Simulation Methodology for Quantitative Study Decision in a Dose Response Setting

Meg Bennetts

Global Pharmacometrics, Pfizer Limited, Sandwich, Kent, CT13 9NJ UK

OBJECTIVES

Tools & Methodology:

- Develop a framework for simulating study data based on current longitudinal models using R and NONMEM® 6.
- Obtain a table of Trial Performance Metrics that quantify study success in terms of how many times a correct decision is made.

Clinical Study:

- Quantify the expected performance of a designed Phase 2b dose-ranging study and probable Phase III dose by evaluating Decision Criteria for FEV_1 and Heart Rate endpoints. The aims of the study were to determine the dose-response relationship of efficacy and safety in moderate asthmatics for a novel drug dosed over 4 weeks.

BACKGROUND

Definitions:

- Truth: The true treatment effect Δ is unknown, but can be estimated from the predictive distribution of Δ, for a given model fitting the current state of knowledge about the drug effect, integrated across fixed and random effects
- Trial: A trial estimate of Δ can be estimated using the formal prospectively defined study data analysis methods. [1, 2, 3]

Study Design:

- The designed, 4 week, study consisted of 5 treatments (three dose levels of novel drug, placebo and active control) with 80 subjects/treatment group. Longitudinal FEV_1, PK and Heart Rate endpoints were to be measured on days 1 and 29.

Models:

- The NONMEM® Models for the endpoints were based on previous studies in the drug program.[4]
 - FEV_1 was a KPD model using a sigmoidal E_{max} PD component with baseline and circadian variation.
 - Heart Rate consisted of a PK model with complex absorption profile and high accumulation, and an E_{max} PD model including covariates for gender, baseline and circadian variation.

SIMULATION METODOLOGY

R: Generate Model Parameters: ①

- NONMEM® Control File Read into R.
- Parameters drawn from a multivariate normal distribution using the MSToolkit[5]
 - Centred on maximum likelihood estimates from the model fit
 - Variance-covariance matrix from the model fit
 - Acceptable ranges for parameters, maximum number of draws to achieve these ranges and number of digits for drawn parameters specified.
- Each draw of the resulting model parameters substituted into the $THETA section of the NONMEM® control file to create corresponding simulation replicate control file.
- $ESTIMATE substituted with $SIMULATE in the replicate control file together with replicate specific FILE names.

NONMEM® : Simulations

- NONMEM® called from R (within loop).
- Truth Simulations ②
 - Variability on parameter uncertainty but no Inter-occasion, inter-individual, circadian or residual variability
 - Within each replicate 1 patient per dose.
- Trial Simulations ③
 - Each paired with its corresponding truth replicate using identical parameter estimates
 - Within each replicate 80 patients per dose
 - Inter-occasion, inter-individual, circadian variation and residual variability as estimated in the model.

R: Statistical Analysis

- NONMEM® output table of longitudinal FEV_1 read into R for each Truth/Trial replicate.
- Protocol defined Trough FEV_1 endpoint derived for Day 29. ④
- Analysis of Trial Replicates to examine the properties of the planned Analyses and to check the sensitivity of the design from Active Control results ⑤

R: Success/Failure Criteria ⑥

- Apply relevant predefined success criteria to each Truth/Trial replicate
 - For Truth this will be a simple calculation of whether the endpoint has passed the Target Value
 - For Trial the success criteria will be based on the results of the analysis such as statistical significance.

