Population pharmacokinetic and exposure-lymphocyte count analysis of FTY720 (Fingolimod/Gilenya) in pediatric patients with multiple sclerosis

Author: Mita M Thapar[1], Colm Farrell[1], Gordon Graham[2], Olivier Petricoul[2]
Institution: [1]ICON Plc, [2]Novartis

1 Objectives

- to develop a linear mixed effects PK model to describe the FTY720-P (Fingolimod/Gilenya) concentrations at steady-state in the multiple sclerosis (MS) pediatric patient population using data from Study CFTY720D2311 (D2311).
- to assess by performing a visual predictive check (VPC) whether the previously developed adult FTY720-P exposure-lymphocyte count model described the pediatric lymphocyte data from study D2311 and to re-estimate the parameters of the exposure-lymphocyte model using a combined dataset from adult and pediatric patients.

2 Methods

Population PK model

Using the previously developed linear mixed effects PK model (see below) for the adult relapsing-remitting (RR) MS population as a starting point, a linear mixed effect model was developed to describe the FTY720-P concentrations at steady-state in the pediatric MS patients using data from Study D2311 alone. Age, weight and dose group were investigated (both as time-varying and time-invariant) covariates in the present analysis.

$$\mu = (\beta_0 + b_0) + (\beta_1 . weight) + (\beta_2 . gender) + (\beta_3 . dose)$$

$$Y \sim MVN\left\{\begin{bmatrix}\mu_1\\ \mu_2\\ \dots\\ \mu_n\end{bmatrix}, \sigma^2\begin{bmatrix}1 & \rho & \rho^2 & \\ & 1 & \rho & \\ & & \dots & \dots\\ & & & 1\end{bmatrix}\right\}, b_0 \sim N(0, \sigma_0^2)$$

where μ_1, $\mu_{2\dots\dots}$, μ_n correspond to the log of the estimated mean steady-state FTY720-P concentrations at n visits, β_0 is the intercept and b_0 is the random effect associated with the intercept, β_1 = weight effect on the intercept, β_2 = gender effect on the intercept and β_3 = dose effect on the intercept. The residual model was proposed to be additive on the log-scale.

Exposure-lymphocyte count model

An exploratory VPC was performed to assess whether the previously developed adult exposure-lymphocyte model (see below) described the pediatric lymphocyte data from Study D2311.

$$I_{max.cov} = I_{max} . e^{\eta_{Imax}} . (1 + \beta_2 . gender)$$

$$fC_p = \begin{cases}\dfrac{I_{max.cov} . C_p}{IC_{50} + C_p} & for\ FTY720\\ 0\ for\ placebo & \end{cases}$$

$$\mu = \beta_0 + \eta_{BSL} + \beta_1 . (WT - 70) + fC_p$$

$$\log(Y) \sim N(\mu, \sigma_{res}^2)\ and\ \begin{bmatrix}\eta_{BSL}\\ \eta_{Imax}\end{bmatrix} \sim MVN\left\{\begin{bmatrix}0\\0\end{bmatrix}, \begin{bmatrix}\sigma_{BSL}^2 & cov\\ cov & \sigma_{Imax}^2\end{bmatrix}\right\}$$

where, IC_{50} is the FTY720-P steady-state concentration that achieves 50% of the maximum extent of reduction (I_{max}), β_2 = gender effect on I_{max}, μ= model estimated log absolute lymphocyte count, β_0 = typical log lymphocyte level at baseline in all patients and the steady-state level in placebo-treated patients, β_1 = weight effect on baseline and η_{Imax} and η_{BSL} are the random effects associated with the I_{max} and lymphocyte count at baseline (β_0). σ_{res}^2 is the variance of the additive residual variability on the log scale.

The previously developed model in adults did not describe the pediatric data and therefore the parameters of the exposure-lymphocyte model were re-estimated using a combined dataset from adult and pediatric patients to determine which parameters were different between the adult and pediatric populations. A binary covariate on each model parameter (baseline, IC50 and Imax) was added to estimate the difference between adult and pediatric data.
The final population PK and exposure-lymphocyte count models were evaluated by performing a prediction-corrected VPC. NONMEM® program version VII level 3.0 was used for all analyses using PDx-Pop (Version 5.2) as an interface.

3 Results

Population PK model

- Following administration of 0.5 mg per day of FTY720 in a pediatric population, the typical steady-state FTY720-P concentration for a body weight of 70 kg (0.978 ng/mL) was within the 90% CI around the median of the adult target FTY720-P steady-state concentration and higher than the 65% relative bound (0.878 ng/mL) of the adult target FTY720-P steady-state concentration level.
- The steady-state FTY720-P concentrations were 49% lower following administration of 0.25 mg capsule compared to following administration of 0.50 mg capsule formulation thus confirming the dose-proportionality between the two dose levels.
- The steady-state FTY720-P concentrations decreased slightly with increasing body weight. The decrease in concentration was non-linear. An increase of 10 kg weight from 20 to 30 kg and from 70 to 80 kg resulted in a 15.2% and a 5.3% decrease in FTY720-P concentrations, respectively.

Goodness-of-fit plots for the final population PK model
Black circles: Observed concentrations, Red circles: Population predicted concentrations, Blue dashed line: Local regression (Loess) smoothing line

