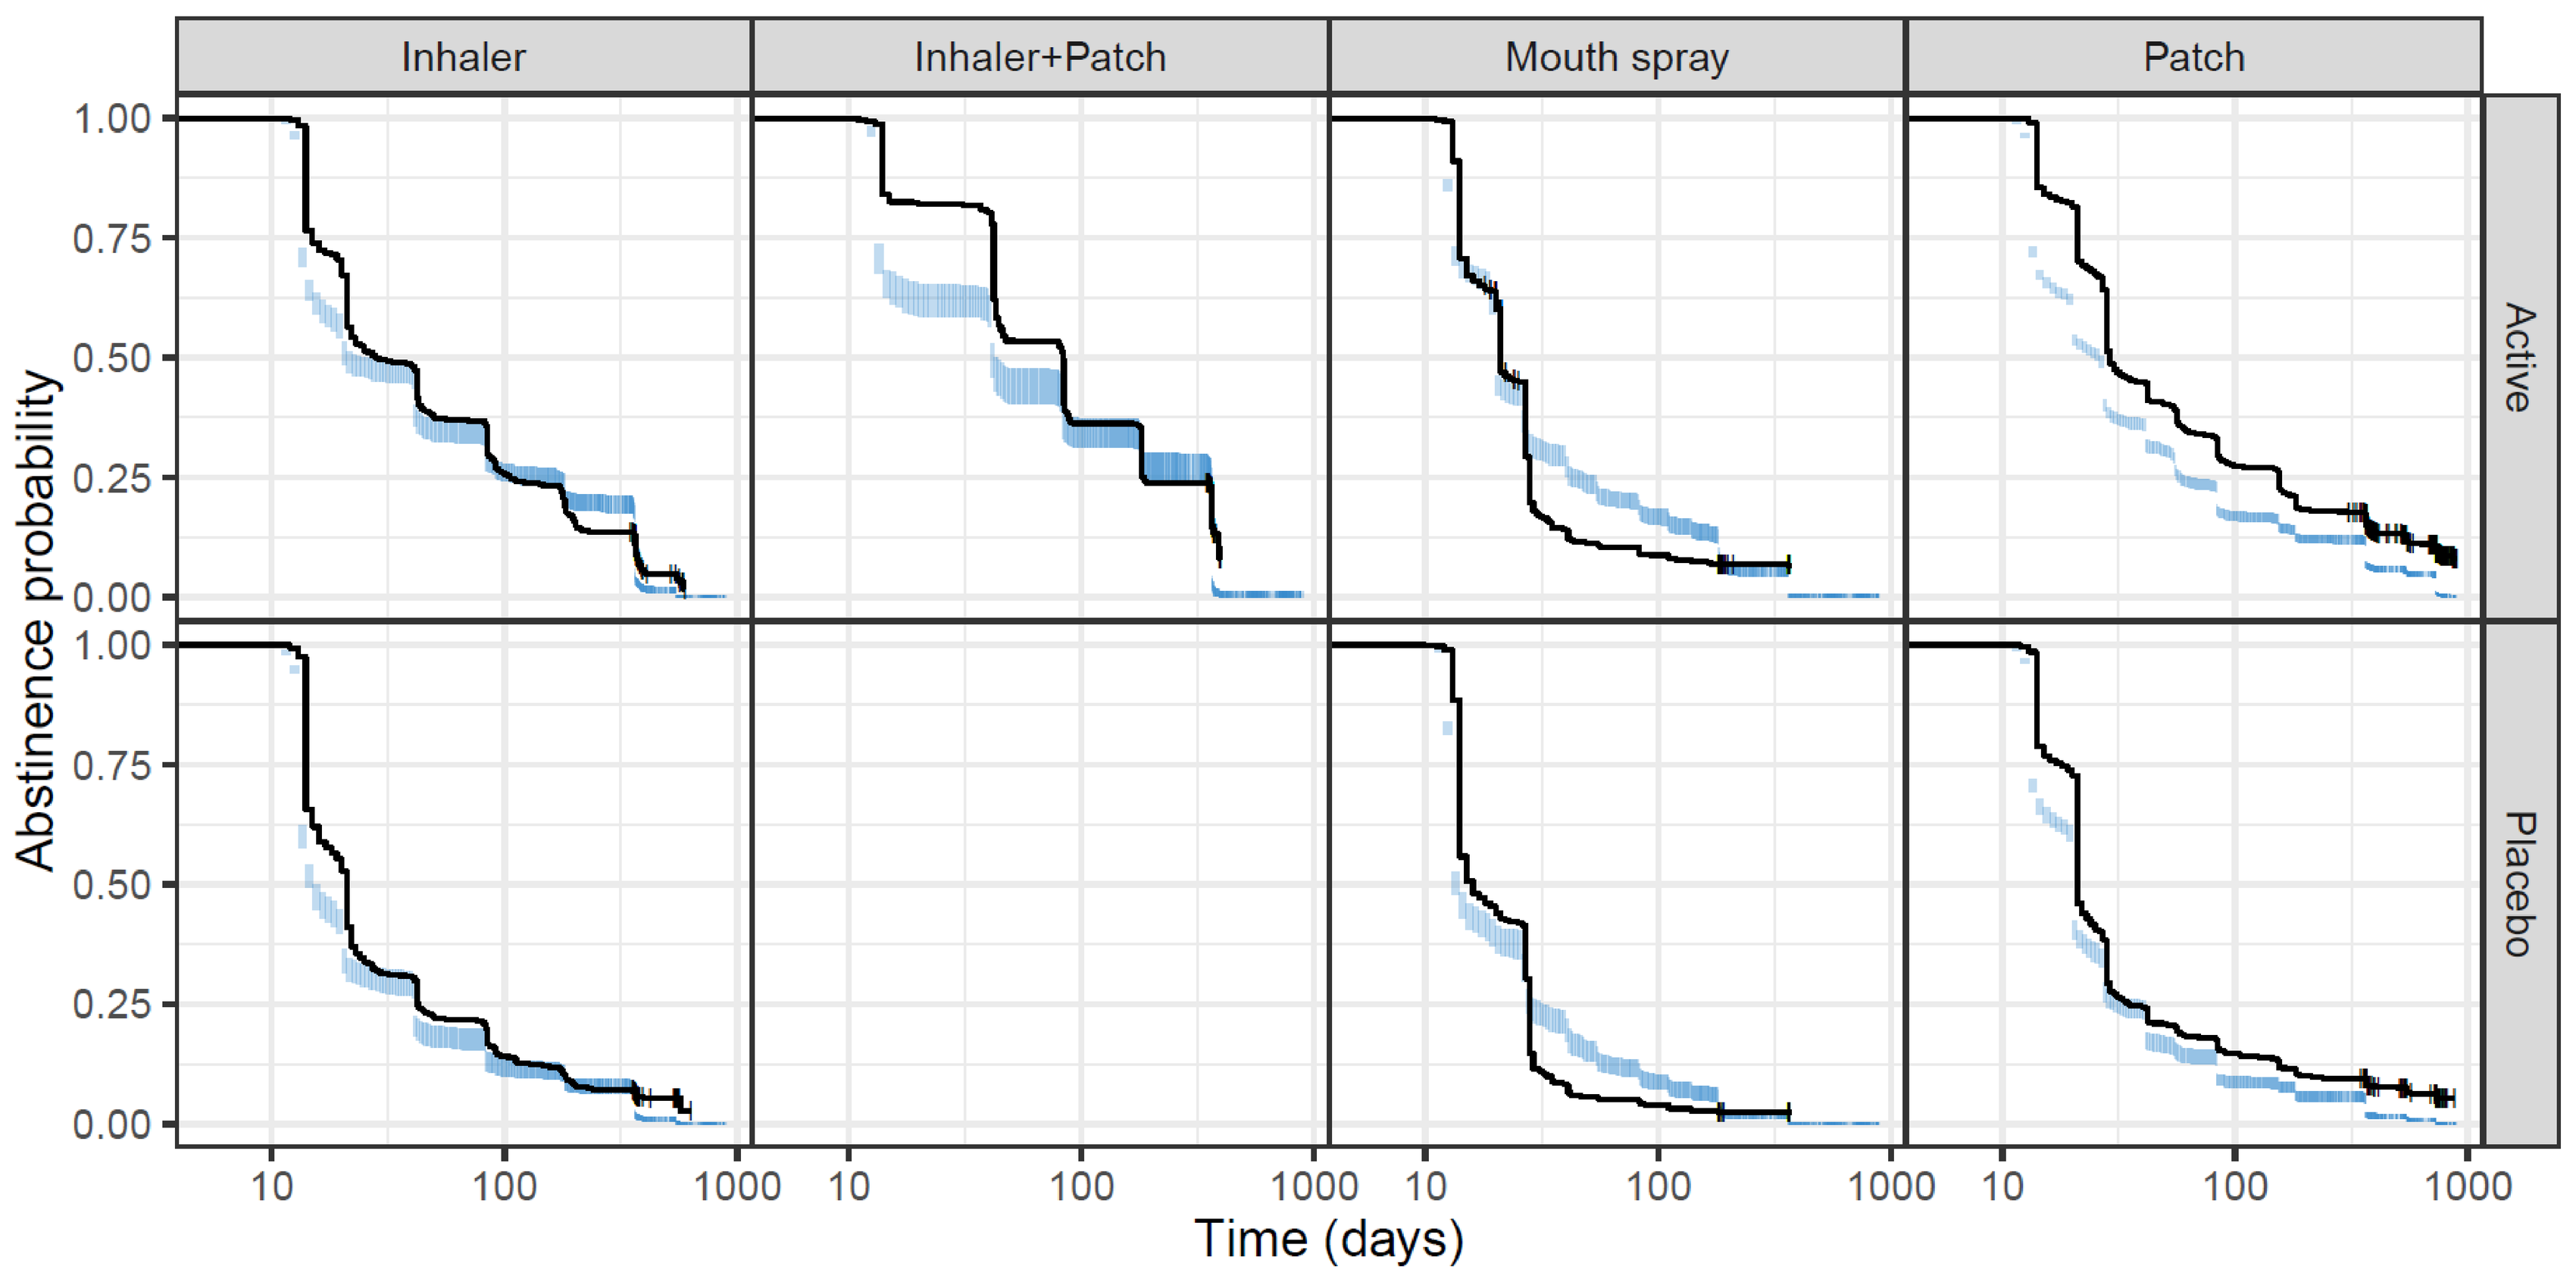
Inhaler
Inhaler+Patch
Mouth spray
Patch
Active
Placebo
Abstinence probability
1.00
0.75
0.50
0.25
0.00
10
100
1000
Time (days)

Figure 2: Kaplan-Meier visual predictive checks for different formulations, stratified by treatment arm. Black lines represent the observed abstinence probability and the areas are the corresponding 95% confidence intervals from 100 simulations. There was no placebo data for combination treatment with inhaler and patch.