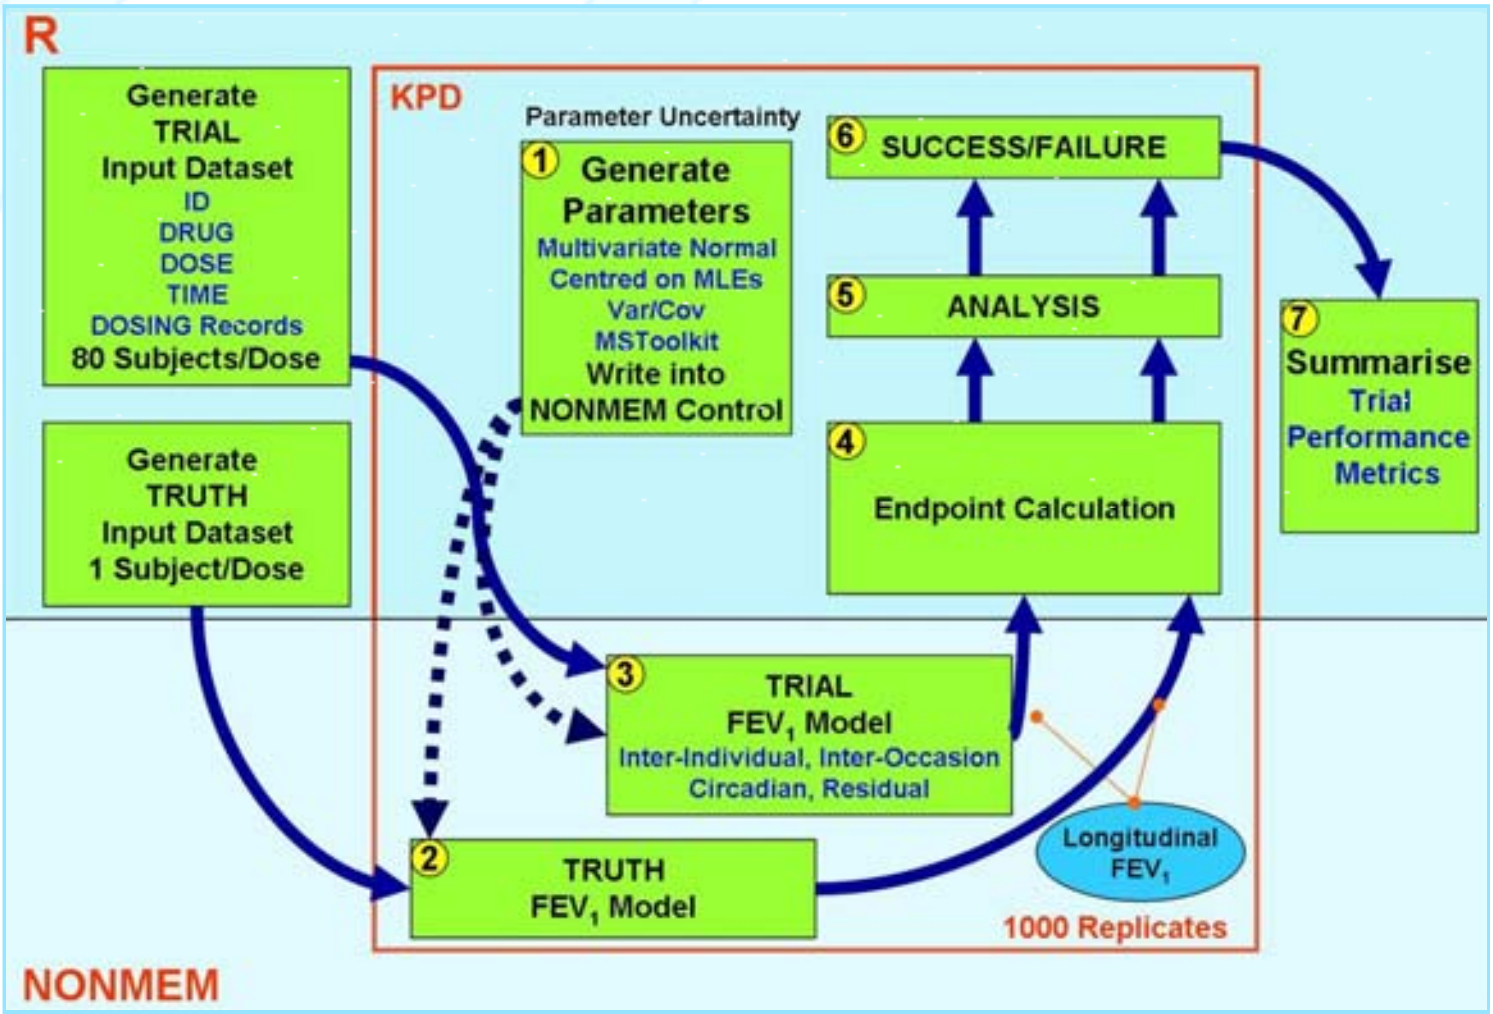
R
Generate TRIAL Input Dataset
ID
DRUG
DOSE
TIME
DOSING Records
80 Subjects/Dose
Generate TRUTH Input Dataset
1 Subject/Dose
KPD
Parameter Uncertainty
① Generate Parameters
Multivariate Normal
Centred on MLEs
Var/Cov
MSToolkit
Write into NONMEM Control
⑥ SUCCESS/FAILURE
⑤ ANALYSIS
④ Endpoint Calculation
⑦ Summarise Trial Performance Metrics
③ TRIAL FEV1 Model
Inter-Individual, Inter-Occasion
Circadian, Residual
② TRUTH FEV1 Model
Longitudinal FEV1
1000 Replicates
NONMEM

CONCLUSIONS: Tools & Methodology

- A workable fit for purpose batch process was developed in R to: sample parameter uncertainty; perform replicate NONMEM® simulations of complex models; derive paired estimates of True and Trial effects; perform statistical analysis of replicate data; apply study success criteria, and summarise across replicates to produce Trial Performance Metrics.

