Landscape on technical and conceptual requirements in Drug/Disease Modelling & Simulation

G. Vlasakakis (1), E. Comets (2), A. Drescher (6), I. Gueorguieva (3), P. Magni (4), N. Terranova (4), O. Della Pasqua (1), E.C. de Lange (5), C. Kloft (6).

(1) GSK, Clinical Pharmacology Modelling & Simulation, Stockley Park, UK; (2) INSERM, UMR 738 Univ Paris Diderot, Sorbonne Paris Cité, Paris, France; (3) Lilly UK, Global PK/PD Department, Erl Wood Manor, Windlesham, Surrey, UK; (4) Dipartimento di Ingegneria Industriale e dell'Informazione, Università degli Studi di Pavia, Pavia, Italy; (5) Leiden University, Leiden, The Netherlands; (6) Freie Universitaet Berlin, Dept. of Clinical Pharmacy and Biochemistry, Berlin, Germany.

Background and Objectives

Concepts of **Drug/Disease Modelling and Simulation (DD M&S)** have internationally gained attractiveness and importance in drug development (known as **model-based drug development**) as well as in therapeutic use (to be named as **model-based patient care**). However, the **role, impact, benefit, weakness and potential** of DD M&S activities provided by different stakeholders involved in this process have not been explored yet. Additionally, questions of what is required and currently available in terms of technical and conceptual skills and competencies have rarely been addressed.

The aim of this project was therefore to perform a survey to collect individual feedback from the DD M&S community:

- to better understand the **current technical and conceptual requirements** for DD M&S and
- to create a landscape including the **identification of core competences as well as opportunities and weaknesses** to be considered as topics for training and education purposes.

Materials & Methods

Target audience and distribution

- Online survey using the SurveyMonkey system open between Nov 2011 and Jan 2012
- Target audience: **'Modellers'** - develop models/perform DD M&S activities,
 'Appliers' - apply/interpret results from DD M&S activities
 'Reviewers' - review DD M&S results
 'Multi' - are involved with all of the above DD M&S activities
- Distribution to /via members of the IMI DDMoRe project consortium (i.e. internal and external)

Structure and design:

- 5 domains: I. Impact and benefit of DD M&S, II. Area of involvement/engagement, III. Technical use/skills and methodological concepts, IV. Gaps & Challenges, V. Willingness to share data/models for training & education (T&E) purposes
- Question types: Single/multiple choice; weighted answers; free text Q

Data analysis:

- Graphical and descriptive statistical evaluation using R and Excel MS
- Data stratification according to type of organisation, level of experience and DD M&S activity

Results

Data base

- In total n= 152 responses returned
- After database cleaning with removal of n=25 responses

➔ **n= 137** responses for data analysis

Level of experience

- 64% senior vs. 36% junior positions (i.e. PhD, PostD)

Type of organisation

- Academia n=74
- Pharma Industry n=59
- SME n=4

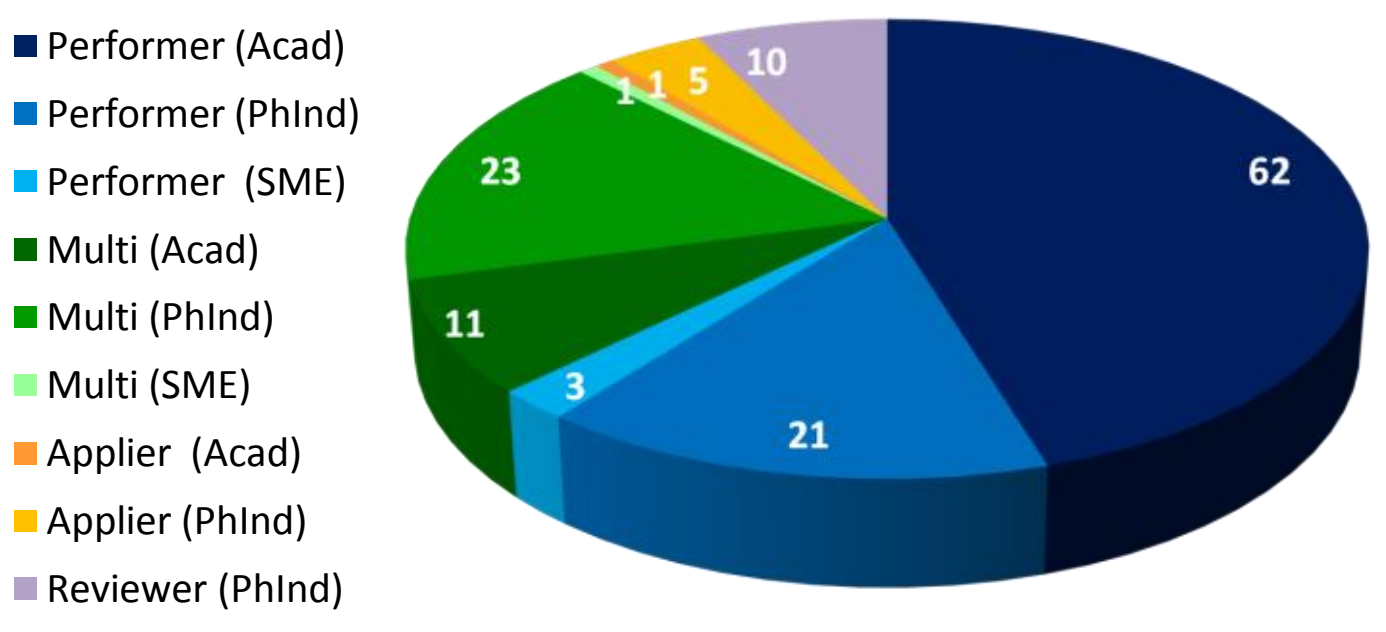

Performer (Acad)
Performer (PhInd)
Performer (SME)
Multi (Acad)
Multi (PhInd)
Multi (SME)
Applier (Acad)
Applier (PhInd)
Reviewer (PhInd)
62
21
3
11
23
1
1
5
10