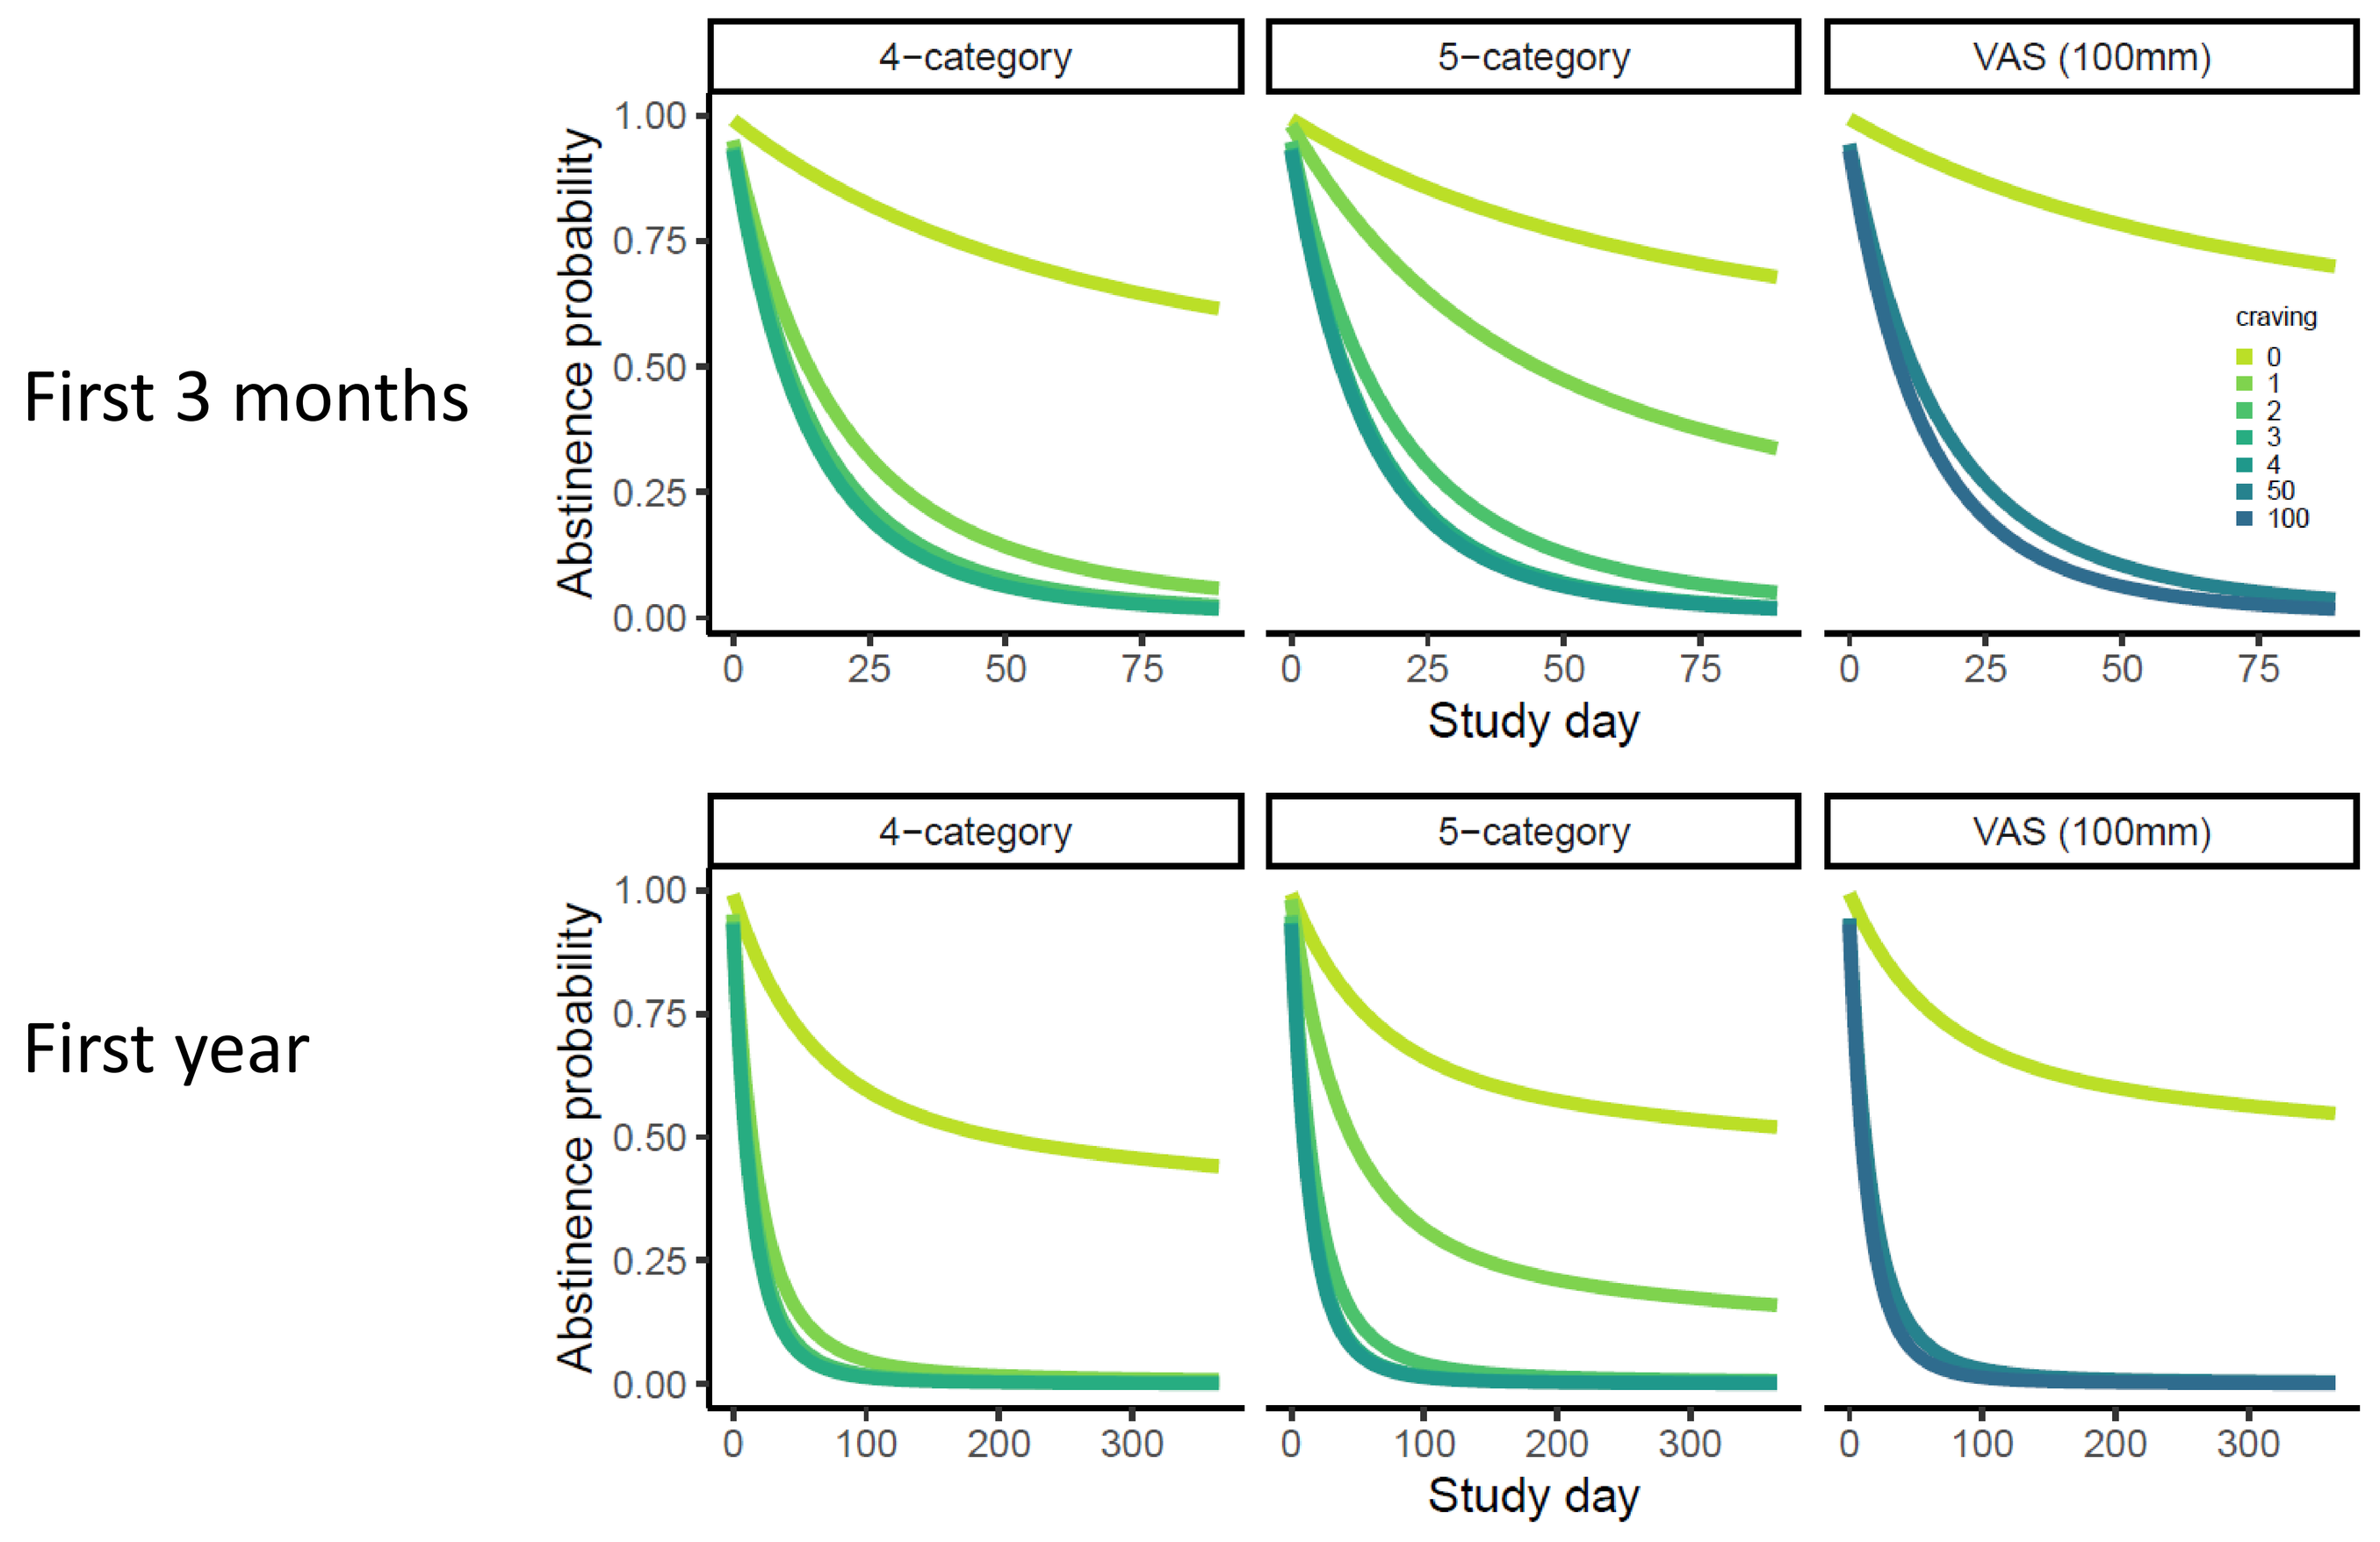
First 3 months
First year
4−category
5−category
VAS (100mm)
Abstinence probability
craving
0
1
2
3
4
50
100
Study day

Figure 3: Plots showing the relationship between craving and smoking abstinence probability based on the model estimates. Plots on the top are for the first 3 months and on the bottom for the first year in the active treatment arm, stratified by different craving scales. VAS is visual analogue scale.