TRIAL PERFORMANCE METRICS IN A DOSE RESPONSE SETTING

Classic 2x2 Contingency Table:

- In the current literature [1,2,3], success is defined at the study level in terms of a go or no go decision.
- A 2x2 contingency table is constructed to compare trial and truth decisions to understand false positive and false negative tradeoffs and to show the proportion of simulated trials where a correct decision and success occur.

Extending this Classic Table for Dose Response Studies: ⑦

- In addition to quantifying the overall study probabilities, in a dose response design, extra granularity can be added by quantifying whether the correct dose would be selected. This is achieved by the following steps:
 - Construct a NxN contingency table, where N is the number of treatments/doses
 - Define the location of treatments/doses with respects to study level go/no go categories
 - E.g. If a dose is commercially unattractive it may be considered a no go even if successful
 - In the analysis employ a step down approach to identify the minimum treatment/dose that meets the success criteria
 - Populate the table with the proportion of Truth/Trial successes at each minimum dose level.

			Trial					
	Minimum Dose		No Go		Go			
			No Dose	Control Dose	High Dose	Medium Dose	Low Dose	Total
Truth	No Go	No Dose	A			B		
		Control Dose						
	Go	High Dose						
		Medium Dose	C			D		
		Low Dose						
		Total	X				Y	100%

Example Trial Performance Metrics

At The Study Level:

- **A** (sum 4 boxes) = Probability of a Correct No Go
- **B** (sum 6 boxes) = Probability of a Incorrect Go
- **C** (sum 6 boxes) = Probability of a Incorrect No Go
- **D** (sum 9 boxes) = Probability of a Correct Go
- (sum 3 boxes) = Probability of Technical Success (At least one dose meets the Truth success criteria)
- (sum 3 boxes) = Probability of a Go Decision (At least one dose meets the Trial success criteria)
- **A + D** = Probability of a Correct Decision

At The Dose Level:

- – Trial Identifies Correct Minimum Dose
- – Trial Minimum Dose Too High
- – Trial Minimum Dose Too Low
- Sum = Probability of Trial replicates correctly identifying the minimum dose with respect to the corresponding Truth Replicate
- Max = Technically the minimum dose that meets the Truth success criteria
- Sum in **D** = Probability of Correct Go Decision but with too low a dose
- Sum in **D** = Probability of Correct Go Decision but with too high a dose
- **X** = Probability that the Trial fails to identify Control as an effective treatment. Including Control in the step-down analysis tests study sensitivity
- **Y** = Probability that the Trial fails to identify a minimally effective dose.

CONCLUSIONS: Clinical Study

- Assessing and comparing the properties of each set of Trial Performance Metrics produced for FEV_1 and HR indicated that the study was likely to be successful with respect to superiority to placebo but highlighted potential issues regarding power, success criteria, comparisons with Control and model assumptions.
- This work enabled the study team to make quantitative decisions regarding choice of study Decision Criteria, analysis methods and probable doses to manufacture for Phase III.

REFERENCES

1. Lalonde, R.L., et al. “Model-Based Drug Development”. Clin Pharm Ther 2007;82:21-32.
2. Kenneth G. Kowalski, Jonathan L. French, Mike K. Smith, Matthew M. Hutmacher. “A Model-Based Framework for Quantitative Decision-Making in Drug Development”. ACoP, Tucson, AZ. March 2008.
3. Kowalski, K.G., Ewy, W., Hutmacher, M.M., Miller, R., and Krishnaswami, S. “Model-Based Drug Development – A New Paradigm for Efficient Drug Development”. Biopharmaceutical Report 2007;15:2-22.
4. Jakob Ribbing, Gai Ling Li, Erno Van Schaick, Lutz Harnisch. “Assessing the therapeutic benefit of a new inhaled QD LABA through population PK-safety & efficacy modelling “. ERS, Vienna, Austria. September 2009.
5. Mike K Smith, Richard Pugh, Romain Francois, Jonathan French. “MSToolkit – An R library for simulating and evaluating clinical trial designs and scenarios”. ACoP, Mashantucket, CT. October 2009.

ACKNOWLEDGEMENTS

The author would like to thank Mike K Smith (Pfizer), Byron Jones (Pfizer), Jakob Ribbing (Pfizer) and Erno Van Schaick (Exprimo NV) for their models, valuable contributions and advice.