Fig. 1: Pie chart of ***primary area in DD M&S*** (n=137)
Acad: academia, PhInd: Pharma industry

Based on the distribution in Fig. 1, the results in Domains I – V were:

- Split into subgroups of **'Modellers'** and **'Multi'** (due to few responders in other groups) representing **n=121** (88%) of responders;
- SME (n=4) was merged with Pharm. Industry ('**Pharma**')

↳ **'Academia'** n= 73 vs. '**Pharma'** n=48 ➔ i.e. **60%** : **40%**

Domain I. "Impact and benefit of DD M&S"

Impact of DD M&S in organisation (Fig. 2)

Major impacts for Academia:

- Methodological development
- Understanding of system characteristics

Major impacts for Pharma:

- Compound development (understand drug characteristics)
- Decision making (eg. dose selection)

➔ For all: **potential for increased impact**

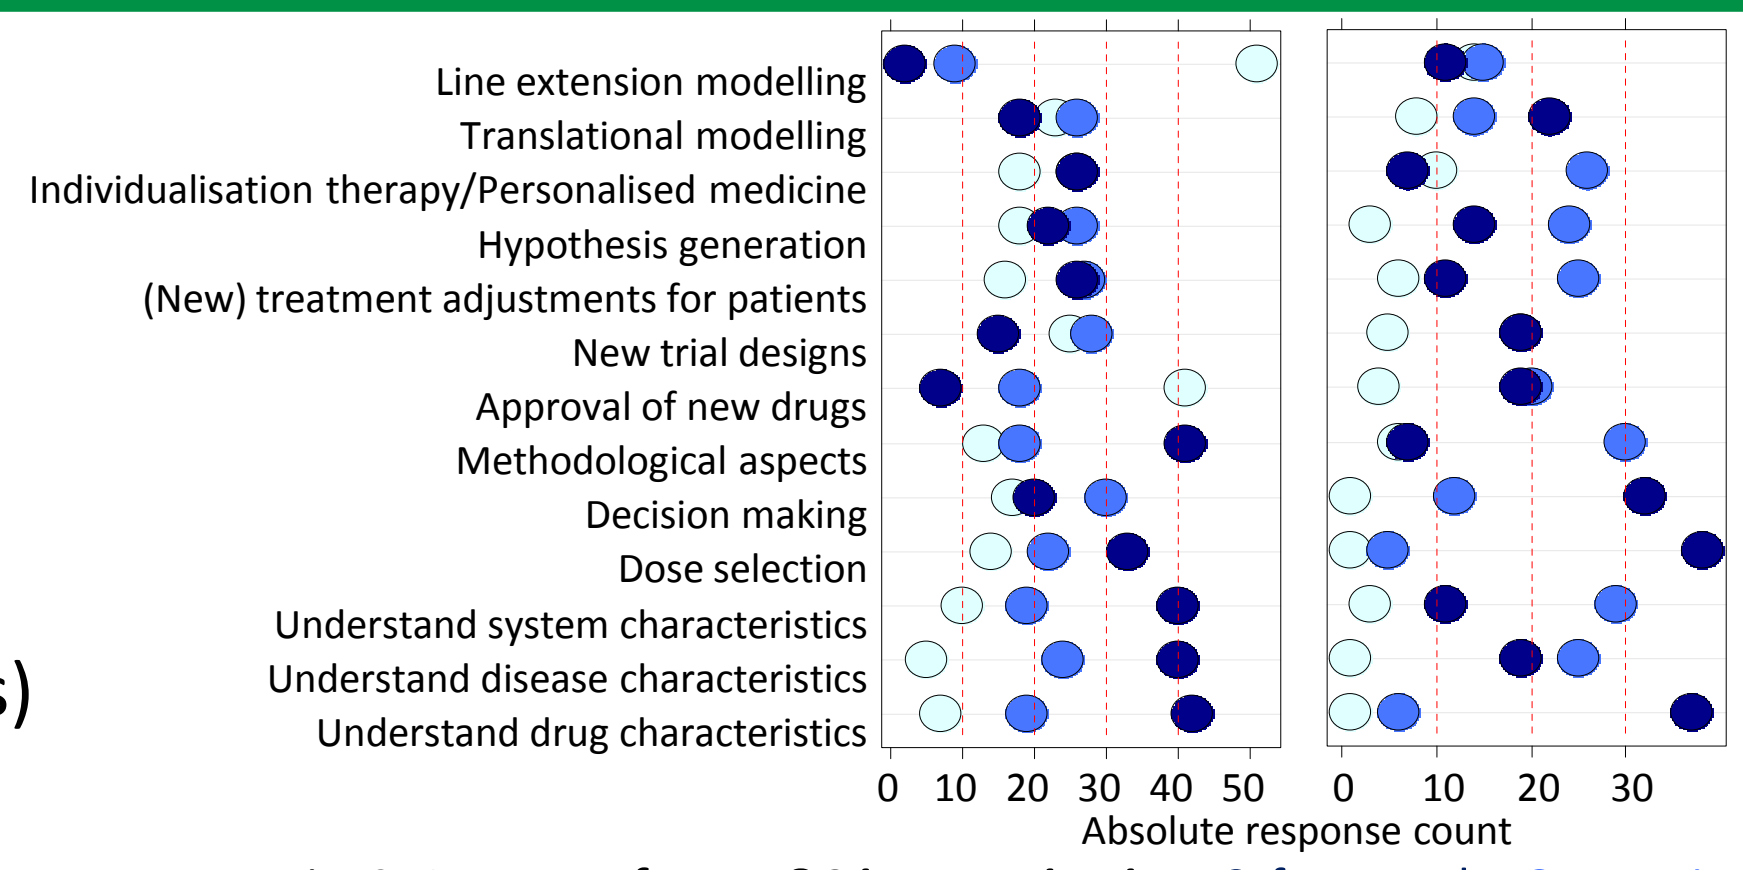

Line extension modelling
Translational modelling
Individualisation therapy/Personalised medicine
Hypothesis generation
(New) treatment adjustments for patients
New trial designs
Approval of new drugs
Methodological aspects
Decision making
Dose selection
Understand system characteristics