Methods

Retrospective data that were available for analysis were collected in 19 separate studies, and included three NRT formulations, specifically: inhaler, mouth spray, or patch, and data from the combination use of inhaler and patch. Smokers, motivated to quit smoking, were included in the active (i.e. NRT) or the placebo treatment arm.

An event (i.e. relapse to smoking) was only assessed at a study visit, therefore the exact time of an event was unknown, and interval-censoring was used. Several time-constant and time-varying hazard functions were explored. A bounded integer model was used to connect integrated craving measured with different craving scales together (similarly as done in [4]). Different ways of relating predicted craving Z-score to hazard of relapse were investigated.

NONMEM 7.4 (ICON plc, Gaithersburg, MD, USA) with the Laplace approximation was used to obtain the likelihood.

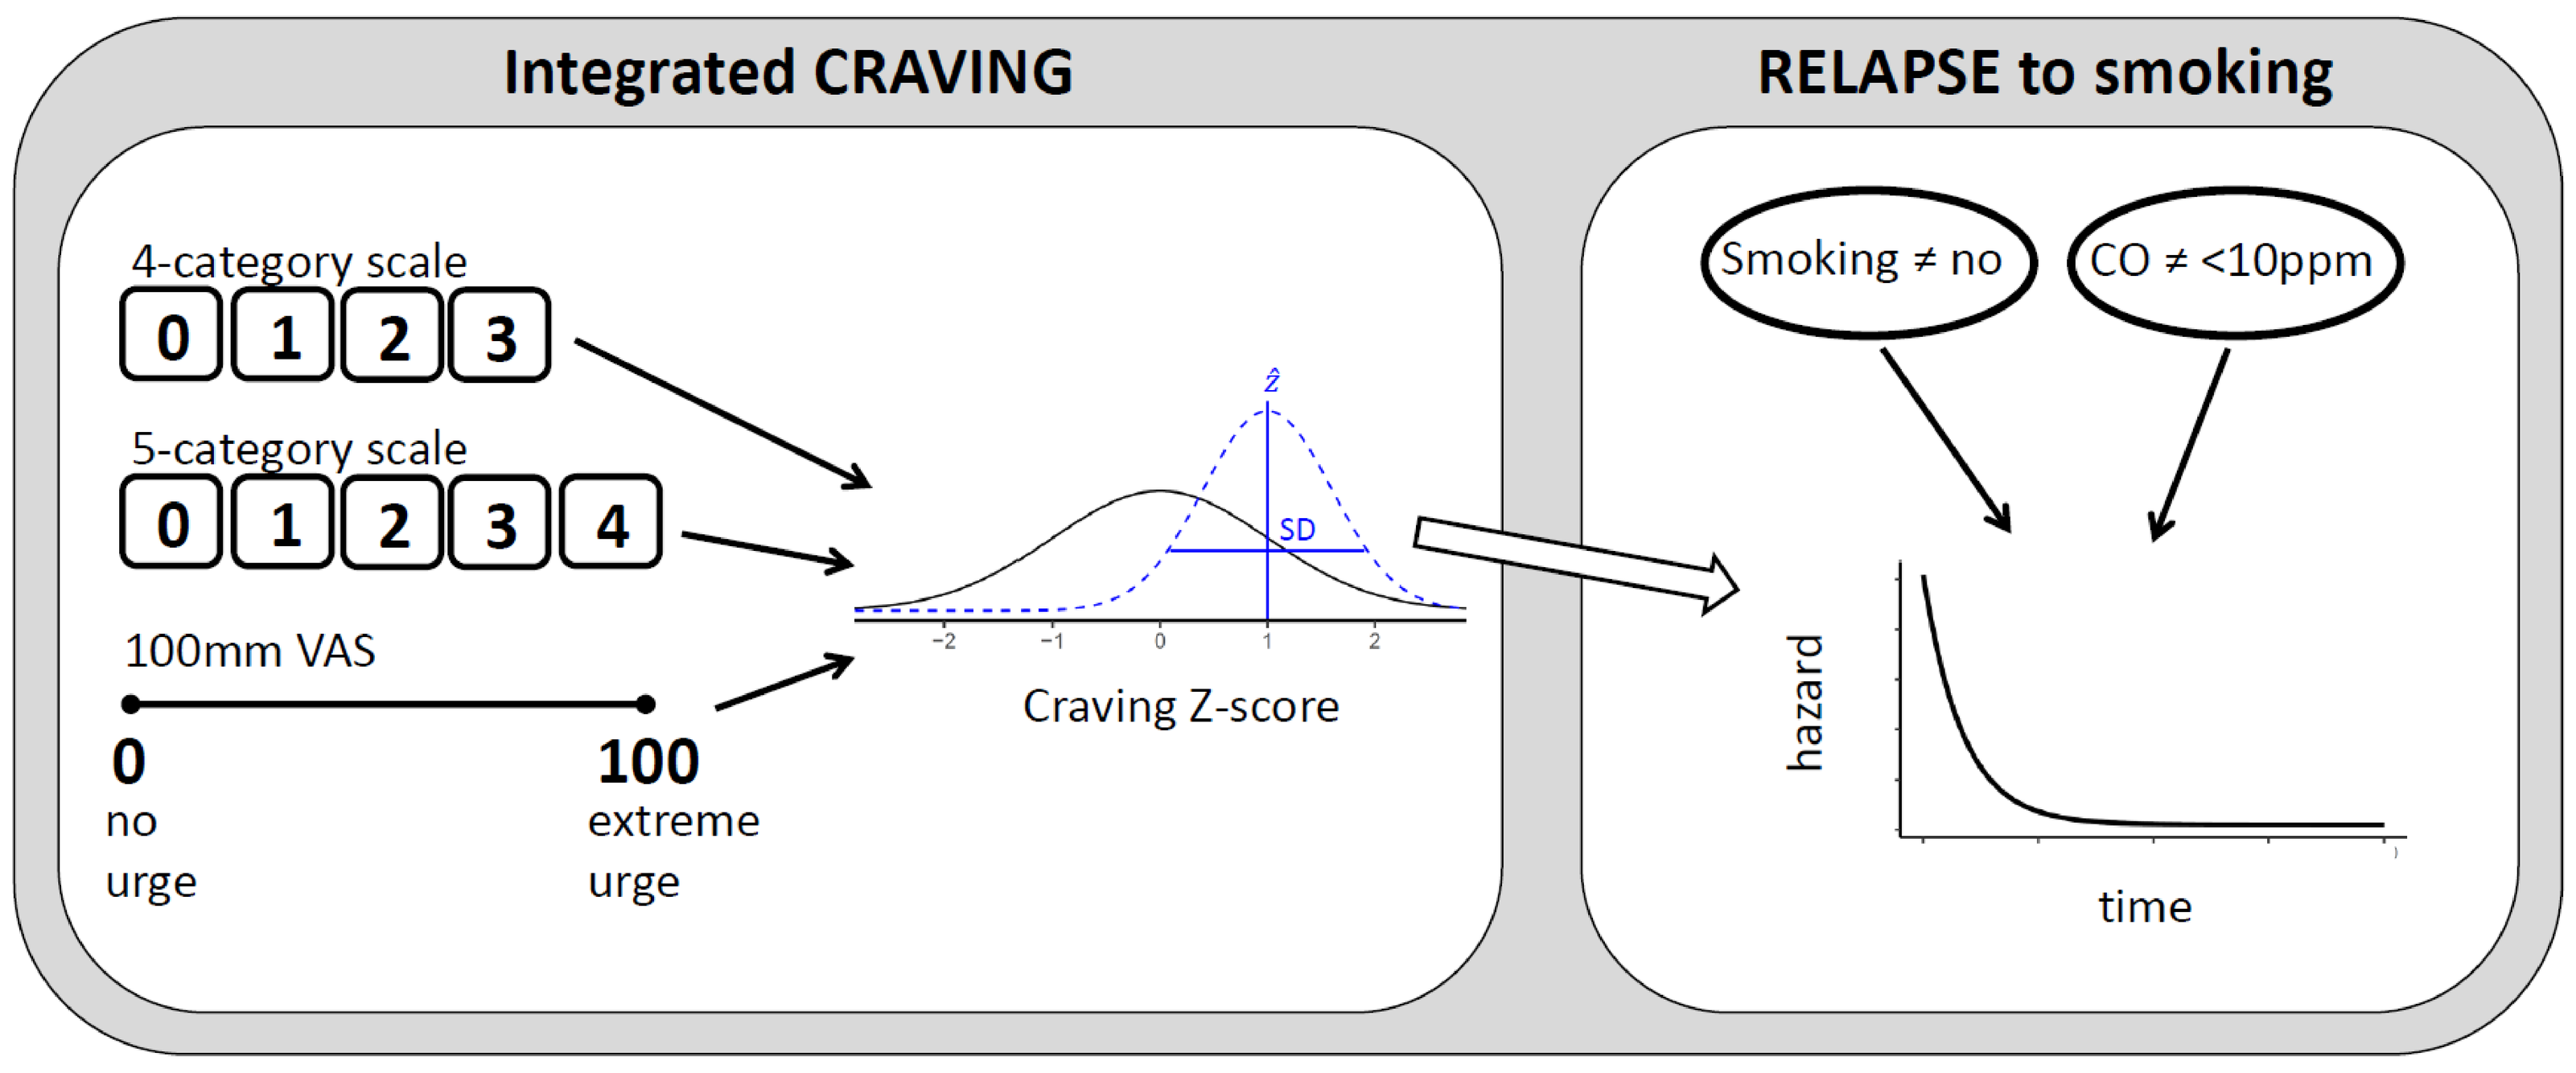
Integrated CRAVING
RELAPSE to smoking
4-category scale
0 1 2 3
5-category scale
0 1 2 3 4
100mm VAS
0
no
urge
100
extreme
urge
SD
Craving Z-score
Smoking ≠ no
CO ≠ <10ppm
hazard
time

Figure 1: Model schematic – integrated craving was assessed with three craving scales: a 4-category scale, a 5-category scale, and a 100mm visual analogue scale (VAS). Abstinence status