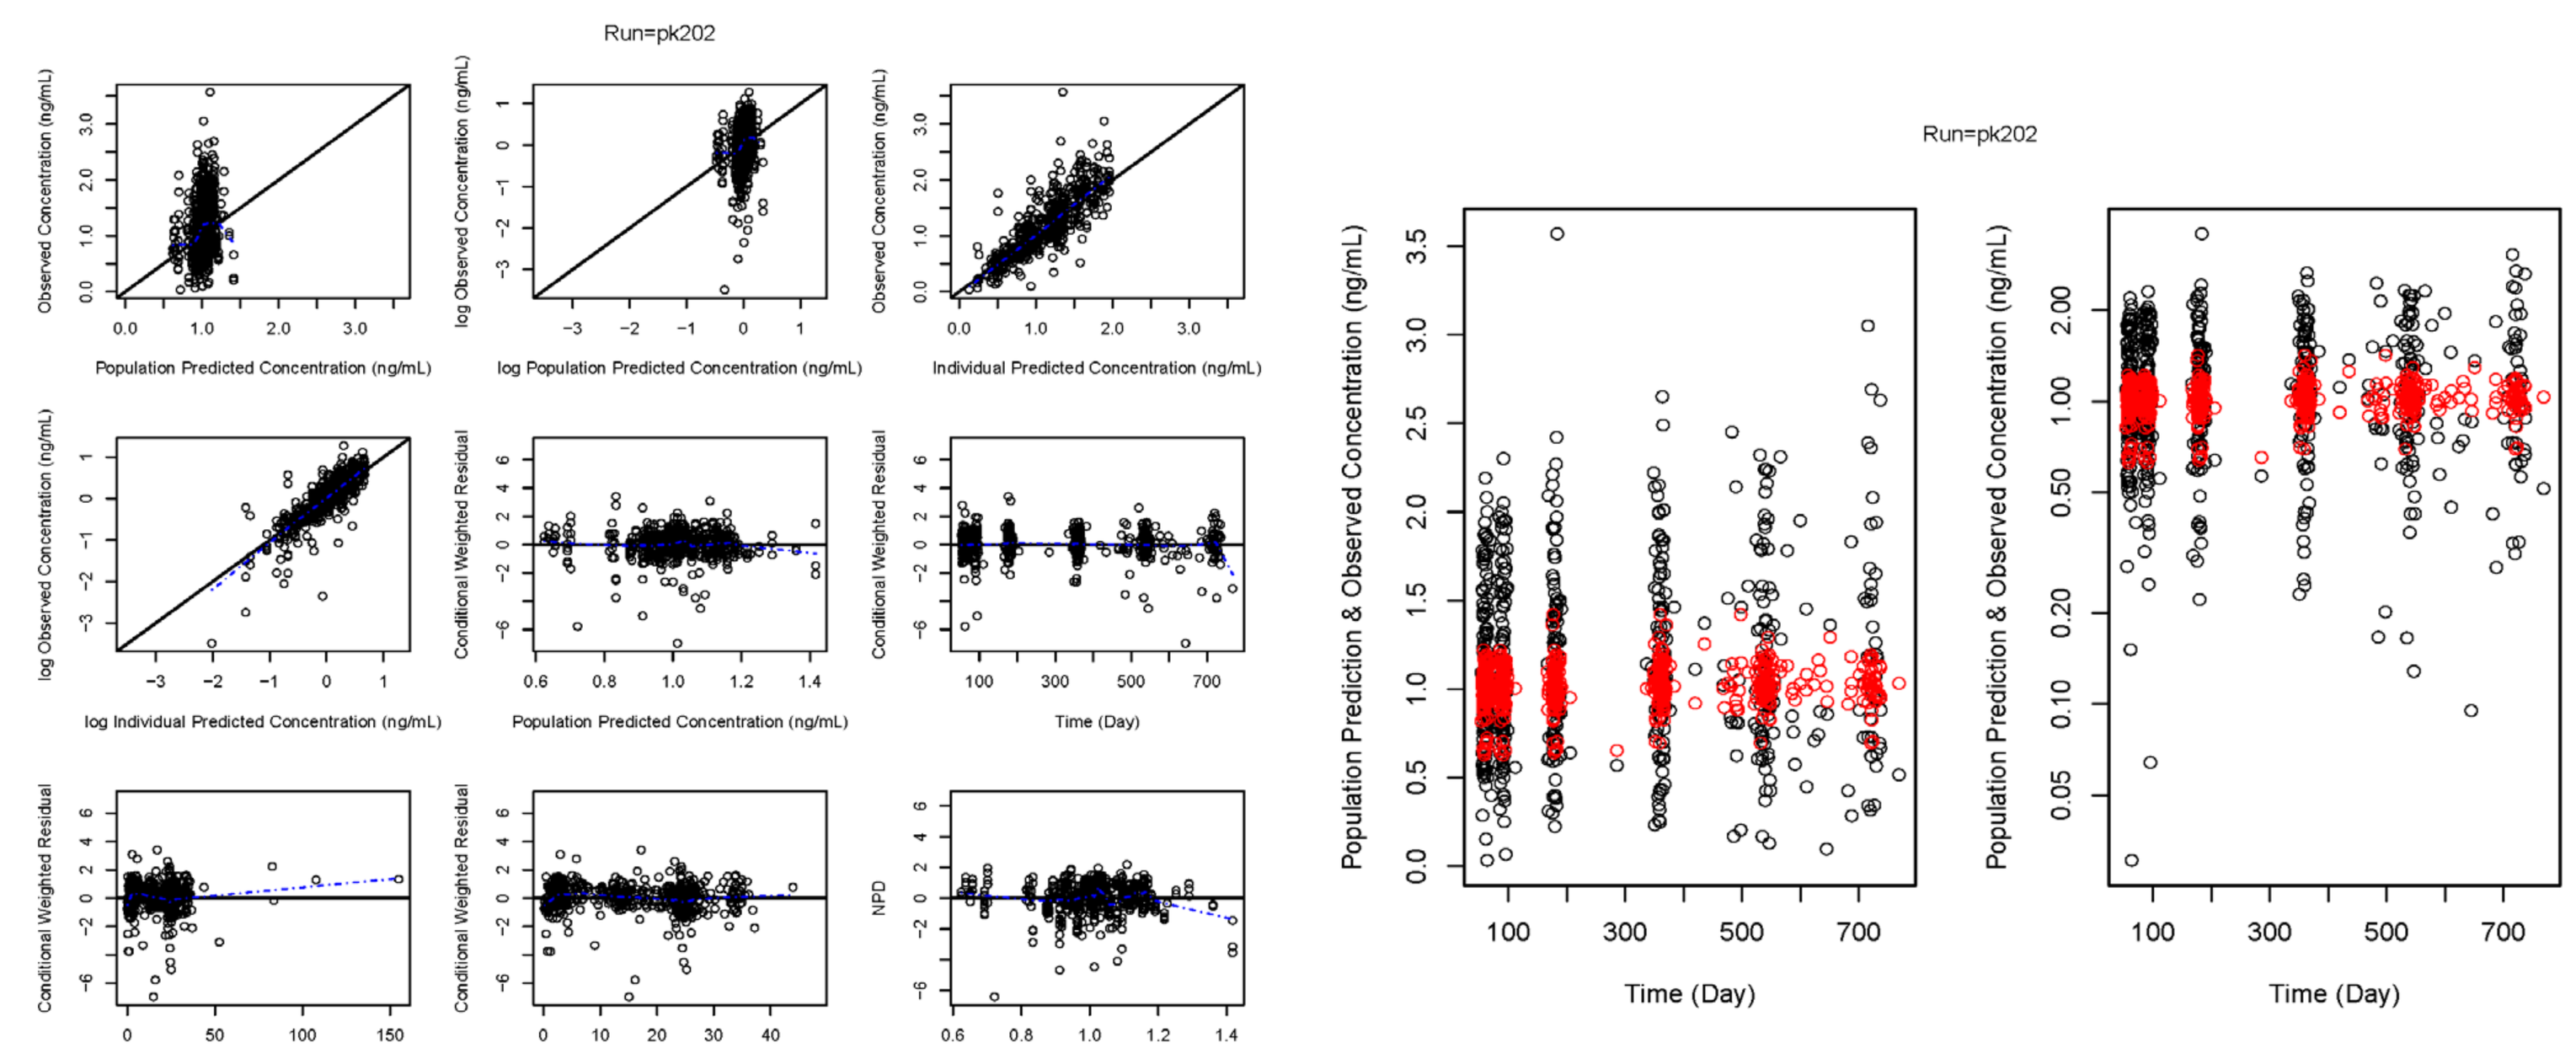
Run=pk202
Observed Concentration (ng/mL)
Population Predicted Concentration (ng/mL)
log Observed Concentration (ng/mL)
log Population Predicted Concentration (ng/mL)
Individual Predicted Concentration (ng/mL)
log Individual Predicted Concentration (ng/mL)
Conditional Weighted Residual
Time (Day)
Time after dose (hr)
NPD
Population Prediction & Observed Concentration (ng/mL)