Understand disease characteristics
Understand drug characteristics
0 10 20 30 40 50
0 10 20 30
Absolute response count

Fig. 2: ***Impact of DD M&S in organisation:*** ● frequently, ● occasionally, ❍ never; left: Academia (n=62-72), right: Pharma (n=40-45)

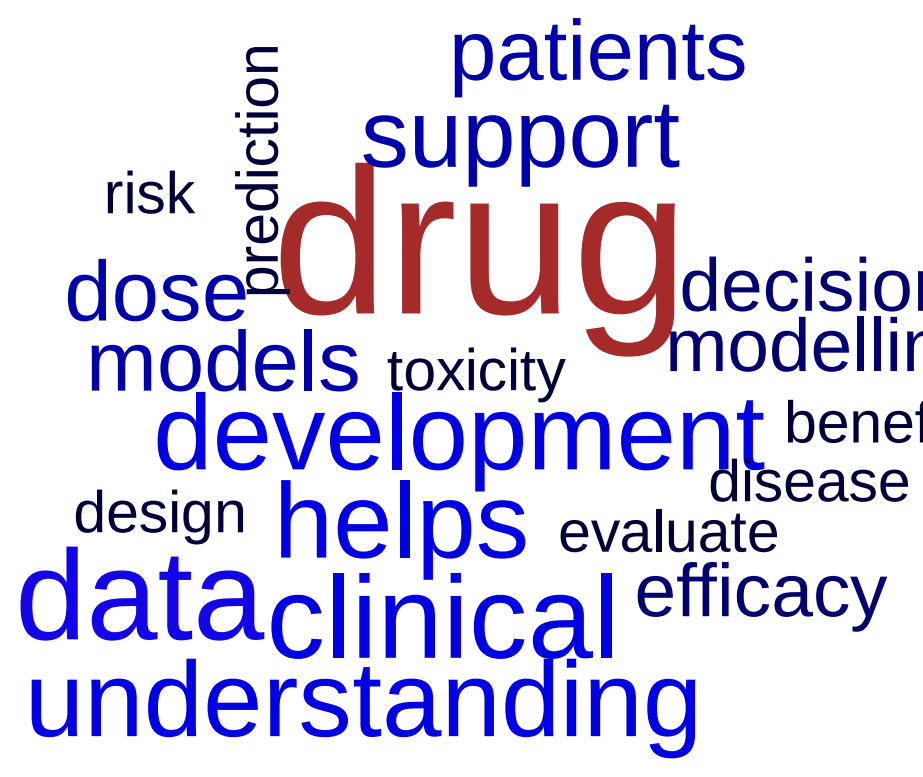

patients
support
risk
prediction
drug
dose
decision
modelling
models
toxicity
development
benefit
disease
design
helps
evaluate
data
clinical
efficacy
understanding

Fig. 3: ***Benefit of DD M&S in organisation (n**=48)*

Benefit of DD M&S in organisation (Fig. 3)

M&S is seen primarily as a tool for

- Understanding drug development
- Using clinical drug data and models
- Supporting drug development and patients' drug dose/dosing

Domain II. "Area of involvement/engagement"

Area of involvement (Fig. 4)

- Academia scored highest for acad. research, (pre-)clinical development and optimisation of therapeutic use
- Pharma scored highest for (pre-)clinical development
- Important differences between for therapeutic use: higher involvement of Academia than Pharma
- Involvement in regulatory authorities: very low for both groups