was a binary response, and was modelled using a time-to-event model, where the event was a relapse to smoking (i.e. non-abstinence).

References
[1] GBD 2017 Risk Factor Collaborators. Global, regional, and national comparative risk assessment of 84 behavioural, environmental and occupational, and metabolic risks or clusters of risks for 195 countries and territories, 1990-2017: a systematic analysis for the Global Burden of Disease Study 2017. The Lancet, 2018; 392: 1923-1994.
[2] Benowitz, N.L., Clinical pharmacology of nicotine: implications for understanding, preventing, and treating tobacco addiction. Clin Pharmacol Ther, 2008; 83(4): 531-541.
[3] Hartmann-Boyce, J., Chepkin, S.C., Ye, W., Bullen, C. & Lancaster, T. Nicotine replacement therapy versus control for smoking cessation. Cochrane Database Syst Rev, 2018; 5: CD000146.
[4] Germovsek, E., Hansson, A., Kjellsson, M.C., Perez Ruixo, J.J., Westin, Å., Soons, P.A., Vermeulen, A., Karlsson, M.O., An exposure-response model relating nicotine plasma concentration to momentary craving across different nicotine replacement therapy formulations, PAGE 27 (2018) Abstr 8649 [www.page-meeting.org/?abstract=8649]

Disclosures: EG, MCK and MOK declare no conflicts of interest; AH, ÅW, PS and AV are (former) employees of subsidiary companies of Johnson & Johnson.

Johnson & Johnson
Apotekarsocieteten