Prediction-corrected VPC for the final population PK model
Open Circle: Observed Lymphocyte Count; Solid Line: Median of Observed Lymphocyte Count; Dashed Lines: 5th and 95th percentile of Observed Lymphocyte Count. Red Shaded Region: 95% Prediction Interval for Median of Predicted Lymphocyte Count; Blue Shaded Regions: 95% Prediction Intervals for the 5th and 95th percentiles of Predicted Lymphocyte Count

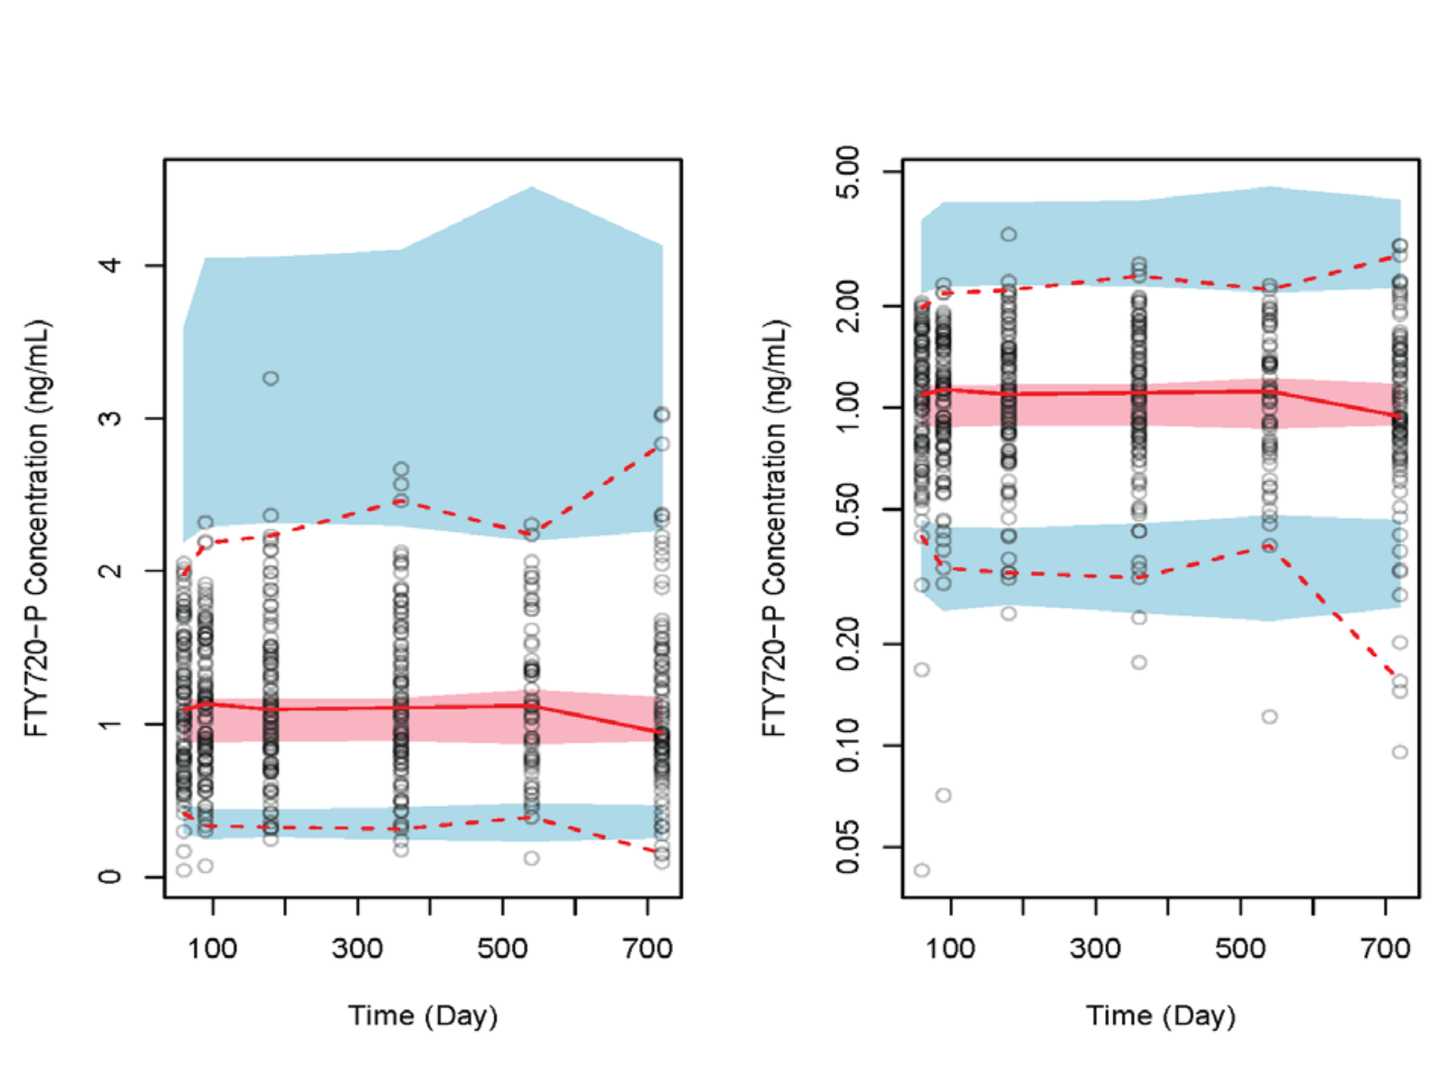
FTY720-P Concentration (ng/mL)
Time (Day)