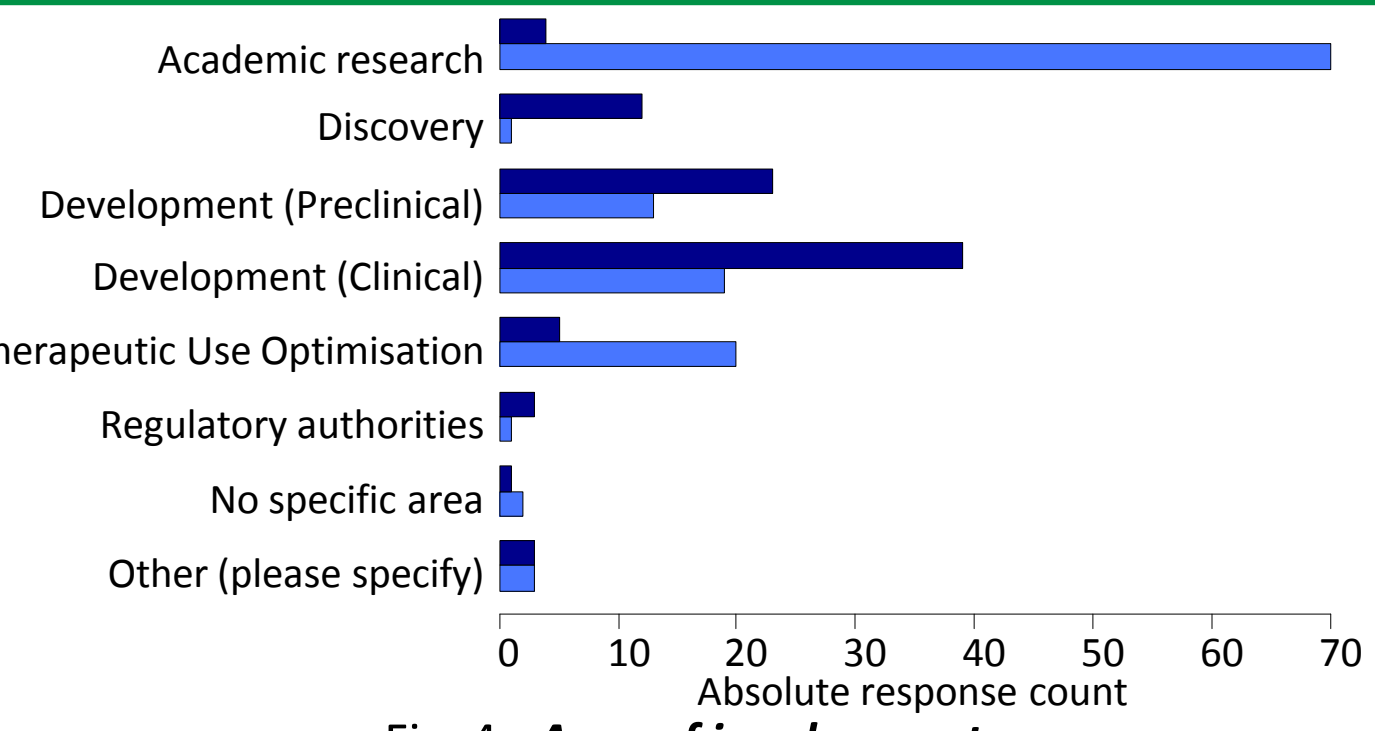

Academic research
Discovery
Development (Preclinical)
Development (Clinical)
Therapeutic Use Optimisation
Regulatory authorities
No specific area
Other (please specify)
0 10 20 30 40 50 60 70
Absolute response count

Fig. 4: ***Area of involvement:***
■ Academia (n=129), ■ Pharma (n=90)

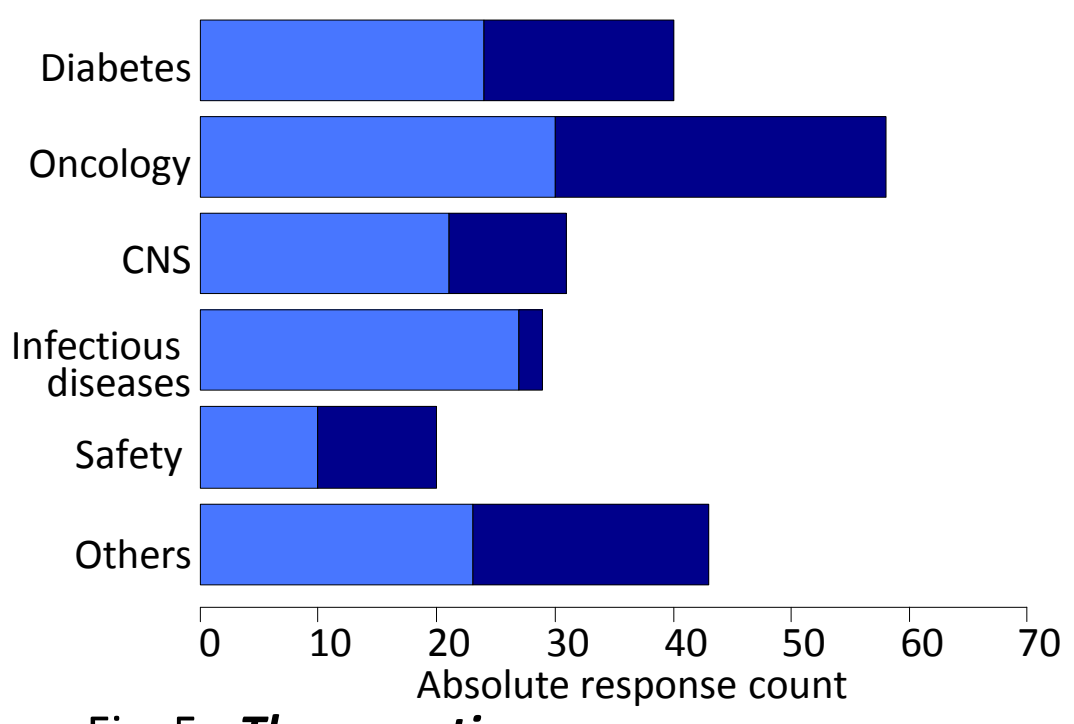

Diabetes
Oncology
CNS
Infectious diseases
Safety
Others
0 10 20 30 40 50 60 70
Absolute response count

Fig. 5: ***Therapeutic area:***
■ Academia (n=135), ■ Pharma (n=86)

Therapeutic area (Fig. 5)

- Equal responses count for Oncology, Diabetes, Safety and CNS of Academia and Pharma.
- An important difference is found for infectious diseases: much higher involvement of Academia than Pharma

Domain III. "Technical use/skills and methodological concepts"

Technical aspects (Fig. 6)

- List of software used by responders: both general statistical software (R, SAS) and specialised programs (NONMEM, Monolix)
- Few responses from the community focusing on systems biology, although they were included in the target audience
- Estimation methods most often cited (additional data): individual regression (used occasionally or frequently by 80%; in population analysis, linearisation-based method FOCE 84%)

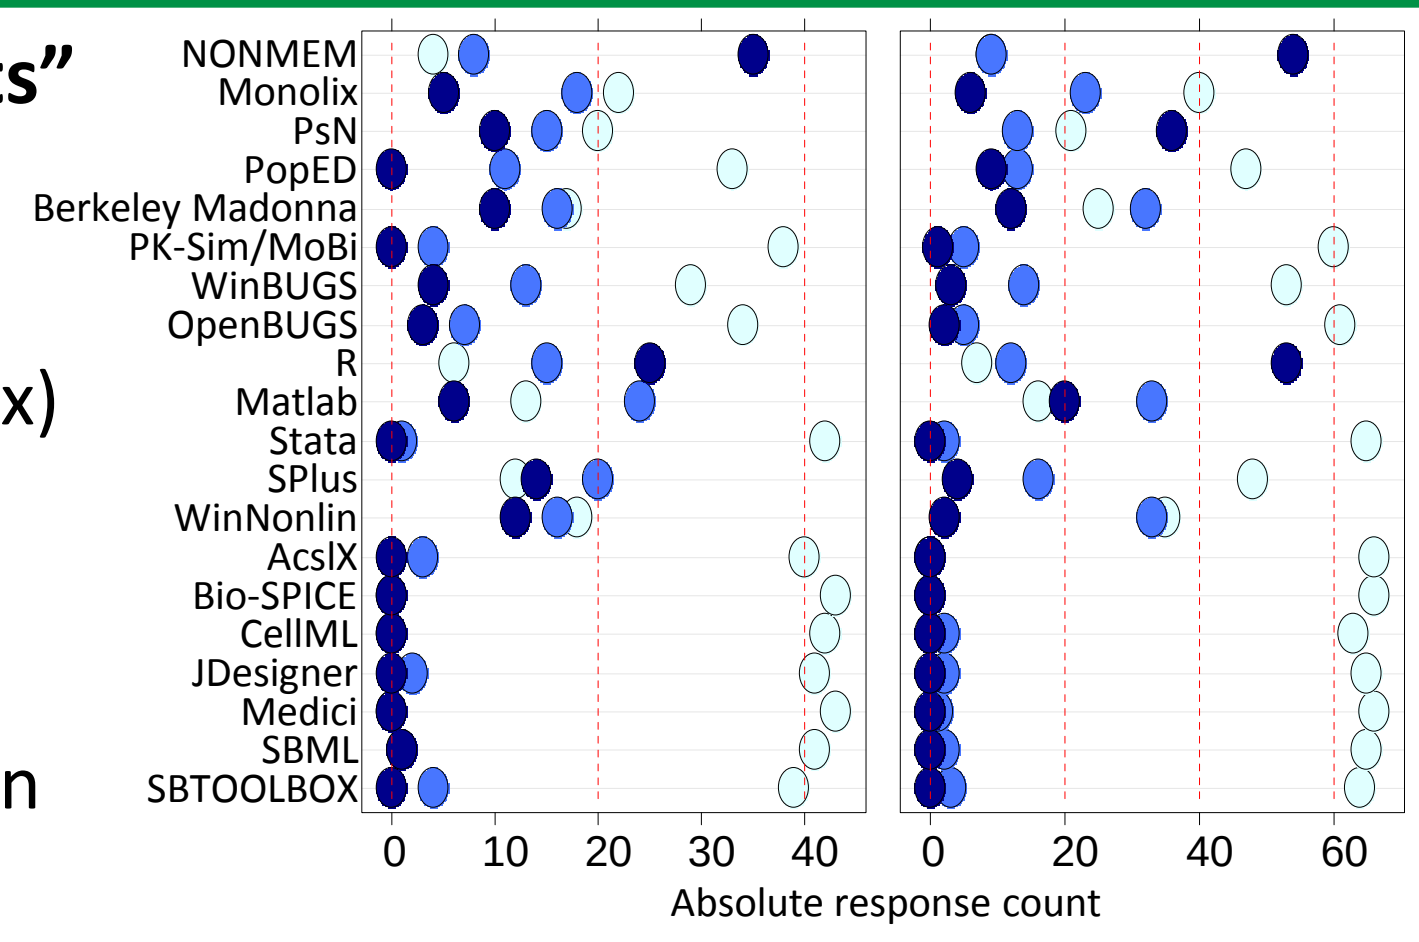

NONMEM
Monolix
PsN
PopED
Berkeley Madonna
PK-Sim/MoBi
WinBUGS
OpenBUGS
R
Matlab
Stata
SPlus
WinNonlin
AcslX
Bio-SPICE
CellML
JDesigner
Medici
SBML
SBTOOLBOX
0 10 20 30 40
0 20 40 60
Absolute response count

Fig. 6: ***Technical aspects:*** ● frequently, ● occasionally, ❍ never;
left: Academia (n=44-72), right: Pharma (n=32-47)