4 Results (cont'd.)

Exposure-lymphocyte count model

- The FTY720-P exposure-lymphocyte relationship in adults is an I_{max} model relating the estimated FTY720-P concentrations at steady state to the absolute lymphocyte count.
- The baseline lymphocyte count was estimated to be 17.2% higher in pediatric patients compared to the adult population. Both I_{max} and IC_{50} were comparable between pediatric and adult populations with I_{max} 6% lower and IC_{50} 12% lower in pediatric versus adult patients.
- The baseline lymphocyte count was slightly higher for higher baseline weights. A 10-kg increase in weight (from 70 to 80 kg) resulted in only a 2% increase in lymphocyte count.

Goodness-of-fit plots for the final exposure-lymphocyte count model
Black circles: Observed lymphocyte count, Red circles: Population predicted lymphocyte count, Blue dashed line: Local regression (Loess) smoothing line

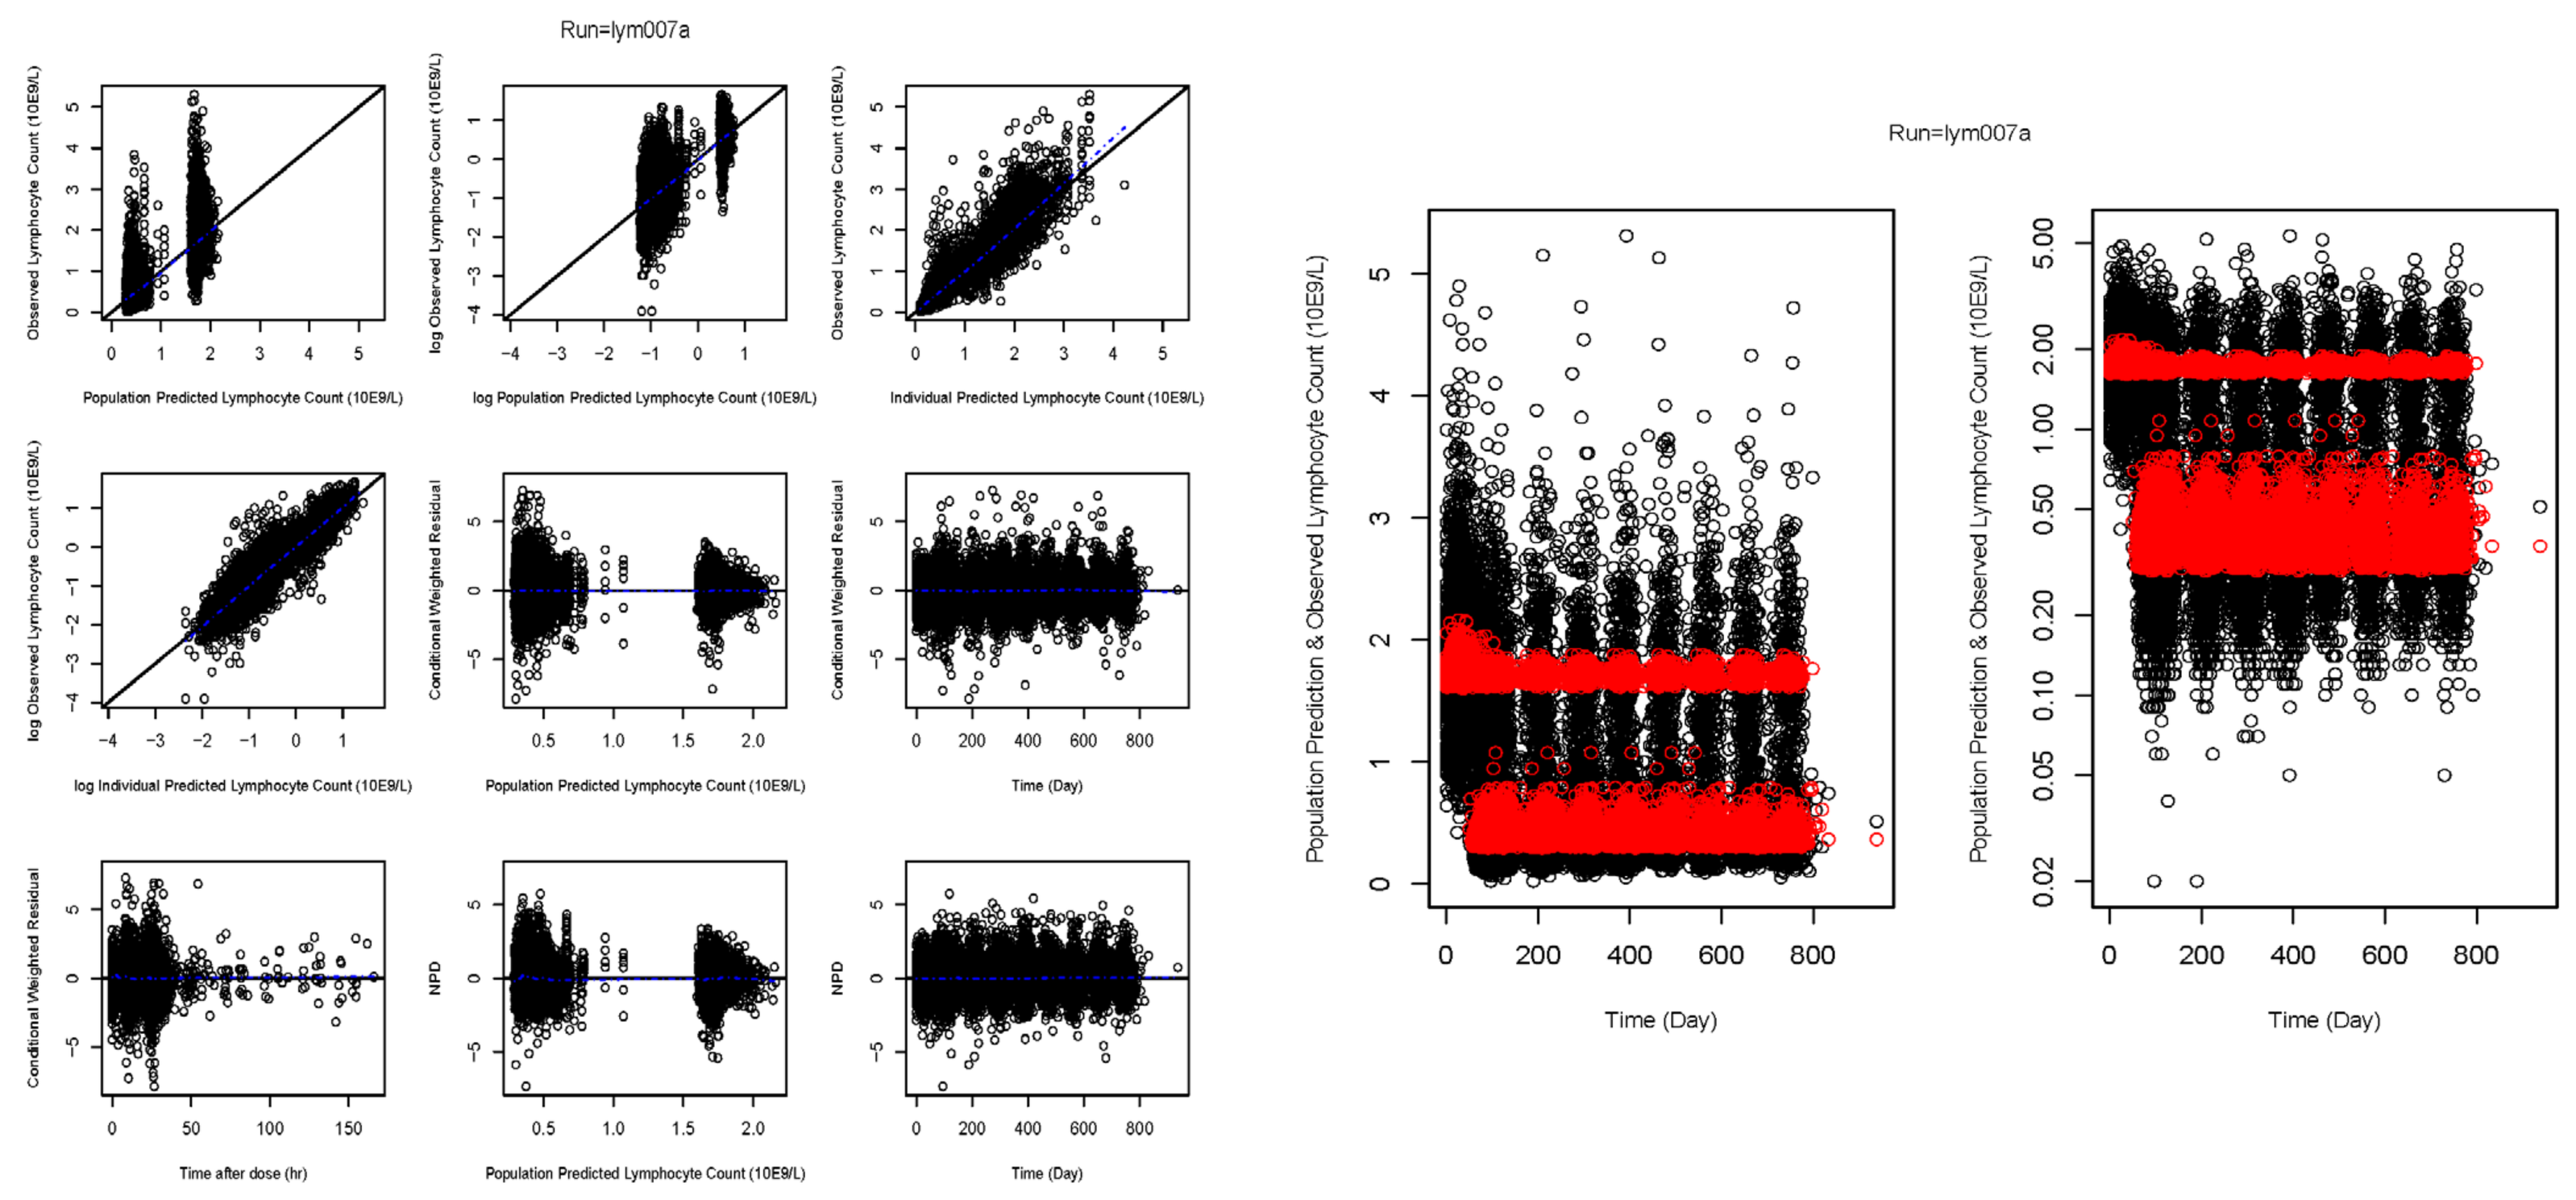
Run=lym007a
Observed Lymphocyte Count (10E9/L)
Population Predicted Lymphocyte Count (10E9/L)
log Observed Lymphocyte Count (10E9/L)
log Population Predicted Lymphocyte Count (10E9/L)
Individual Predicted Lymphocyte Count (10E9/L)
log Individual Predicted Lymphocyte Count (10E9/L)
Conditional Weighted Residual
Time (Day)
Time after dose (hr)
NPD
Population Prediction & Observed Lymphocyte Count (10E9/L)