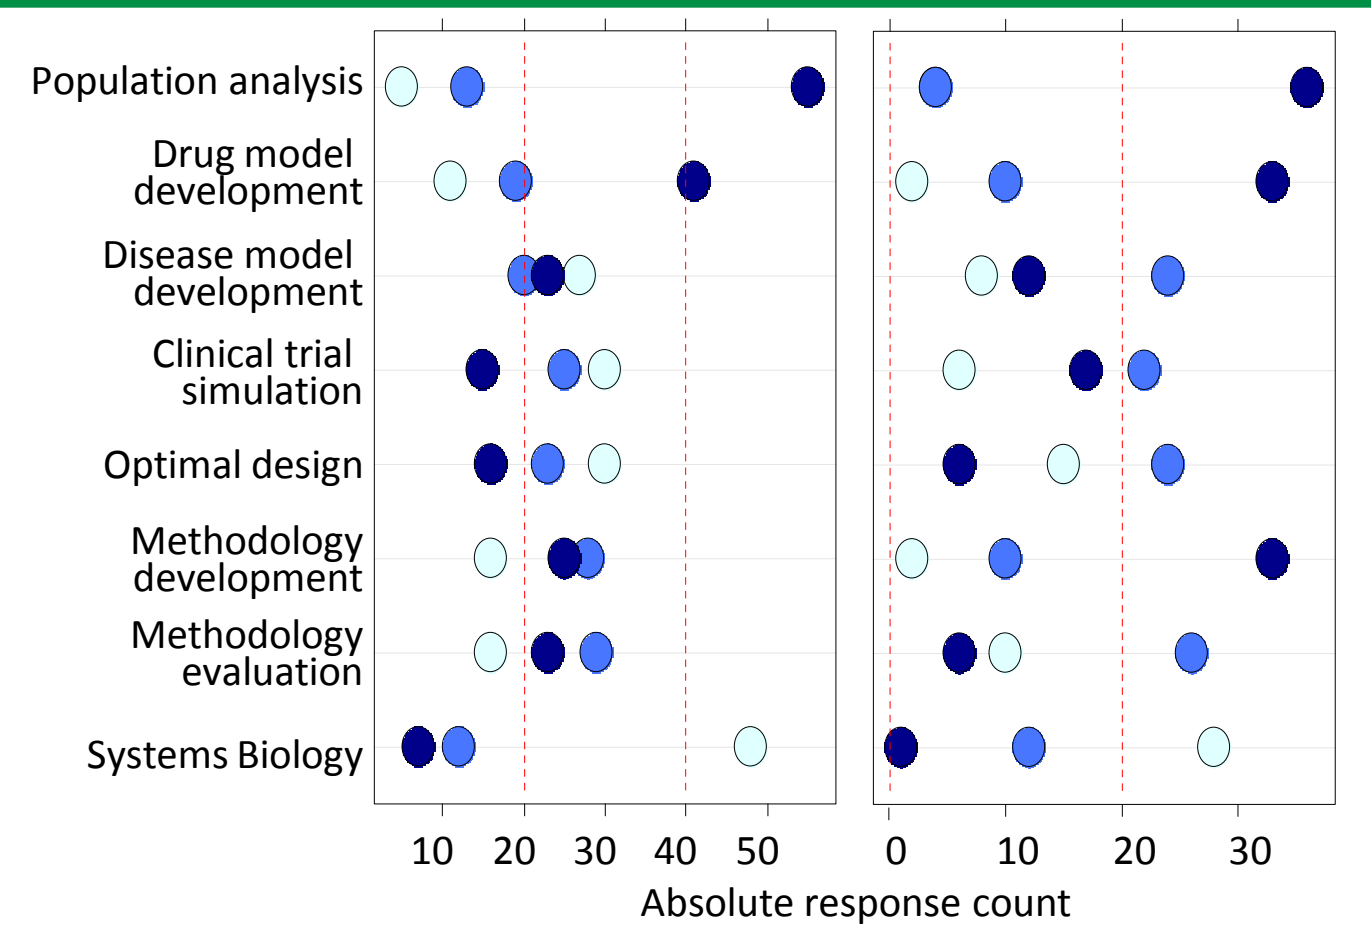

Population analysis
Drug model development
Disease model development
Clinical trial simulation
Optimal design
Methodology development
Methodology evaluation
Systems Biology
10 20 30 40 50
0 10 20 30
Absolute response count

Fig. 7: ***DD M&S activities/method. concepts:*** ● frequently, ● occasionally, ❍ never; left: Academia (n=34-72), right: Pharma (n=23-45)

DD M&S activities/methodol. concepts (Fig.7)

- Most important methodological concepts of DD M&S: population analysis
- Methodological developments performed both in industry in Academia
- As expected, Pharma more often involved in disease model development, clinical trial simulation, and optimal design

Domain IV. "Gaps & Challenges"

Gaps in environment (Fig. 8)

- Significant (major and minor) gaps for all defined categories
- Differences in gaps on 'high-performance computers', 'education' and 'training': More pronounced in Academia than in Pharma

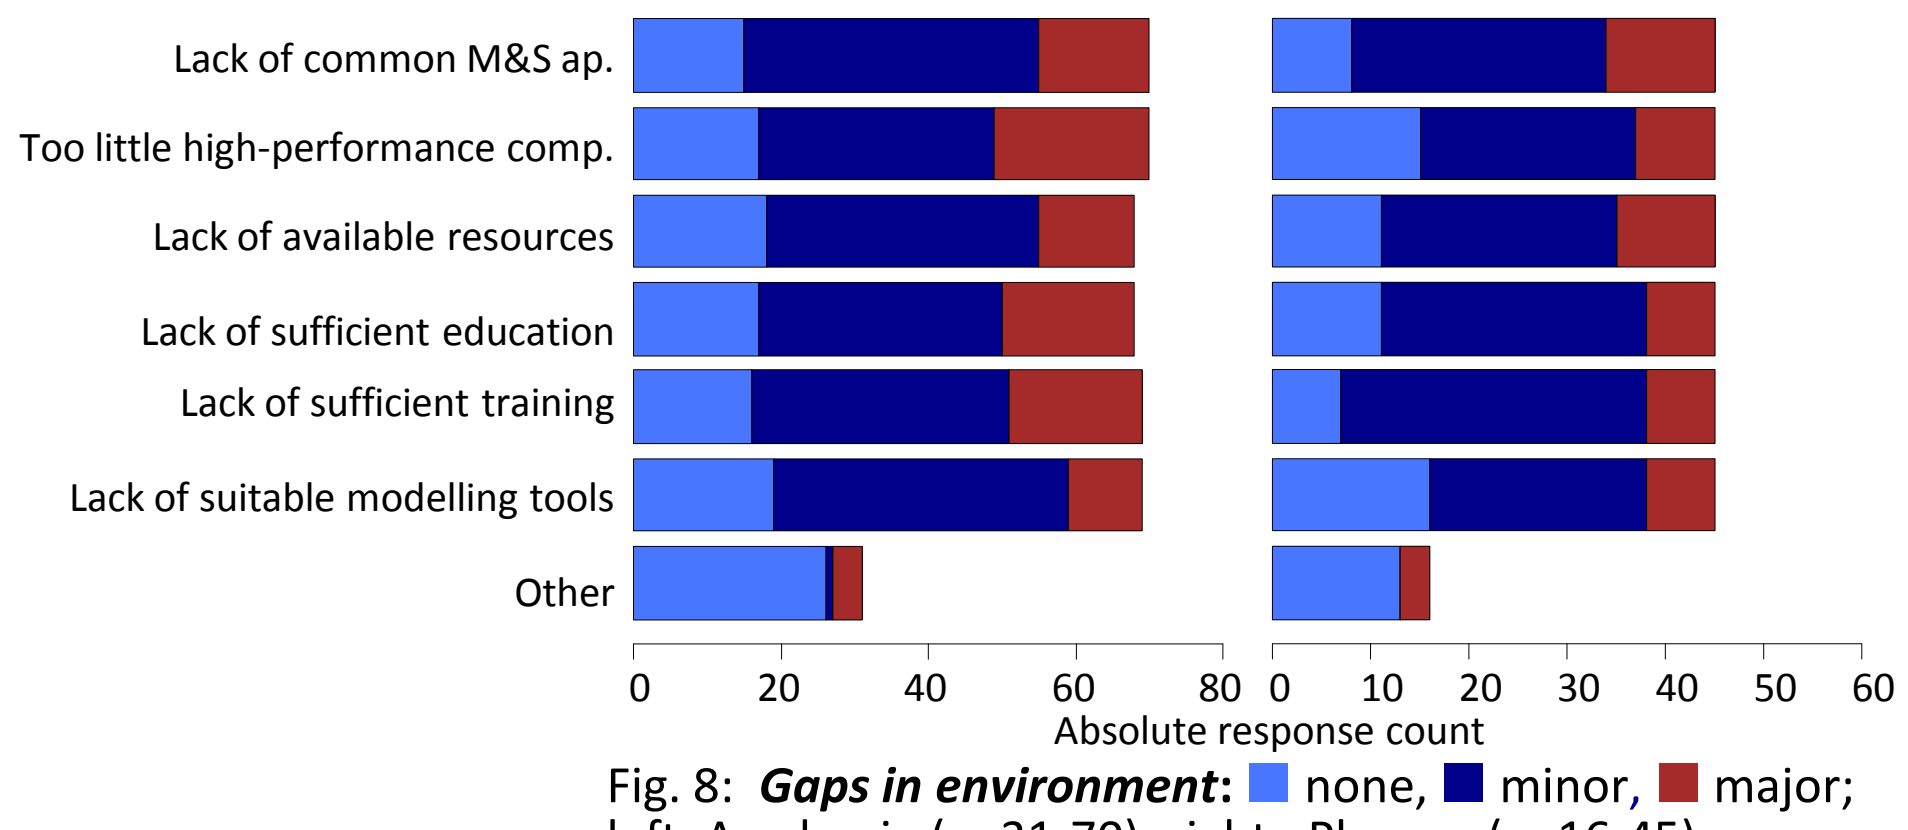

Lack of common M&S ap.
Too little high-performance comp.
Lack of available resources
Lack of sufficient education
Lack of sufficient training
Lack of suitable modelling tools
Other
0 20 40 60 80
0 10 20 30 40 50 60
Absolute response count

Fig. 8: ***Gaps in environment***: ■ none, ■ minor, ■ major;
left: Academia (n=31-70), right: Pharma (n=16-45)