Prediction-corrected VPC for the final exposure-lymphocyte count model
Open Circle: Observed Lymphocyte Count; Solid Line: Median of Observed Lymphocyte Count; Dashed Lines: 5th and 95th percentile of observed concentrations. Red Shaded Region: 95% Prediction Interval for Median of Predicted Lymphocyte Count; Blue Shaded Regions: 95% Prediction Intervals for the 5th and 95th percentiles of Predicted Lymphocyte Count

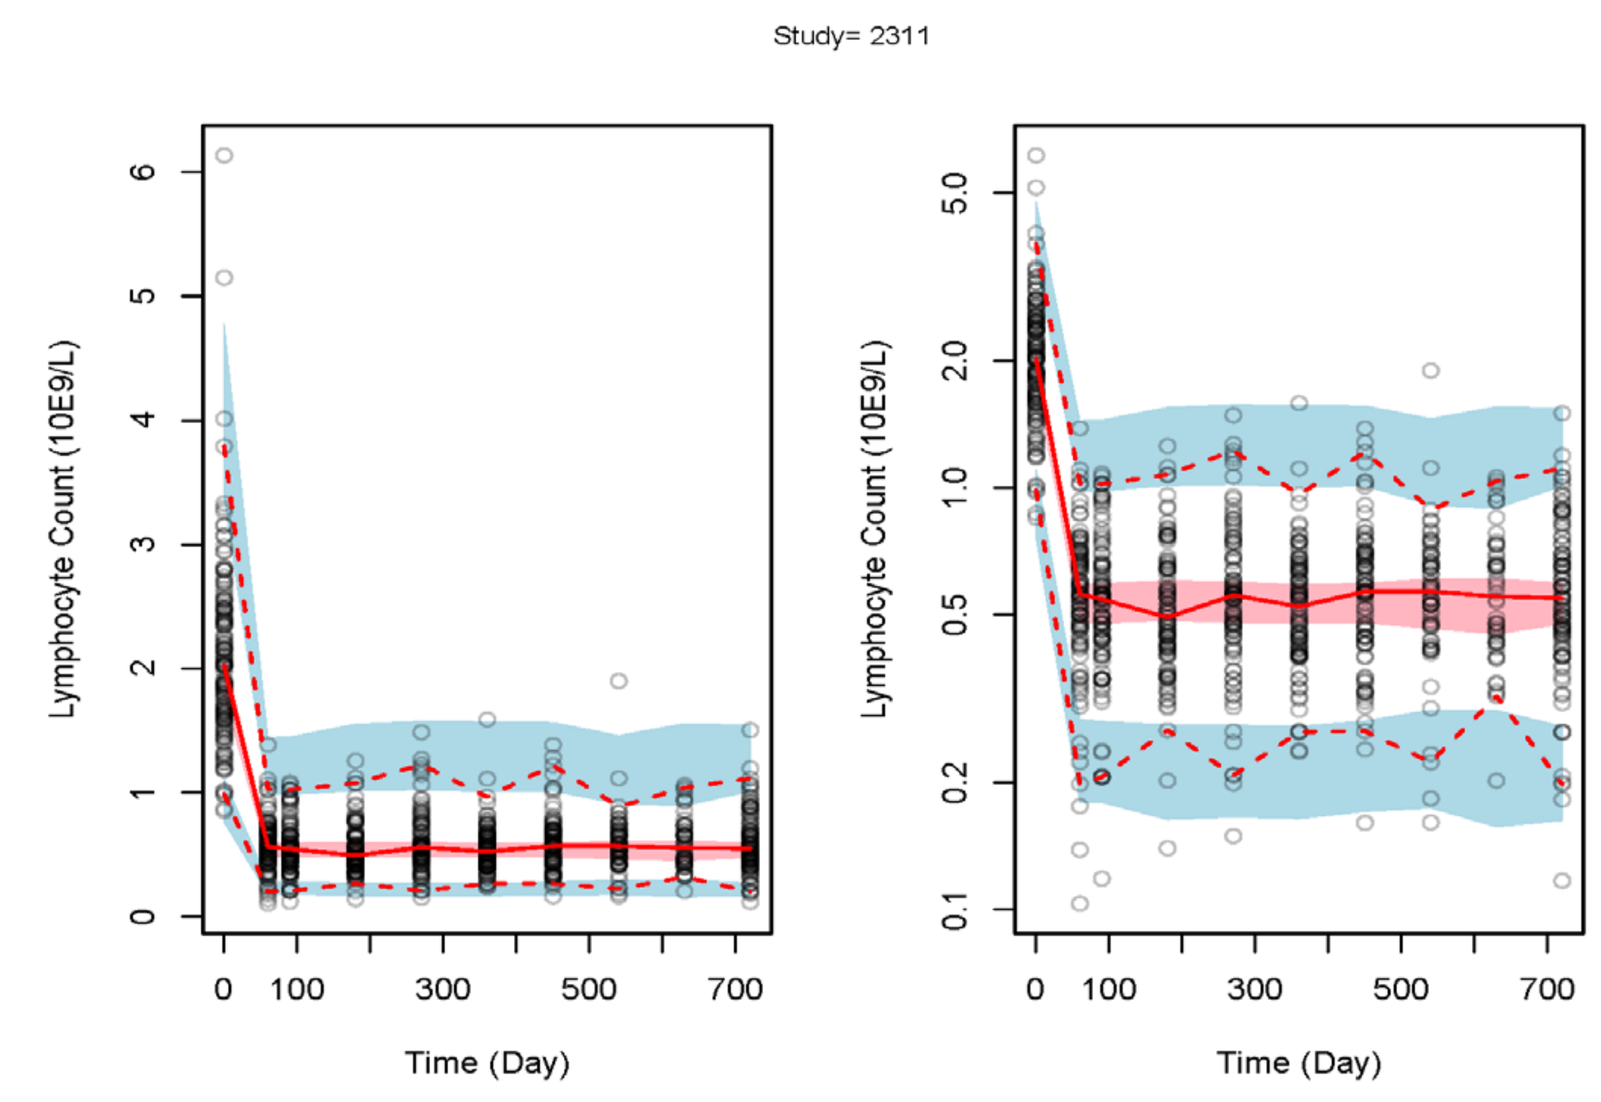
Study= 2311
Lymphocyte Count (10E9/L)
Time (Day)

5 Simulations

Predicted FTY720-P Steady-State Concentrations Using Final Population PK Model

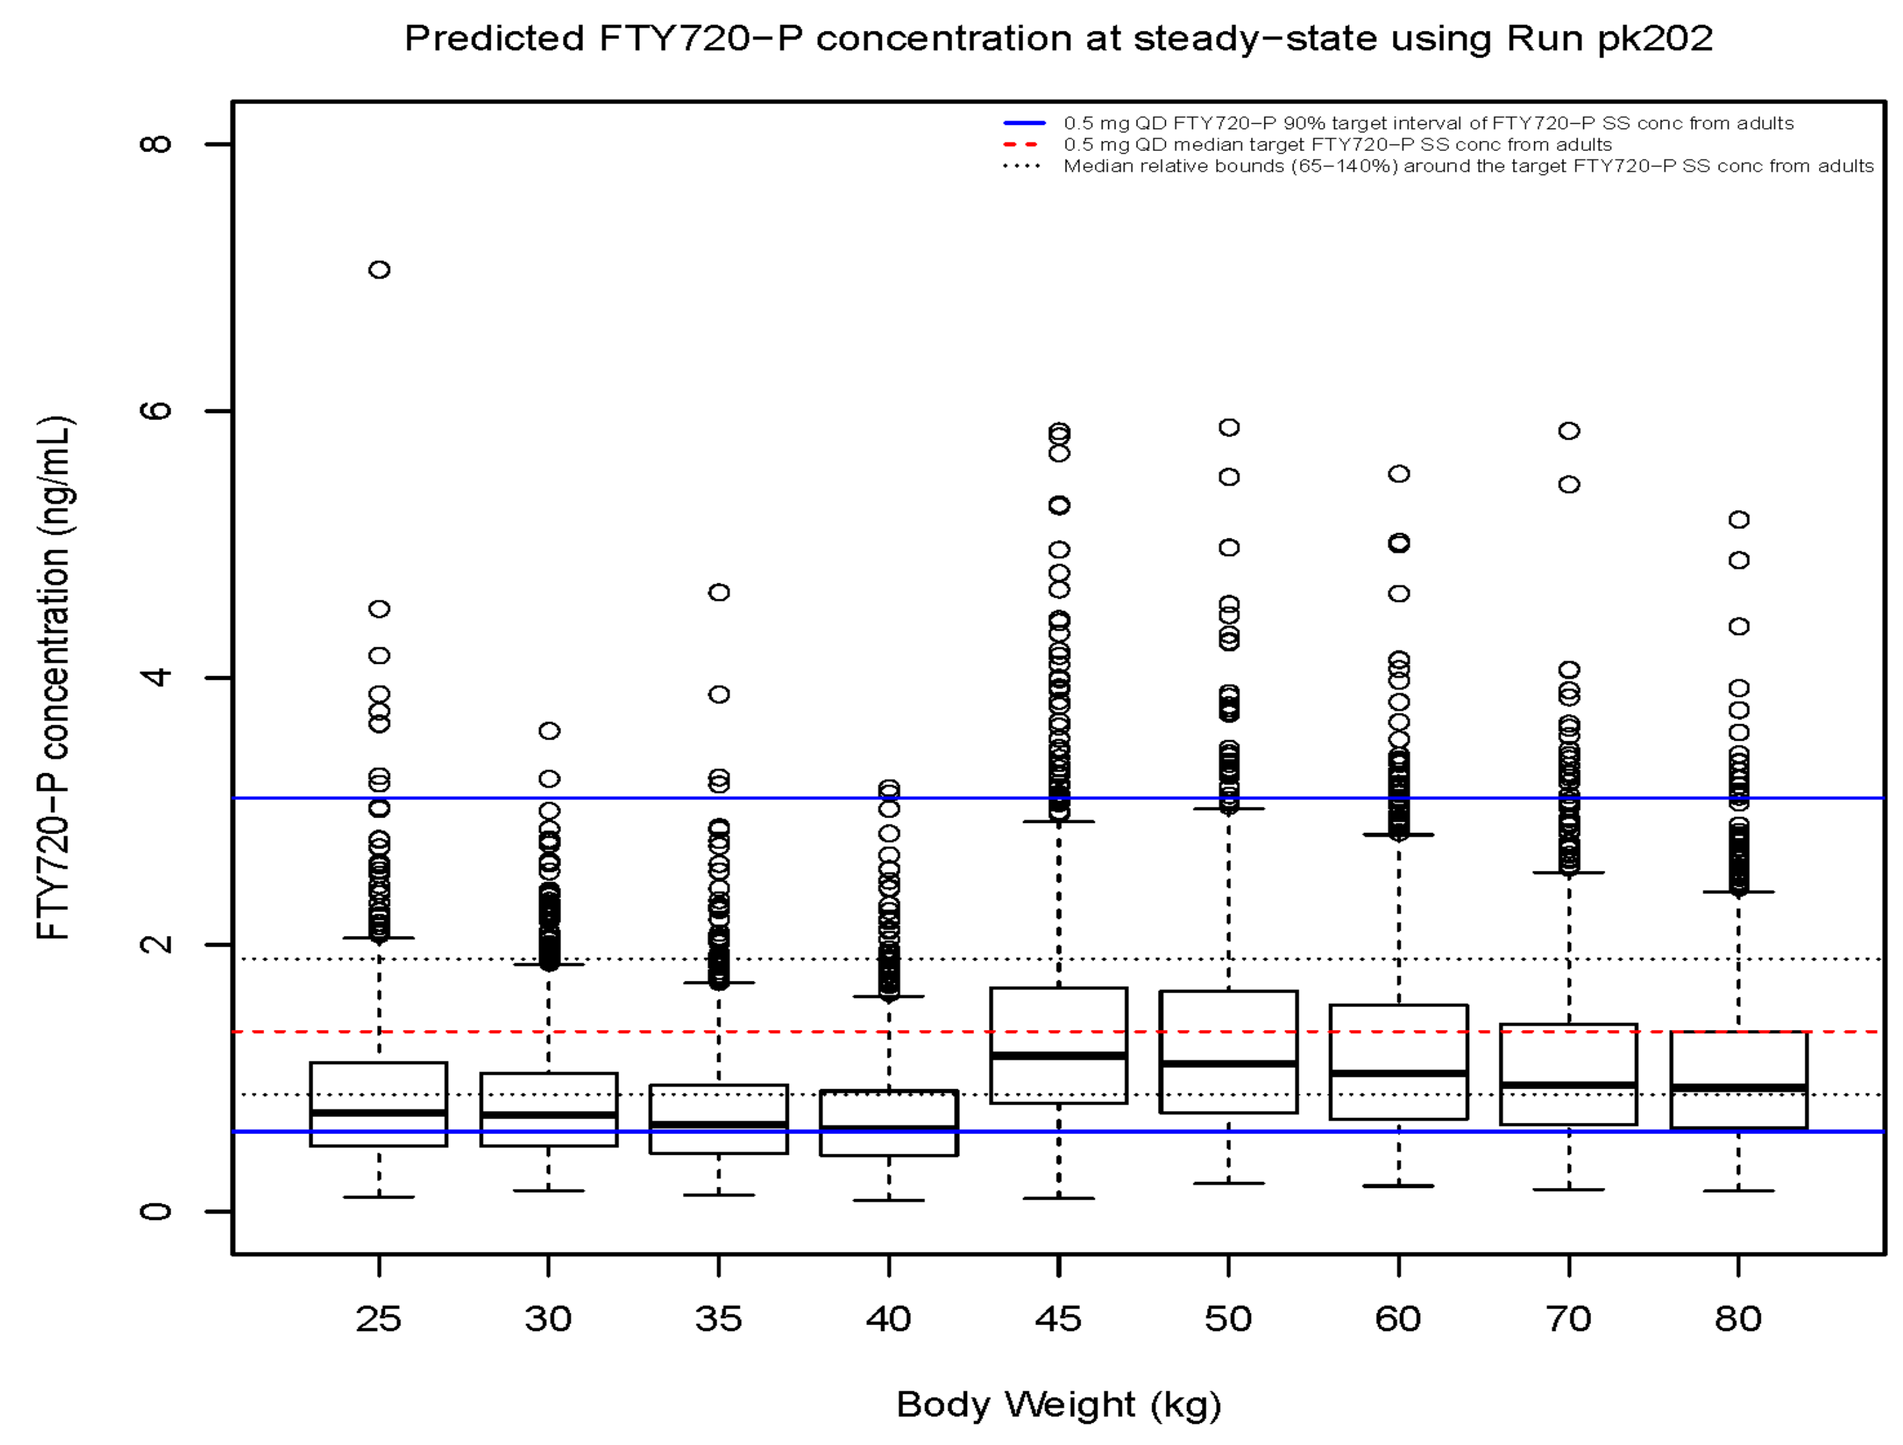
Predicted FTY720−P concentration at steady−state using Run pk202
0.5 mg QD FTY720−P 90% target interval of FTY720−P SS conc from adults
0.5 mg QD median target FTY720−P SS conc from adults
Median relative bounds (65−140%) around the target FTY720−P SS conc from adults
FTY720−P concentration (ng/mL)
Body Weight (kg)