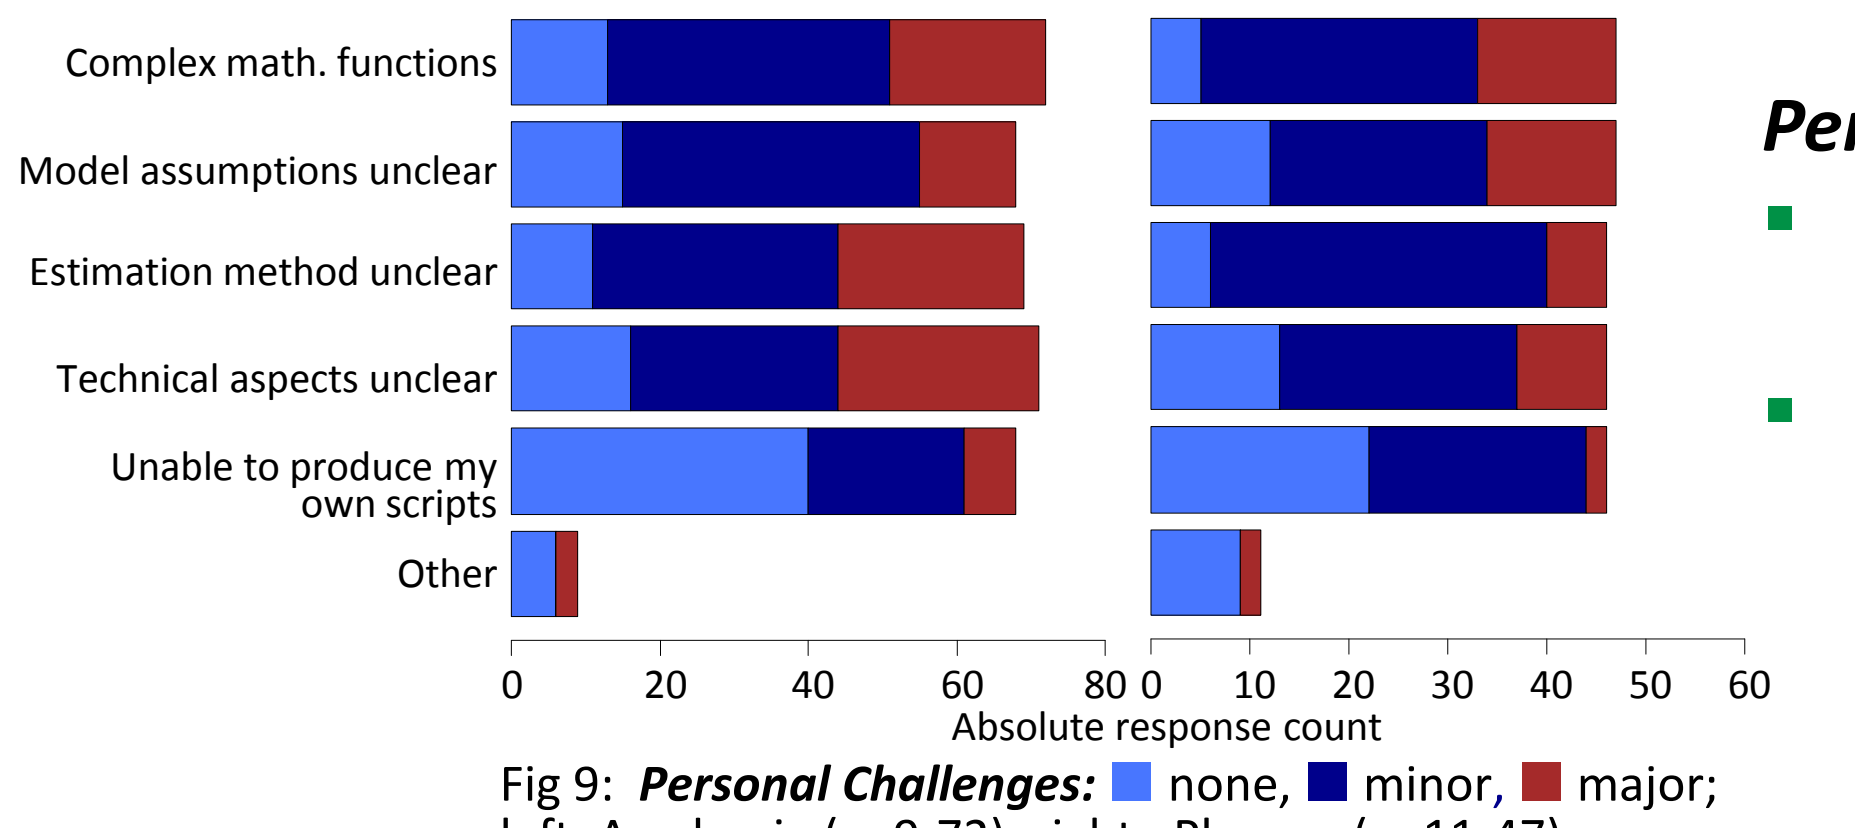

Complex math. functions
Model assumptions unclear
Estimation method unclear
Technical aspects unclear
Unable to produce my own scripts
Other
0 20 40 60 80
0 10 20 30 40 50 60
Absolute response count

Fig 9: ***Personal Challenges:*** ■ none, ■ minor, ■ major;
left: Academia (n=9-72), right: Pharma (n=11-47)

Personal Challenges (Fig. 9)

- Significant (major and minor) challenges for all defined categories
- Major challenges for 'estimation methods' and 'technical aspects': Much more pronounced in Academia than in Pharma

Domain V. "Willingness to share data/models for training & education purposes"

T&E highly benefit from implementing models based on real-life data. Whereas the vast majority are **willing to share published** models (~90%) and data (80%), the majority is **unwilling for unpublished** models or data: here, only less than half and only one third is willing to share models and data, respectively, for T&E purposes

Conclusions and Perspectives

The survey provided a landscape of role, impact, weaknesses and requirements in DD M&S primarily based on experts who develop or perform DD M&S. DD M&S has a **perceived impact** from under-standing fundamental mechanisms to supporting decision making both in drug development and therapeutic use of medicines. However, its **potential seem to be not fully exploited** yet. Areas of involvement of Academia and Pharma seem partly complementary. Due to the various levels of complexity in methodology, a **large panel of competences** is required to ensure performance and communication within and outside DD M&S community. More than 2/3 of responders, although often trained in several aspects of DD M&S, would **participate in further training** if given the chance.

In next steps, the landscape shall be used to **develop a competence framework** for **future training and education curricula**, which will go beyond technical and statistical aspects; understanding of the clinical implications of modelling assumptions as well as communication skills are urgently needed.

Acknowledgement: We thank David Wille (GSK) for statistical input. The research leading to these results has received support from the Innovative Medicines Initiative Joint Undertaking under grant agreement n° 115156, resources of which are composed of financial contribution from the European Union's Seventh Framework Programme (FP7/2007-2013) and EFPIA companies' in kind contribution.
The DDMoRe project is also financially supported by contributions from Academic and SME partners.