6 Conclusions

Population PK model

- Following administration of 0.5 mg per day of FTY720 in a pediatric population, the typical steady-state FTY720-P concentration for a body weight of 70 kg (0.978 ng/mL) was slightly below the adult target FTY720-P steady-state concentration level (median: 1.35 ng/mL, (90% CI: 0.62, 3.1)) but within the 90% CI around the median of the adult target FTY720-P steady-state concentration. The typical steady-state FTY720-P concentration in pediatrics was also higher than the 65% relative bound (0.878 ng/mL) of the adult target FTY720-P steady-state concentration level.
- The steady-state FTY720-P concentrations were dose proportional following 0.25 mg and 0.50 mg capsule formulation of FTY720 in the pediatric population.
- The steady-state FTY720-P concentrations decreased slightly with an increasing weight. The decrease in concentration was non-linear. An increase of 10 kg weight from 20 to 30 kg and from 70 to 80 kg resulted in a 15.2% and a 5.3% decrease in FTY720-P concentrations, respectively.

Exposure-lymphocyte count model

- The baseline lymphocyte count was estimated to be 17.2% higher in pediatric patients compared to the adult population.
- Both I_{max} and IC_{50} were comparable between pediatric and adult populations with I_{max} 6% lower and IC_{50} 12% lower in pediatric versus adult patients.
- The baseline lymphocyte count was slightly higher for the higher baseline weight. A 10-kg increase in weight (from 70 to 80 kg) resulted in only a 2% increase in lymphocyte count.